In Vitro Characterization of a Novel TROP2-Targeting Antibody-Drug Conjugate OBI-992

Tzer-Min Kuo,[1] Ting-Yu Chang,[1] Jye-Yu Huang,[1] Wei-Chien Tang,[1] Chun-Jung Lin,[1] Yi-Chen Wu,[1] Chi-Huan Lu,[1] Hao-Cheng Weng,[1] Yu-Jung Chen,[1] Yu-Hsuan Tsao,[1] Cheng-Yen Wei,[1] Lifen Shen,[1] Wan-Fen Li,[1] and Ming-Tain Lai[1]

[1]OBI Pharma, Inc., Taipei, Taiwan

AACR 2024
3130

BACKGROUND

- Antibody-drug conjugates (ADCs) are promising anticancer therapeutics that have the potential for precise targeting using monoclonal antibodies conjugated to cytotoxic payloads through a chemical linker.[1,2]
- Trophoblast cell surface antigen 2 (TROP2), a transmembrane glycoprotein widely expressed in epithelial cancers, has emerged as an attractive target for ADC development.[3,4]
- Several ADCs are US Food and Drug Administration-approved or under clinical development, including the approved ADC sacituzumab govitecan, which is composed of the sacituzumab antibody conjugated to the topoisomerase inhibitor 1 (TOP1) SN-38 payload, and the investigational datopotamab deruxtecan (Dato-DXd), composed of the TROP2 antibody datopotamab conjugated to a DXd payload.[4]
- Exatecan, a water-soluble camptothecin analog, exhibits enhanced suppression of TOP1 activity and superior tumor suppression capabilities compared with other TOP1 inhibitor payloads.[5]
- OBI-992 is an investigational ADC composed of a TROP2 antibody (R4702) conjugated to the exatecan payload via a hydrophilic enzyme-cleavable linker (**Figure 1**).
- Pharmacokinetic and pharmacodynamic data on OBI-992 and its antitumor activity in vivo are presented in AACR 2024 posters 7179 and 1893, respectively.

Figure 1. OBI-992 is Composed of the TROP2 Antibody R4702 Linked to the Payload Exatecan via a Hydrophilic Linker

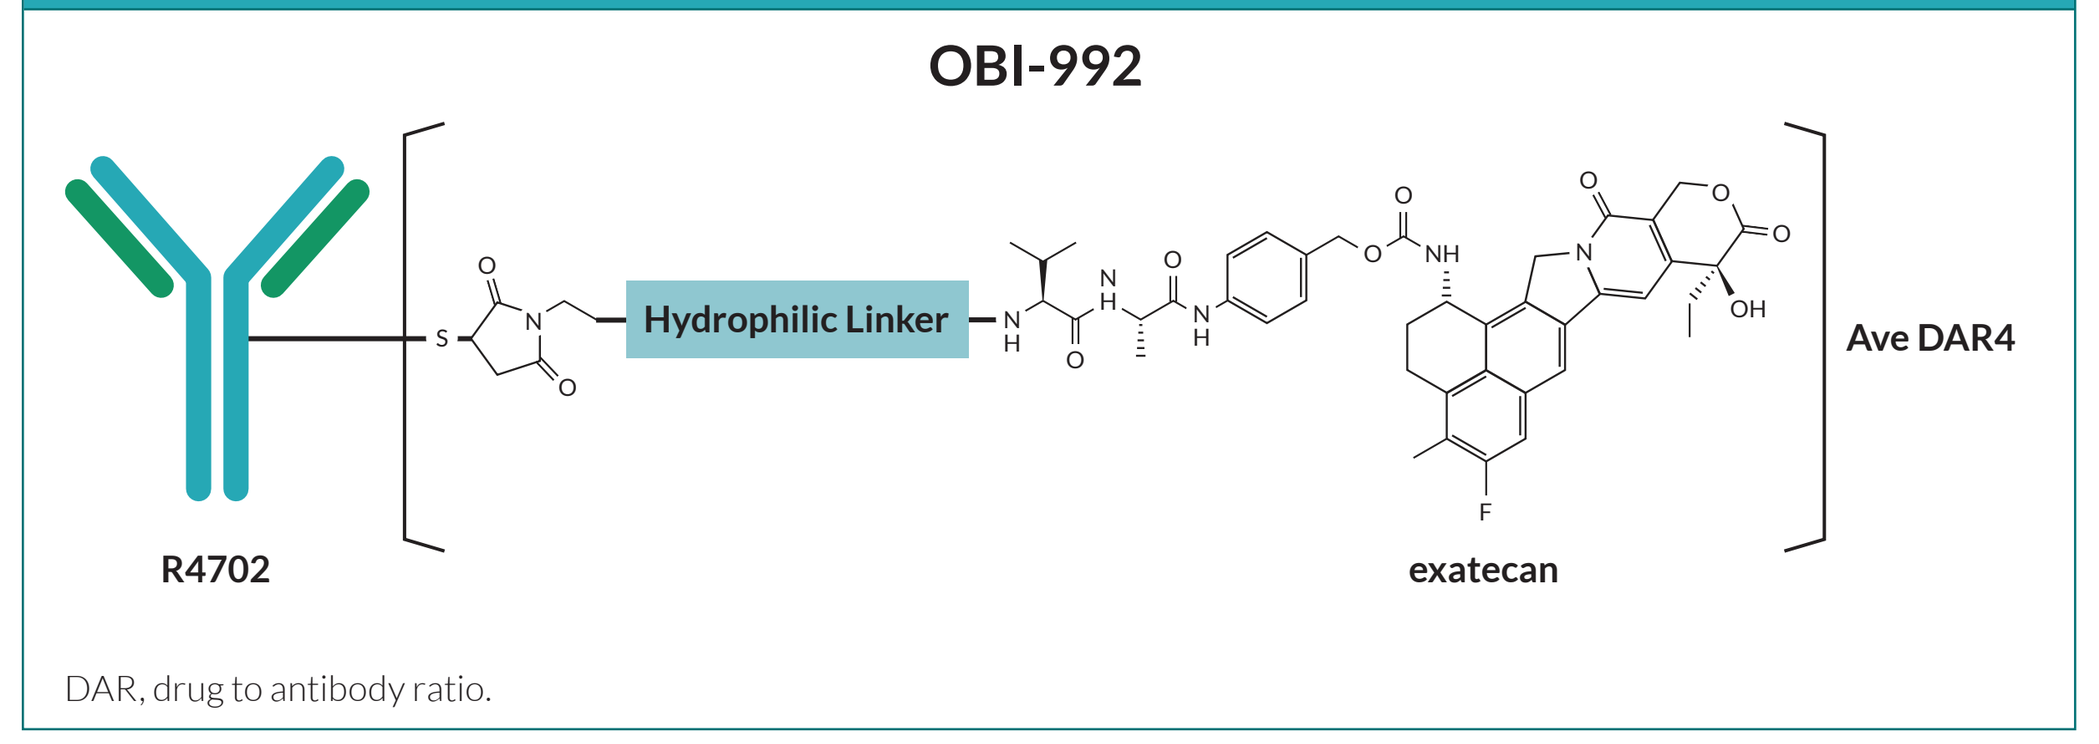
OBI-992
Hydrophilic Linker
Ave DAR4
R4702
exatecan

DAR, drug to antibody ratio.

METHODS

- The prognostic roles of TROP2 and TOP1 were determined by analyzing the "*Kaplan-Meier Plotter*" database[6] for over 1055 patients with colon cancer.
- An enzyme-linked immunosorbent assay (ELISA) study was conducted to investigate the binding epitope of R4702.
 - The peptides of indicated TROP2 domains were incubated with R4702, and absorbances at 450 nm were measured.
- In competition binding assays, BxPC3 cells were sequentially treated with an unlabeled antibody, followed by incubation with a fluorescein-labeled antibody; mean fluorescence intensity was quantified using flow cytometry.
- Cell binding assays and surface plasmon resonance (SPR) were carried out to measure non-specific binding activity and binding affinity, respectively.
 - In cell binding assays, each TROP2 antibody was first immobilized on the protein A sensor chip. Data were fit globally to different binding schemes that corresponded to a 1:1 model binding in sensogram plots that were generated by SPR kinetic analysis.
 - In cell surface binding assays, MCF7 and NCI-N87 cells were incubated with TROP2 antibodies, and mean fluorescence intensity was quantified.
 - Whole blood samples from two donors were incubated with fluorescein-labeled ADCs for 4 hours to assess the association with peripheral blood leukocytes. Mean fluorescence intensity of each leukocyte population was measured to represent the strength of cell association. Herceptin served as the human immunoglobulin G1 (IgG1) isotype control.
 - To assess neutrophil viability, differentiated neutrophils were treated with ADCs for 6 days. Viable neutrophils (CD66b+/PI-) were identified by fluorescence-activated cell sorting (FACS); absolute counts were determined by referring to Count-Bright Beads.
- DNA damage activity, cytotoxicity, and immunogenic cell death (ICD) effects of 3 TOP1 inhibitors exatecan, DXd, and SN-38, were evaluated.
 - Luciferase assays of ICD markers including extracellular ATP and HMGB1 in TOP1 inhibitor (TOPi)-treated MC38 with over-expressed TROP2 were carried out.
 - A western blot analysis of DNA damage markers, RPA2 pS4/S8 and γH2AX in MDA-MB-231 cells treated with TOPi for 3 hours was performed.
 - Cell viability was measured in cells treated with TOPi for 6 days.
 - The cytotoxicity of OBI-992 and other ADCs was measured using 3 two-dimensional (2D) cancer cell lines (breast cancer MDA-MB-231, gastric cancer NCI-N87, and pancreatic cancer Capan-1) and 1 three-dimensional (3D) culture of prostate cancer DU145 cells.
- To examine ADC-resistant cell lines, western blot analyses of proteins were carried out in DLD1 cells.
 - TROP2 and breast cancer resistance protein (BCRP) expression and potential mutations in TOP1 were investigated in TROP2 ADC-induced resistant colon cancer cells.
 - To identify a TOP1 mutation in the ADC-resistant DLD1 cells, the RNA of TOP1 was extracted and sequenced by standard reverse transcription-polymerase chain reaction amplification.
 - The crystal structure of the TOP1 inhibitor binding site of TOP1 (PDB = 1T8I) was conducted using PyMOL software.
- The synergistic effects of the combination of OBI-992 with poly (ADP-ribose) polymerases (PARP) inhibitors were evaluated in vitro.

OBJECTIVE

To identify the prognostic value of TOP1 and TROP2 and to investigate and compare the characteristics of OBI-992 and its components with that of benchmarks sacituzumab govitecan and datopotamab deruxtecan.

RESULTS

- High mRNA expression levels of both *TROP2* and *TOP1* significantly correlated with worse overall survival in patients with colon cancer (**Figure 2**).

Figure 2. Evaluation of Prognostic Value of *TROP2(TACSTD2)* and *TOP1* mRNA Expression

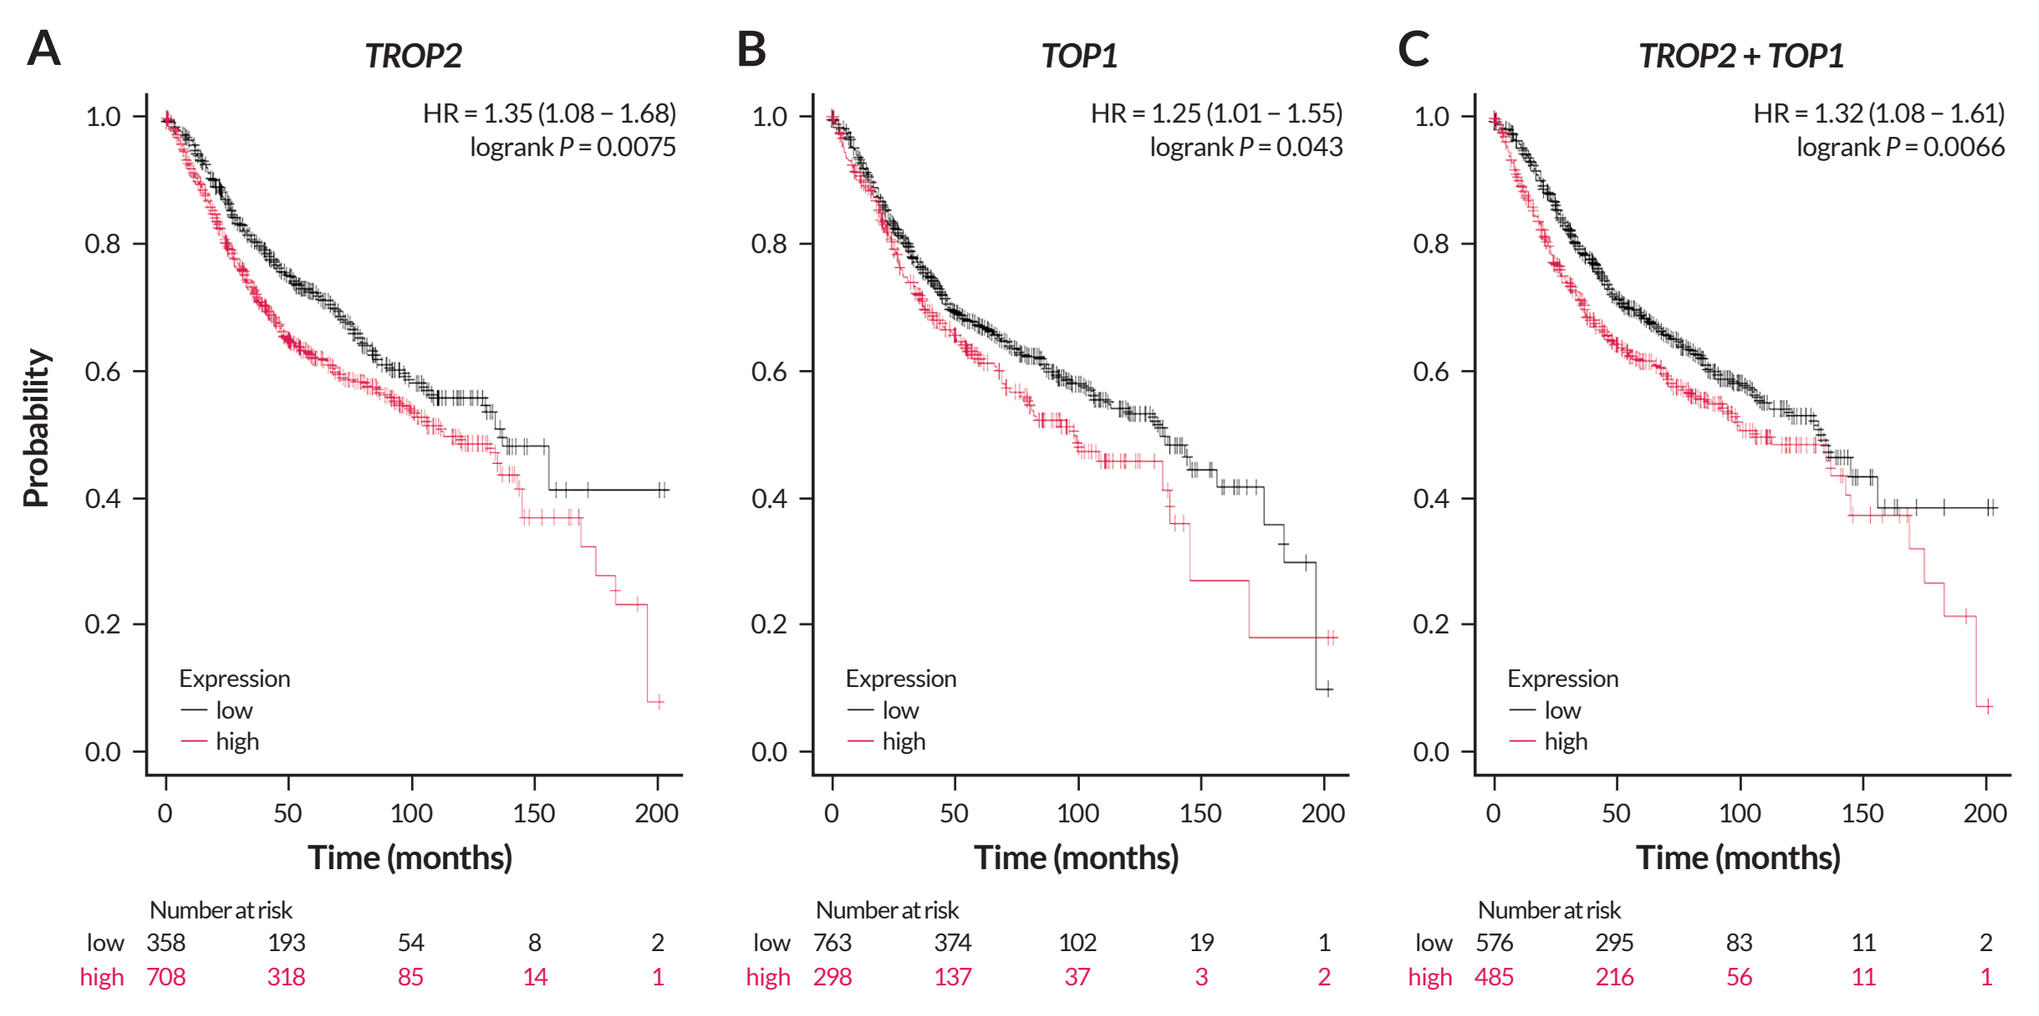
A TROP2 HR = 1.35 (1.08 – 1.68) logrank P = 0.0075
B TOP1 HR = 1.25 (1.01 – 1.55) logrank P = 0.043
C TROP2 + TOP1 HR = 1.32 (1.08 – 1.61) logrank P = 0.0066
Probability
Time (months)
Expression low high

The association of (A) *TROP2*, (B) *TOP1*, and (C) the combination of *TROP2* and *TOP1* mRNA expression levels with overall survival using colon cancer sample data from the Kaplan-Meier Plotter.[6]
HR, hazard ratio; TOP1, topoisomerase 1; TROP2, trophoblast cell surface antigen 2.

- The binding epitope of the TROP2 antibody R4702 is distinct from that of sacituzumab and datopotamab (**Figure 3**).

Figure 3. R4702 Binds to a Distinct Region on TROP2

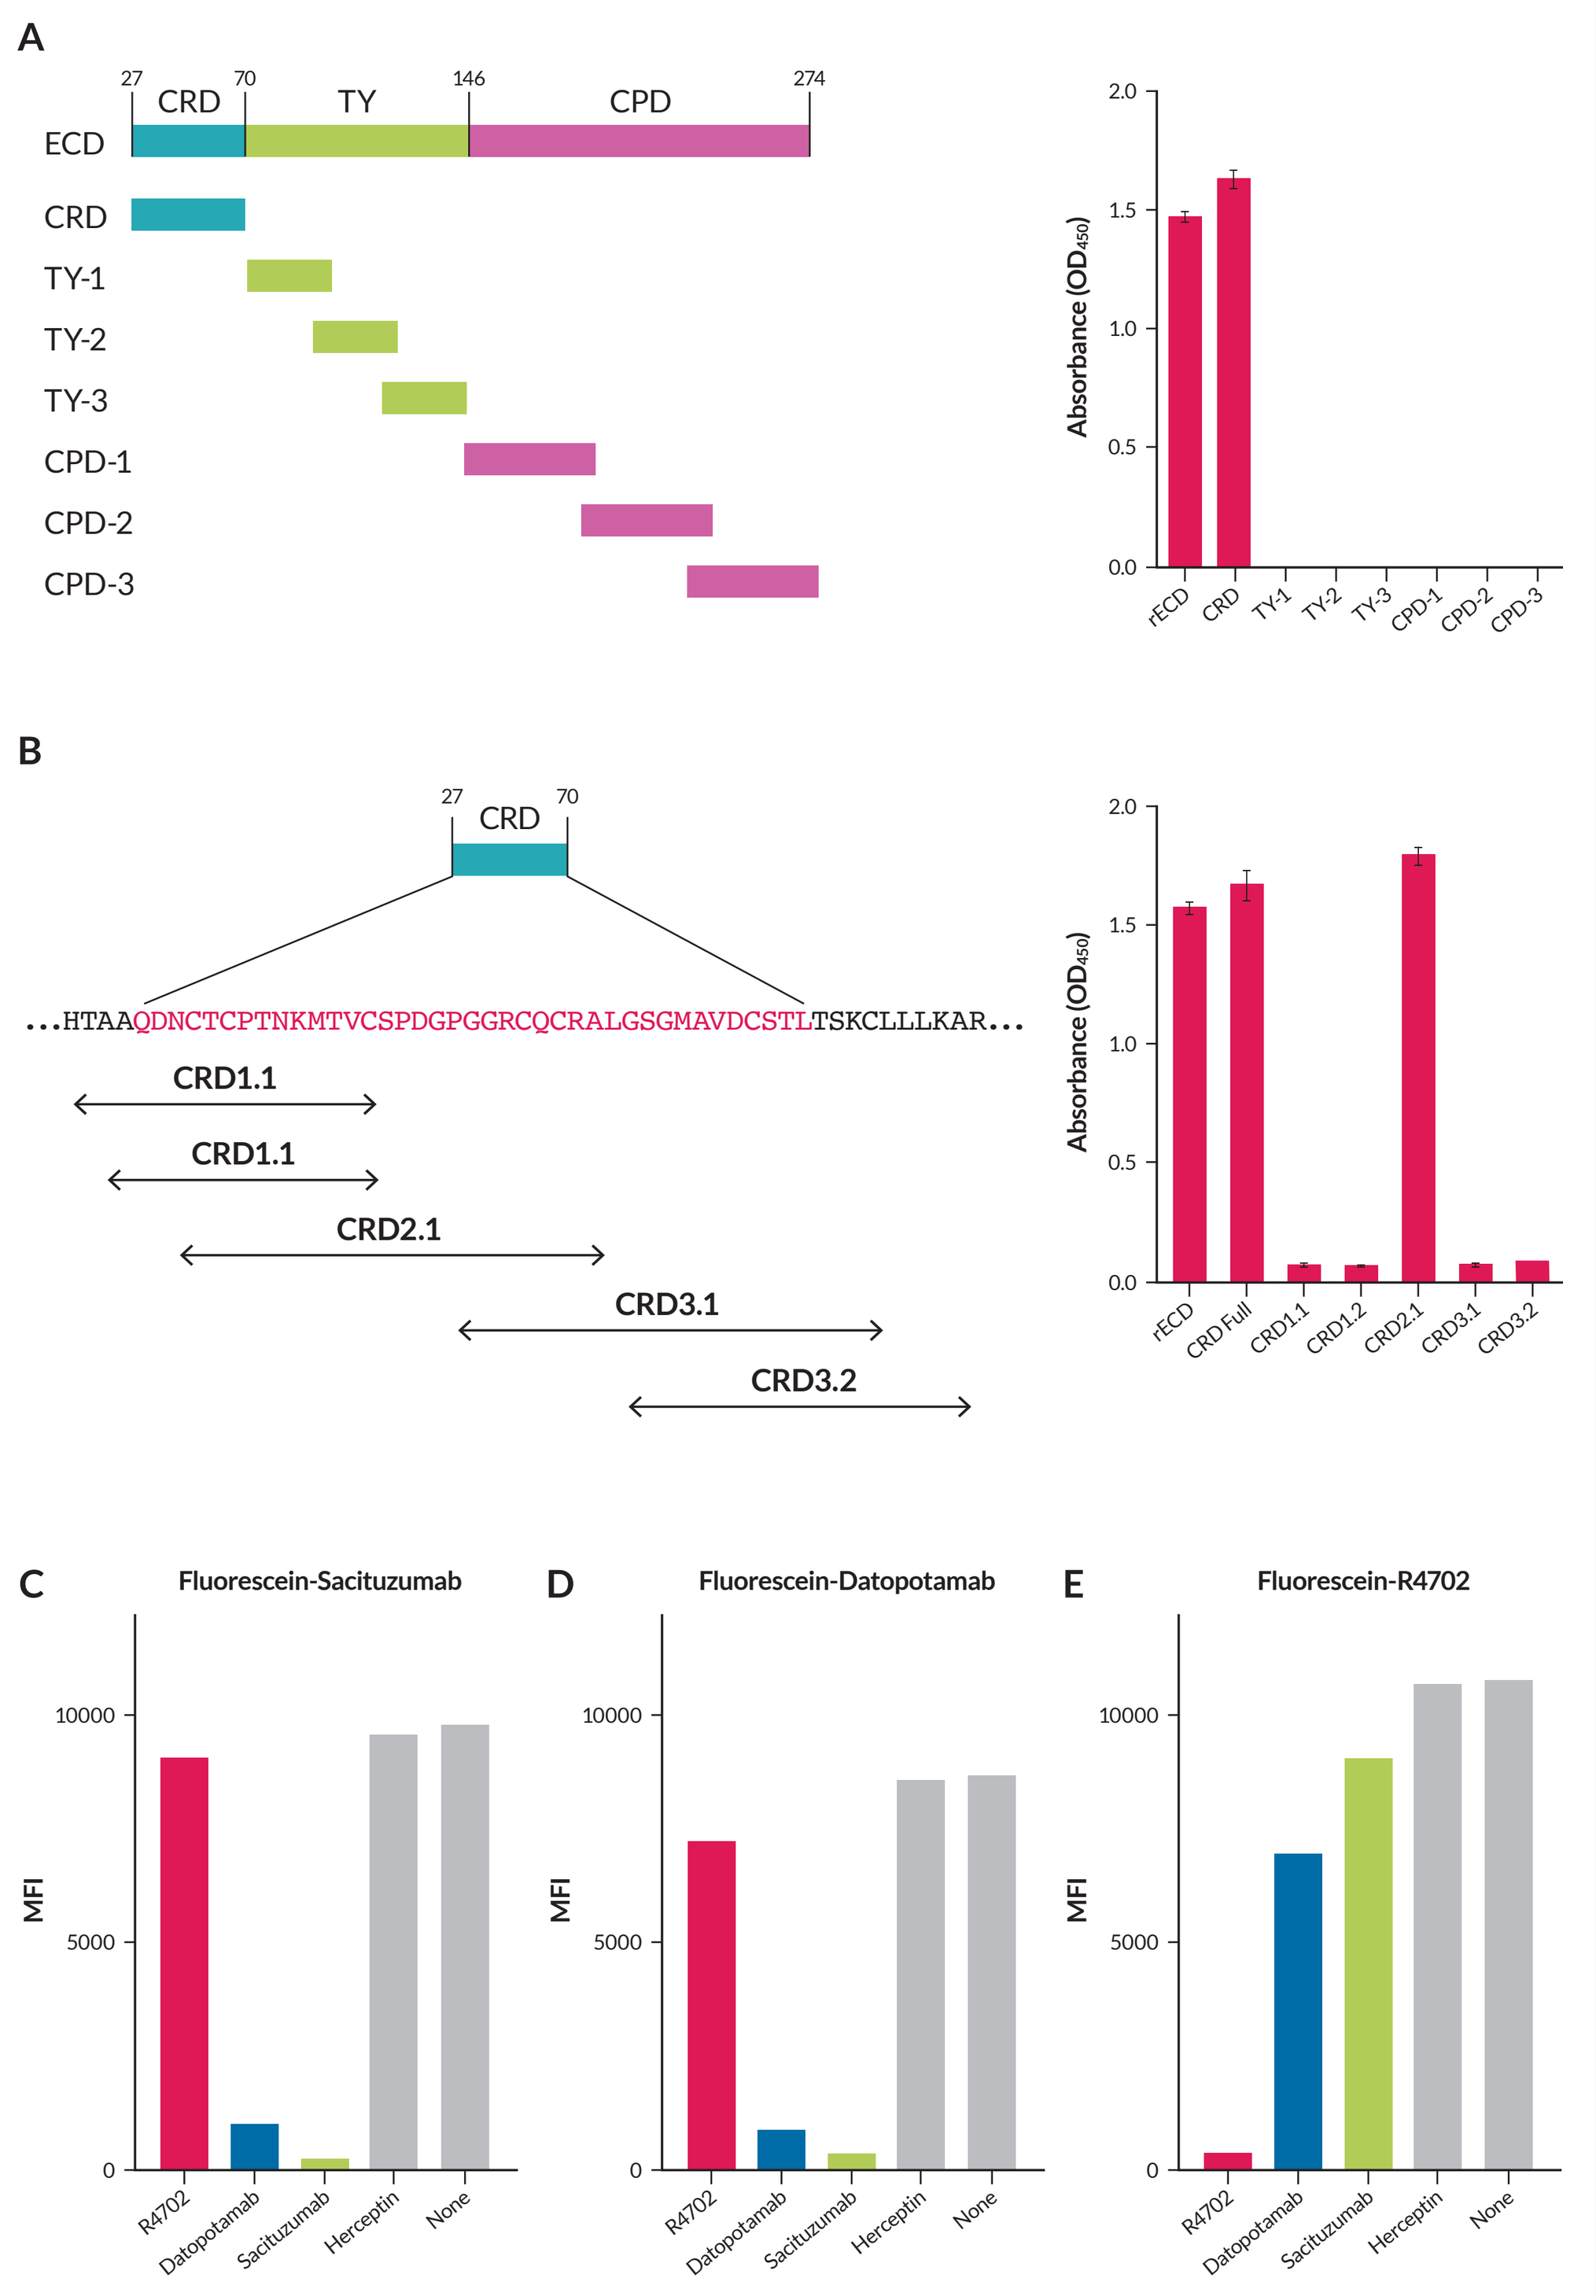
A
ECD CRD TY CPD
B
...HTAAQDNCTCPTNKMTVCSPDGPGGRCQCRALGSGMAVDCSTLTSKCLLLKAR...
CRD1.1
CRD2.1
CRD3.1
CRD3.2
Absorbance (OD450)
C Fluorescein-Sacituzumab
D Fluorescein-Datopotamab
E Fluorescein-R4702
MFI

(A-B) ELISA assay of R4702 binding epitope screen. (C-E) Competition binding assays.
ELISA, enzyme-linked immunosorbent assay; MFI, mean fluorescence intensity; TROP2, trophoblast cell surface antigen 2.

- R4702 and OBI-992 demonstrated a higher binding affinity and less non-specific binding than datopotamab (**Figure 4**).

Figure 4. R4702 Exhibits Higher Binding Affinity to the TROP2 Antigen Relative to Datopotamab

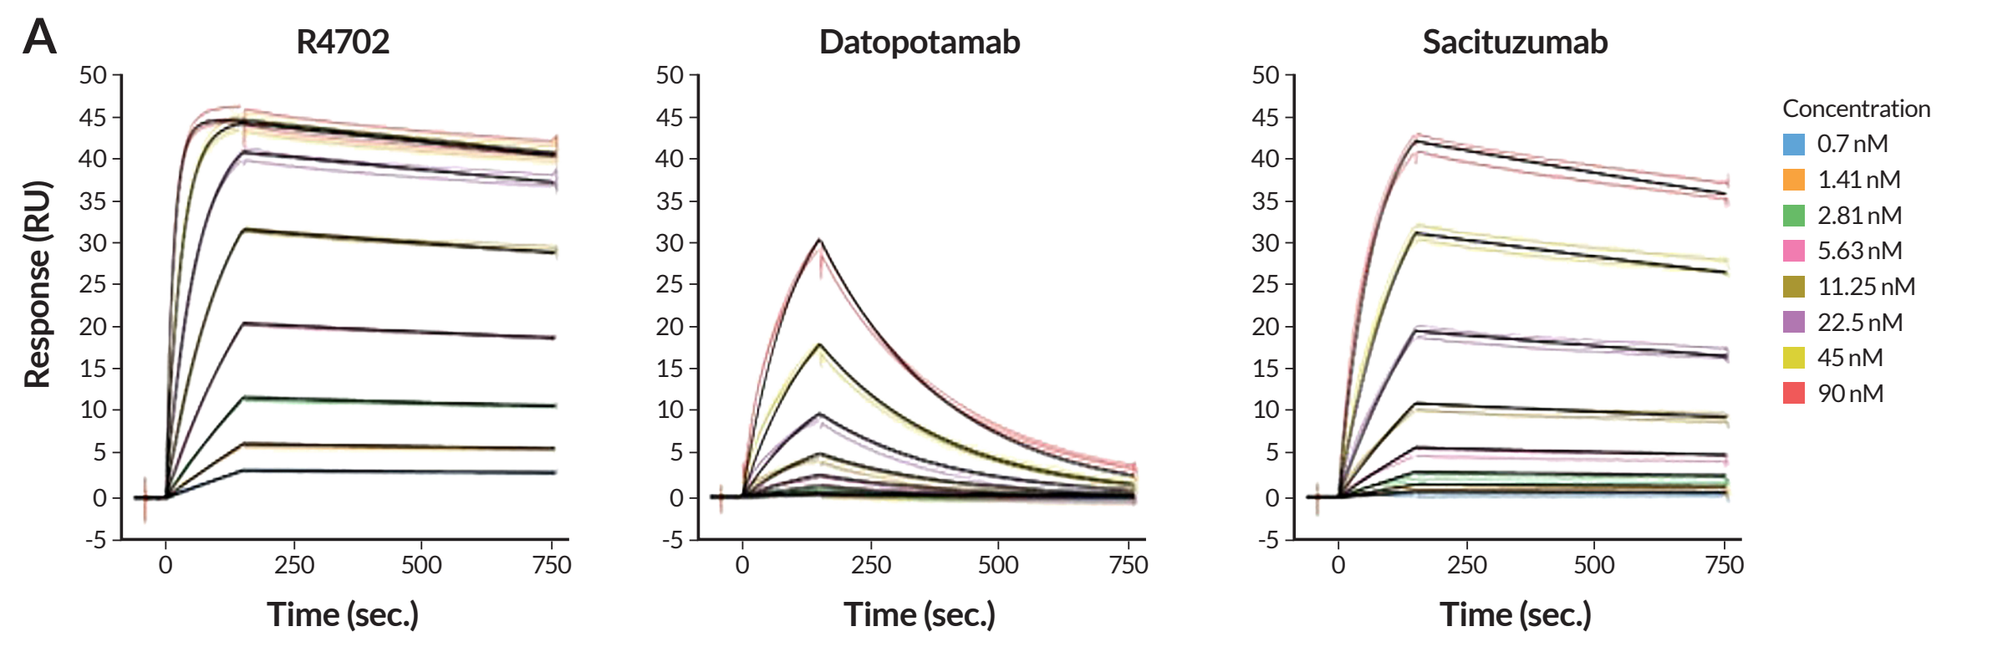
A
R4702
Datopotamab
Sacituzumab
Response (RU)
Time (sec.)
Concentration 0.7 nM 1.41 nM 2.81 nM 5.63 nM 11.25 nM 22.5 nM 45 nM 90 nM

B

Binding affinity of TROP2 antibodies on SPR analysis

TROP2 antibody	KD (M)	Ka (1/Ms)	Kd (1/s)
R4702	2.01E−10	7.45E+05	1.50E−04
Datopotamab	6.43E−08	6.35E+04	4.09E−03
Sacituzumab	1.67E−09	1.58E+05	2.65E−04

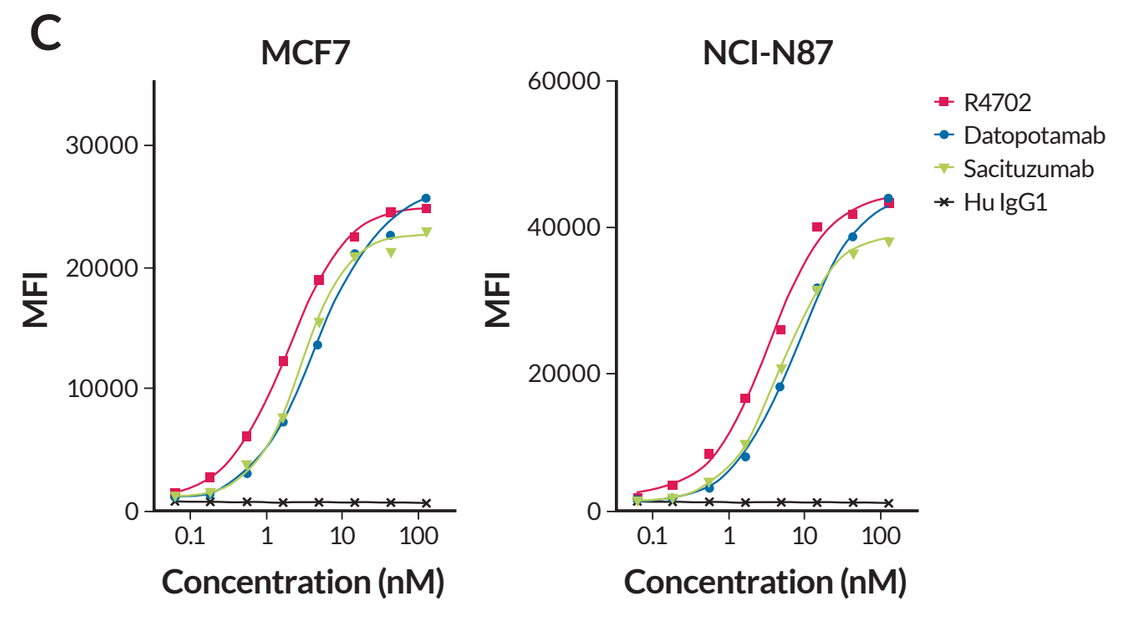
C
MCF7
NCI-N87
MFI
Concentration (nM)
R4702 Datopotamab Sacituzumab Hu IgG1

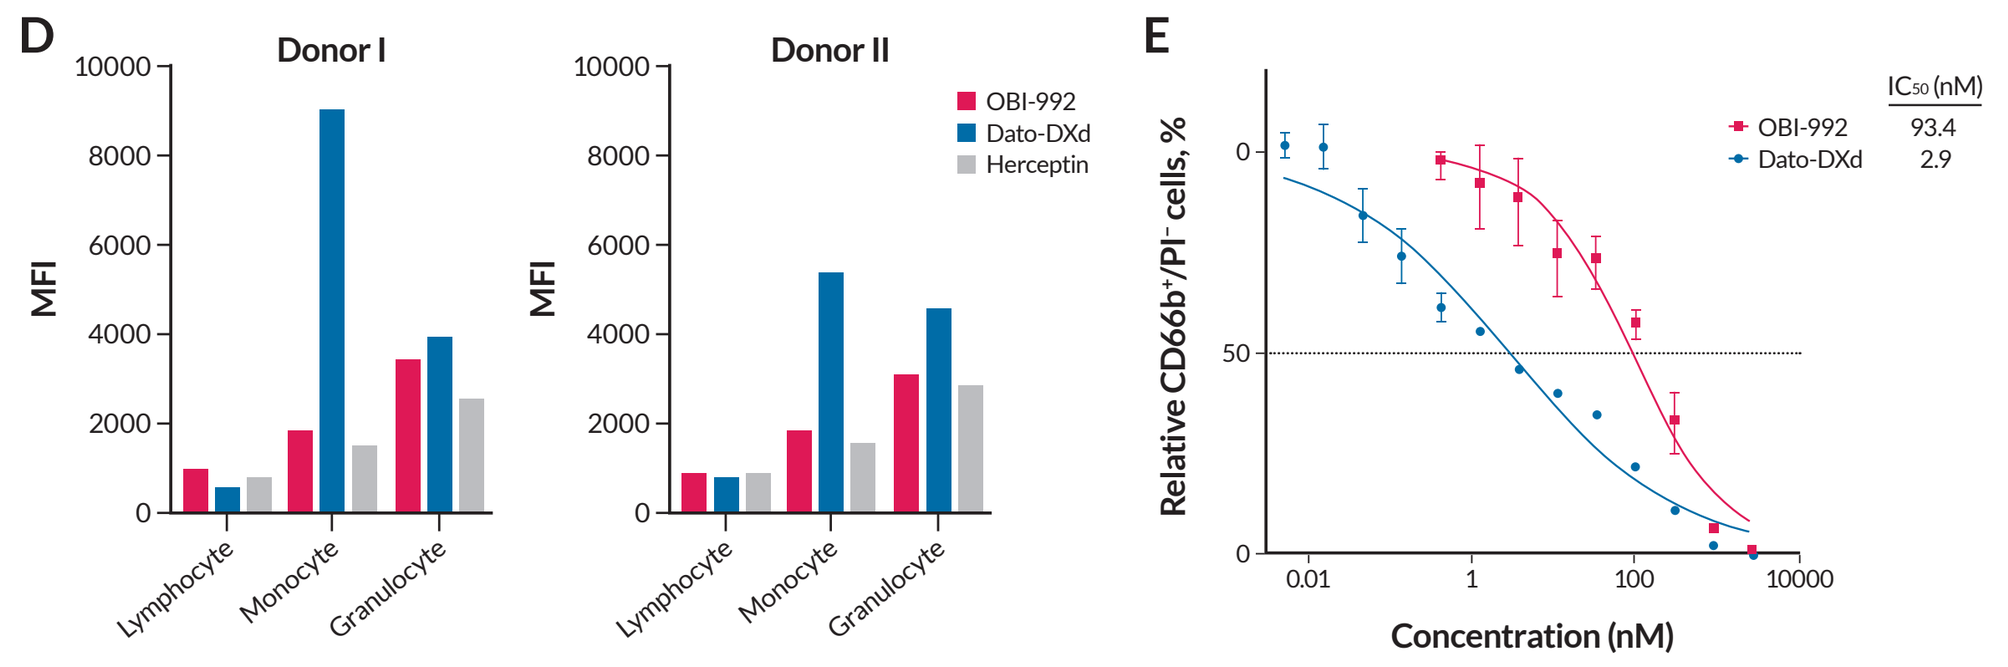
D
Donor I
Donor II
OBI-992 Dato-DXd Herceptin
Lymphocyte Monocyte Granulocyte
E
Relative CD66b+/PI- cells, %
IC50 (nM) OBI-992 93.4 Dato-DXd 2.9
Concentration (nM)

(A) Sensogram plots generated by SPR kinetic analysis demonstrated the association and dissociation characteristics between human TROP2 and TROP2 antibodies (B) Dynamic affinity of TROP2 antibodies to human TROP2 protein. (C) Cell surface binding assay. (D) ADC association with peripheral blood leukocytes. (E) Neutrophil viability with ADC treatment.
Dato-DXd, datopotamab deruxtecan; IC_{50}, half-maximal inhibitory concentration; Ka, association rate constant; Kd, dissociation rate constant; KD, dissociation constant; MFI, mean fluorescence intensity; RU, response units; SPR, surface plasmon resonance; TROP2, trophoblast cell surface antigen 2.

- The OBI-992 payload exatecan induced immunogenic cell death and demonstrated potent cancer-killing effects (**Figure 5**).

Figure 5. Expression of ICD Markers, DNA Damage, and Cytotoxicity Induced by TOP1 Inhibitors

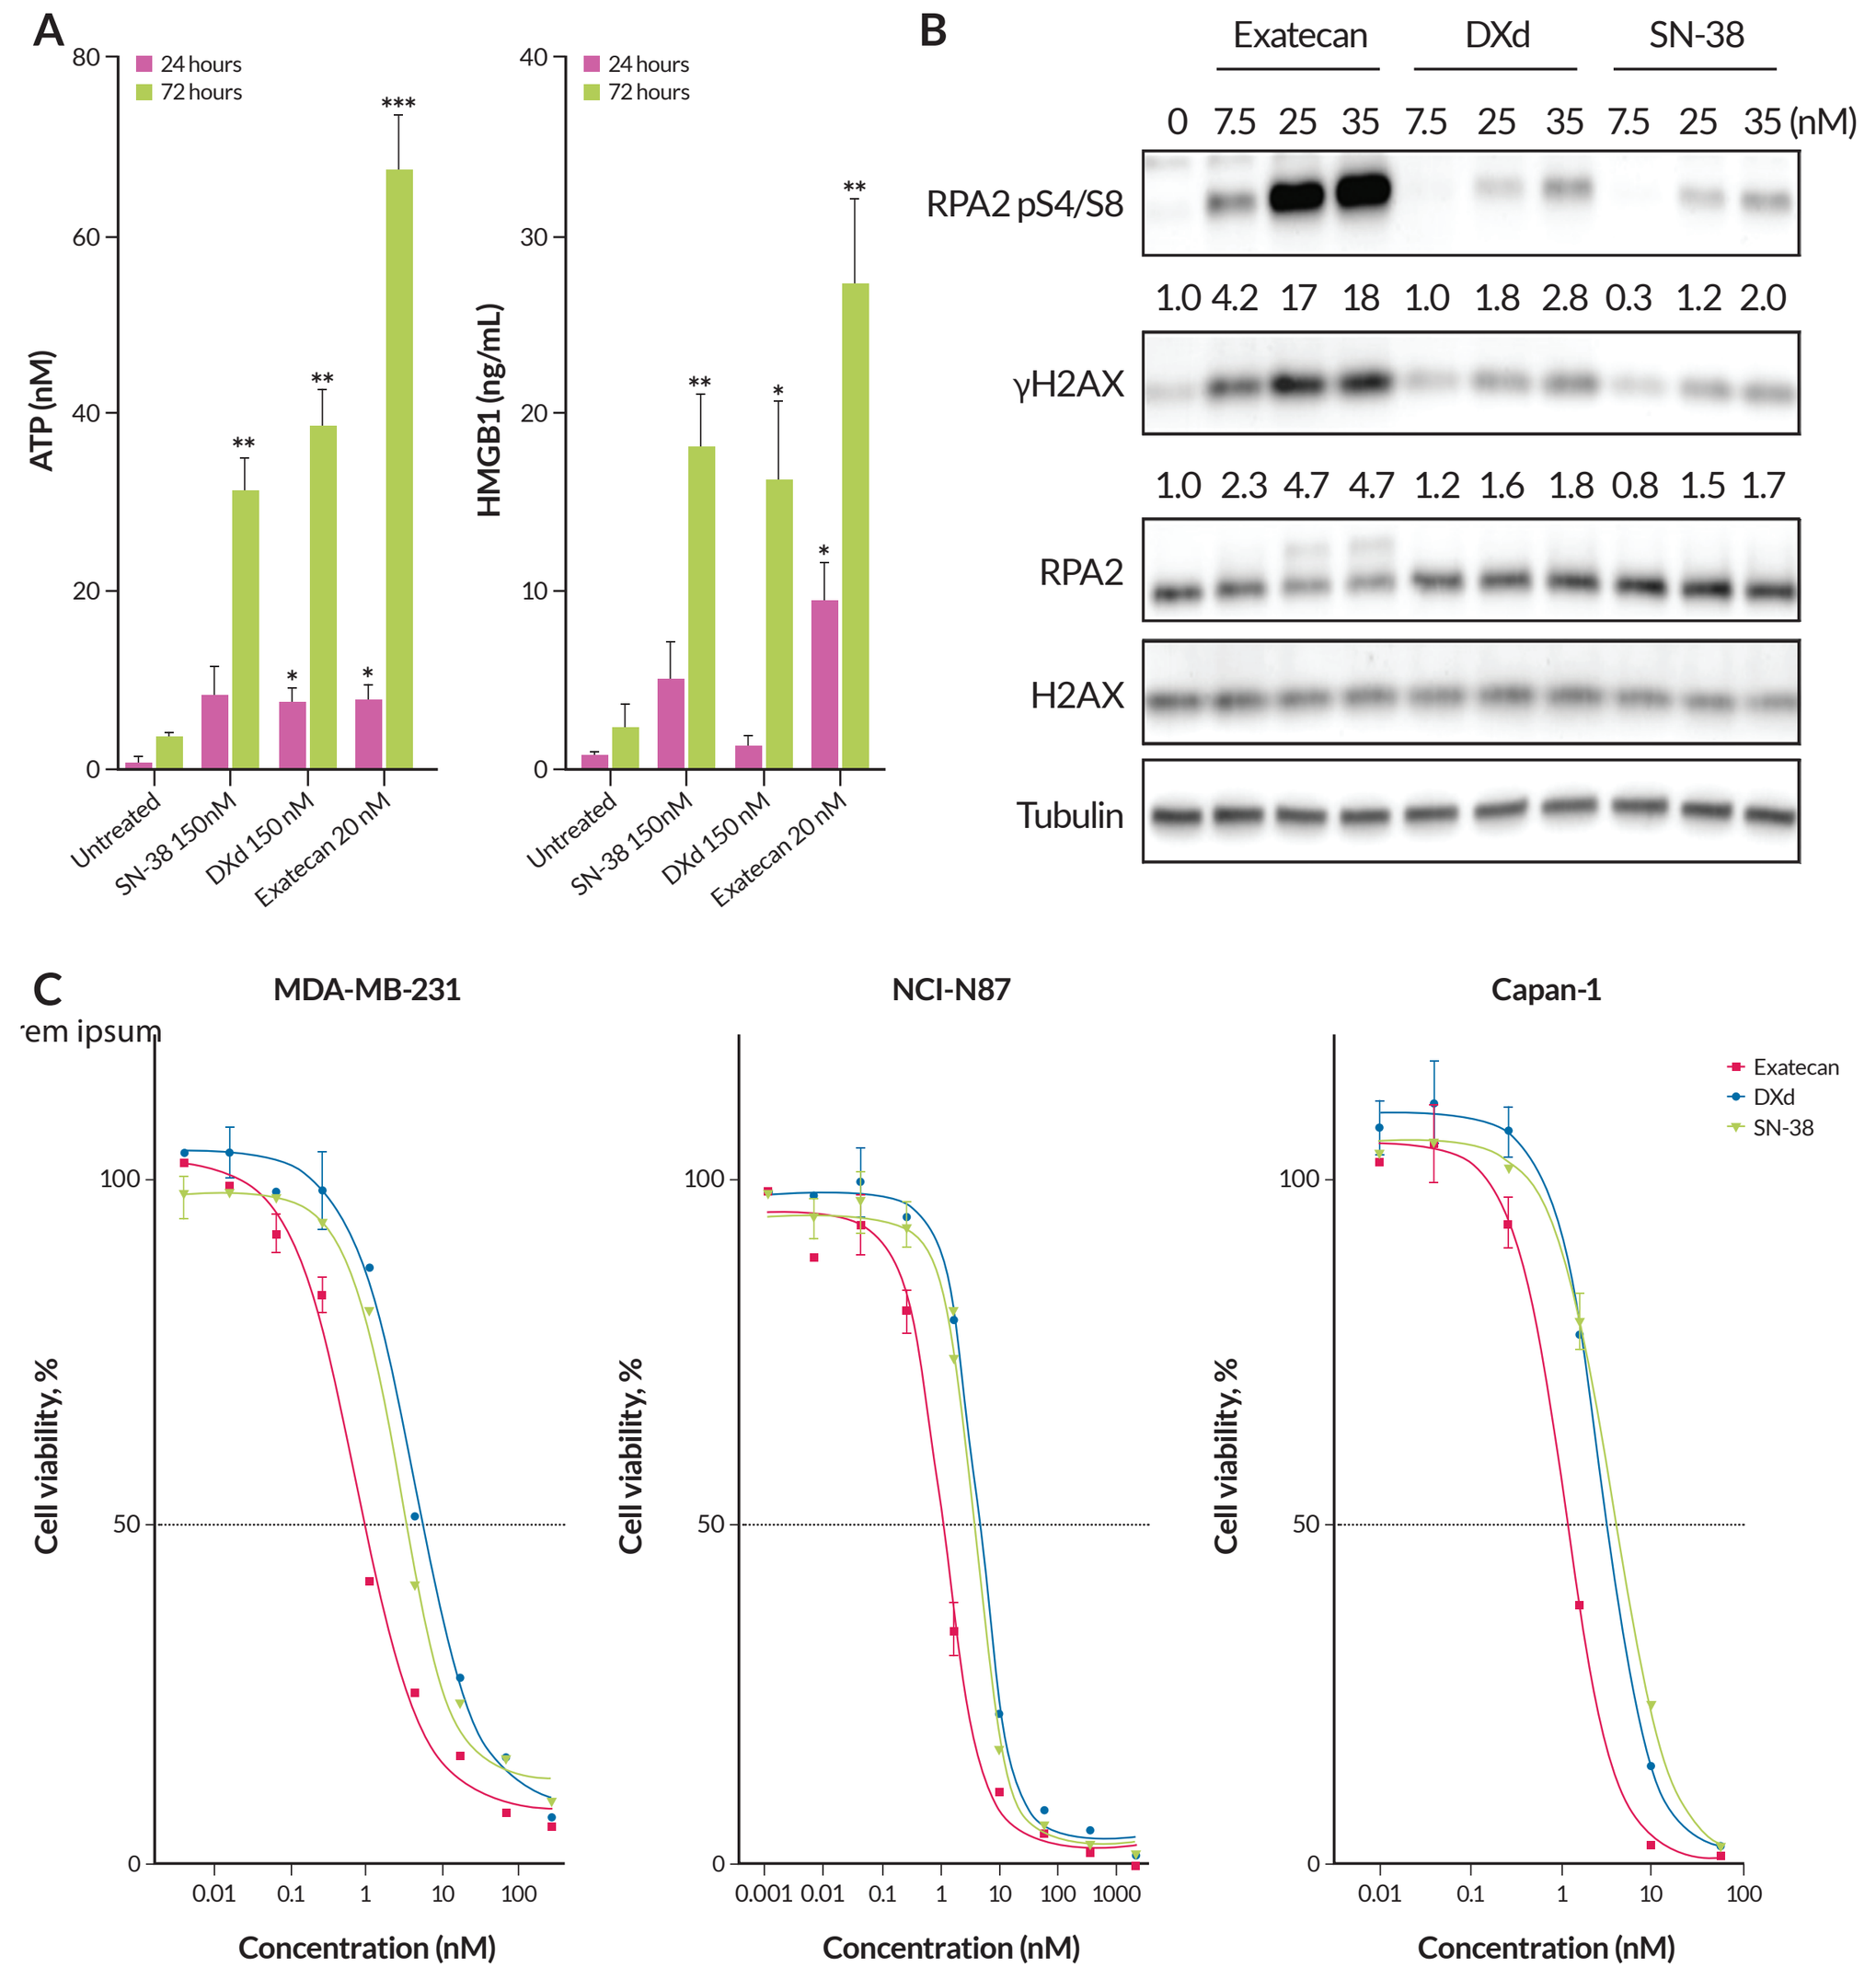
A
24 hours 72 hours
ATP (nM)
HMGB1 (ng/mL)
Untreated SN-38 150nM DXd 150nM Exatecan 20nM
B
Exatecan DXd SN-38
0 7.5 25 35 7.5 25 35 7.5 25 35 (nM)
RPA2 pS4/S8
1.0 4.2 17 18 1.0 1.8 2.8 0.3 1.2 2.0
γH2AX
1.0 2.3 4.7 4.7 1.2 1.6 1.8 0.8 1.5 1.7
RPA2
H2AX
Tubulin
C
MDA-MB-231
NCI-N87
Capan-1
Cell viability, %
Concentration (nM)
Exatecan DXd SN-38

(A) Luciferase assays of ICD markers extracellular ATP (left) and HMGB1 (right) (B) Western blot analysis of DNA damage markers, RPA2 pS4/S8 and γH2AX. The data in this figure result from separate gel electrophoresis runs with consistent loading of the same samples. (C) Cell viability of indicated cells treated with TOPi. Percentages of cell viability were calculated, and results are shown with mean and SD (n=3).
ICD, immunogenic cell death; TOP1, topoisomerase 1; TOPi, topoisomerase inhibitor.
*$P \le .05$; **$P \le .01$; and ***$P \le .001$.

- OBI-992 demonstrated high cytotoxic activity in both 2D and 3D cultures (**Figure 6**).

Figure 6. Positive Correlation Between OBI-992 Cytotoxicity and TROP2 Expression in Cancer Cells

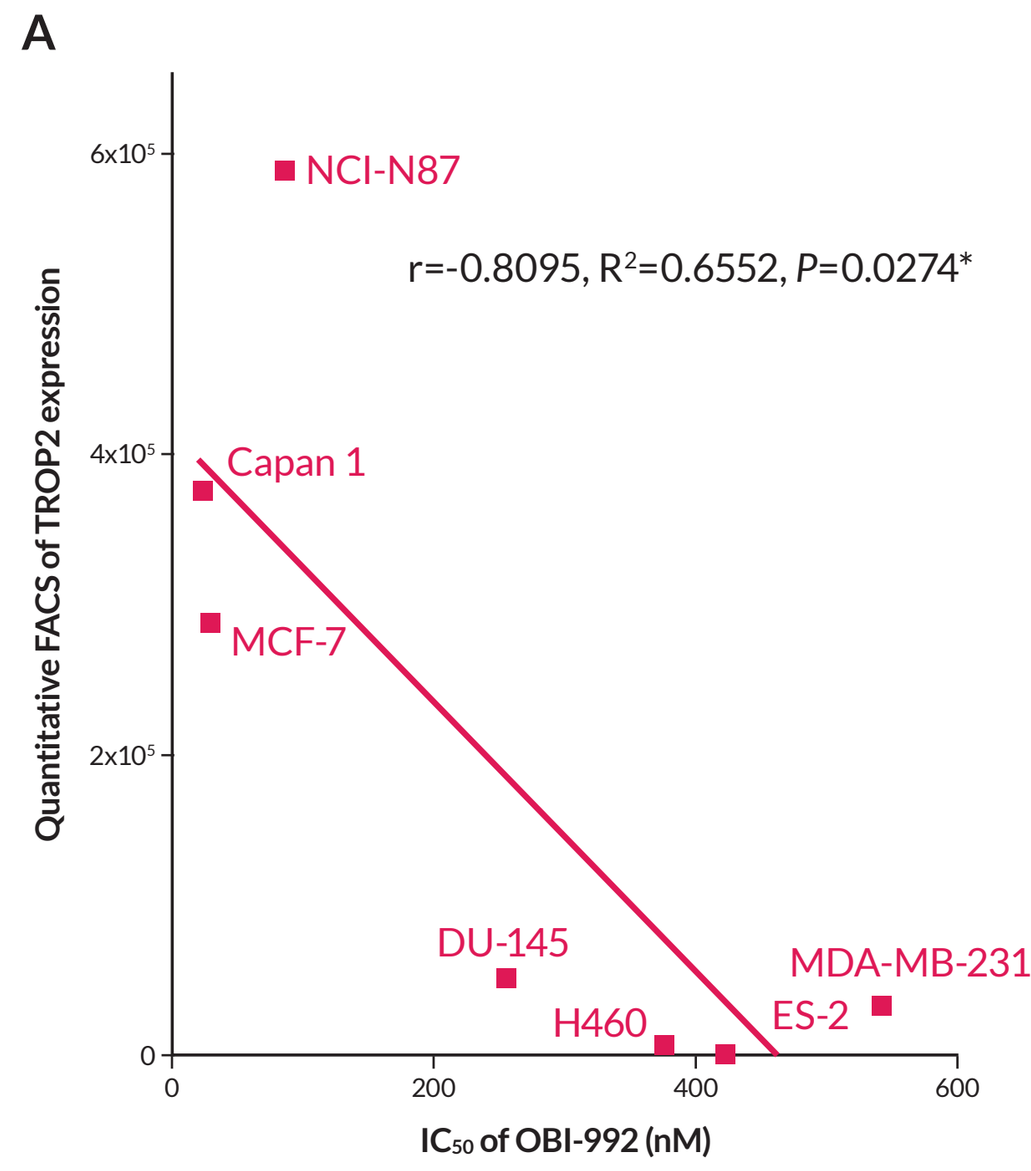
A
r=-0.8095, R2=0.6552, P=0.0274*
Quantitative FACS of TROP2 expression
IC50 of OBI-992 (nM)
NCI-N87 Capan 1 MCF-7 DU-145 H460 ES-2 MDA-MB-231

	Quantitative FACS of TROP2	OBI-992 IC_{50} (nM)
NCI-N87	588946	86.5
Capan1	376355	23.1
MCF-7	288128	29.5
DU-145	52120	255.8
MDA-MB-231	33862	543.6
H460	6743	376.9
ES-2	1976	422.8

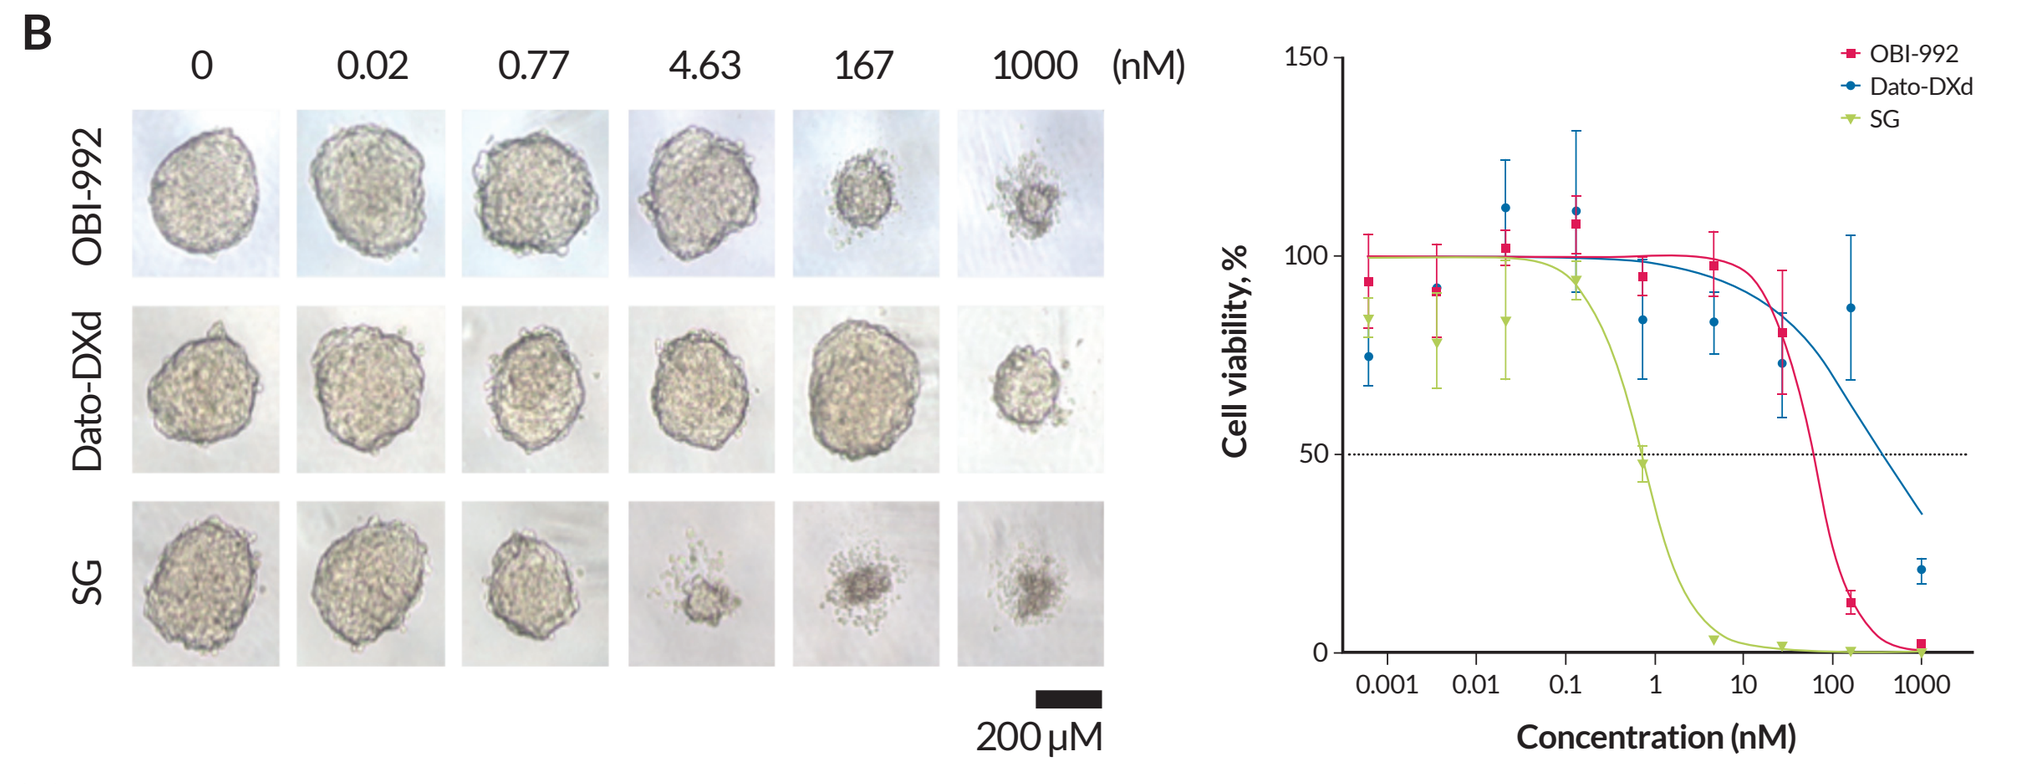
B
0 0.02 0.77 4.63 167 1000 (nM)
OBI-992
Dato-DXd
SG
200 μM
Cell viability, %
Concentration (nM)
OBI-992 Dato-DXd SG

(A) Cytotoxicity of OBI-992 in multiple cancer types. Quantitative FACS values were used to represent TROP2 expression.
(B) Cell viability of prostate cancer DU145 3D spheroid cells treated with the indicated TROP2-ADC for 6 days. Representative images of the spheroid are shown (left). Percentages of cell viability are shown with mean and SEM values (n=3) (right).
3D, three-dimensional; Dato-DXd, datopotamab deruxtecan; FACS, fluorescence-activated cell sorting; IC_{50}, half-maximal inhibitory concentration; R^2, Pearson correlation r value; SG, sacituzumab govitecan, SEM, standard error of mean; TROP2, trophoblast cell surface antigen 2.

- An R364H mutation in TOP1 was identified in OBI-992-resistant cells, with a slight decrease in TROP2 expression and no major increase in BCRP protein expression (**Figure 7**).

Figure 7. Characterization of TROP2-ADC-Resistant Cell Lines

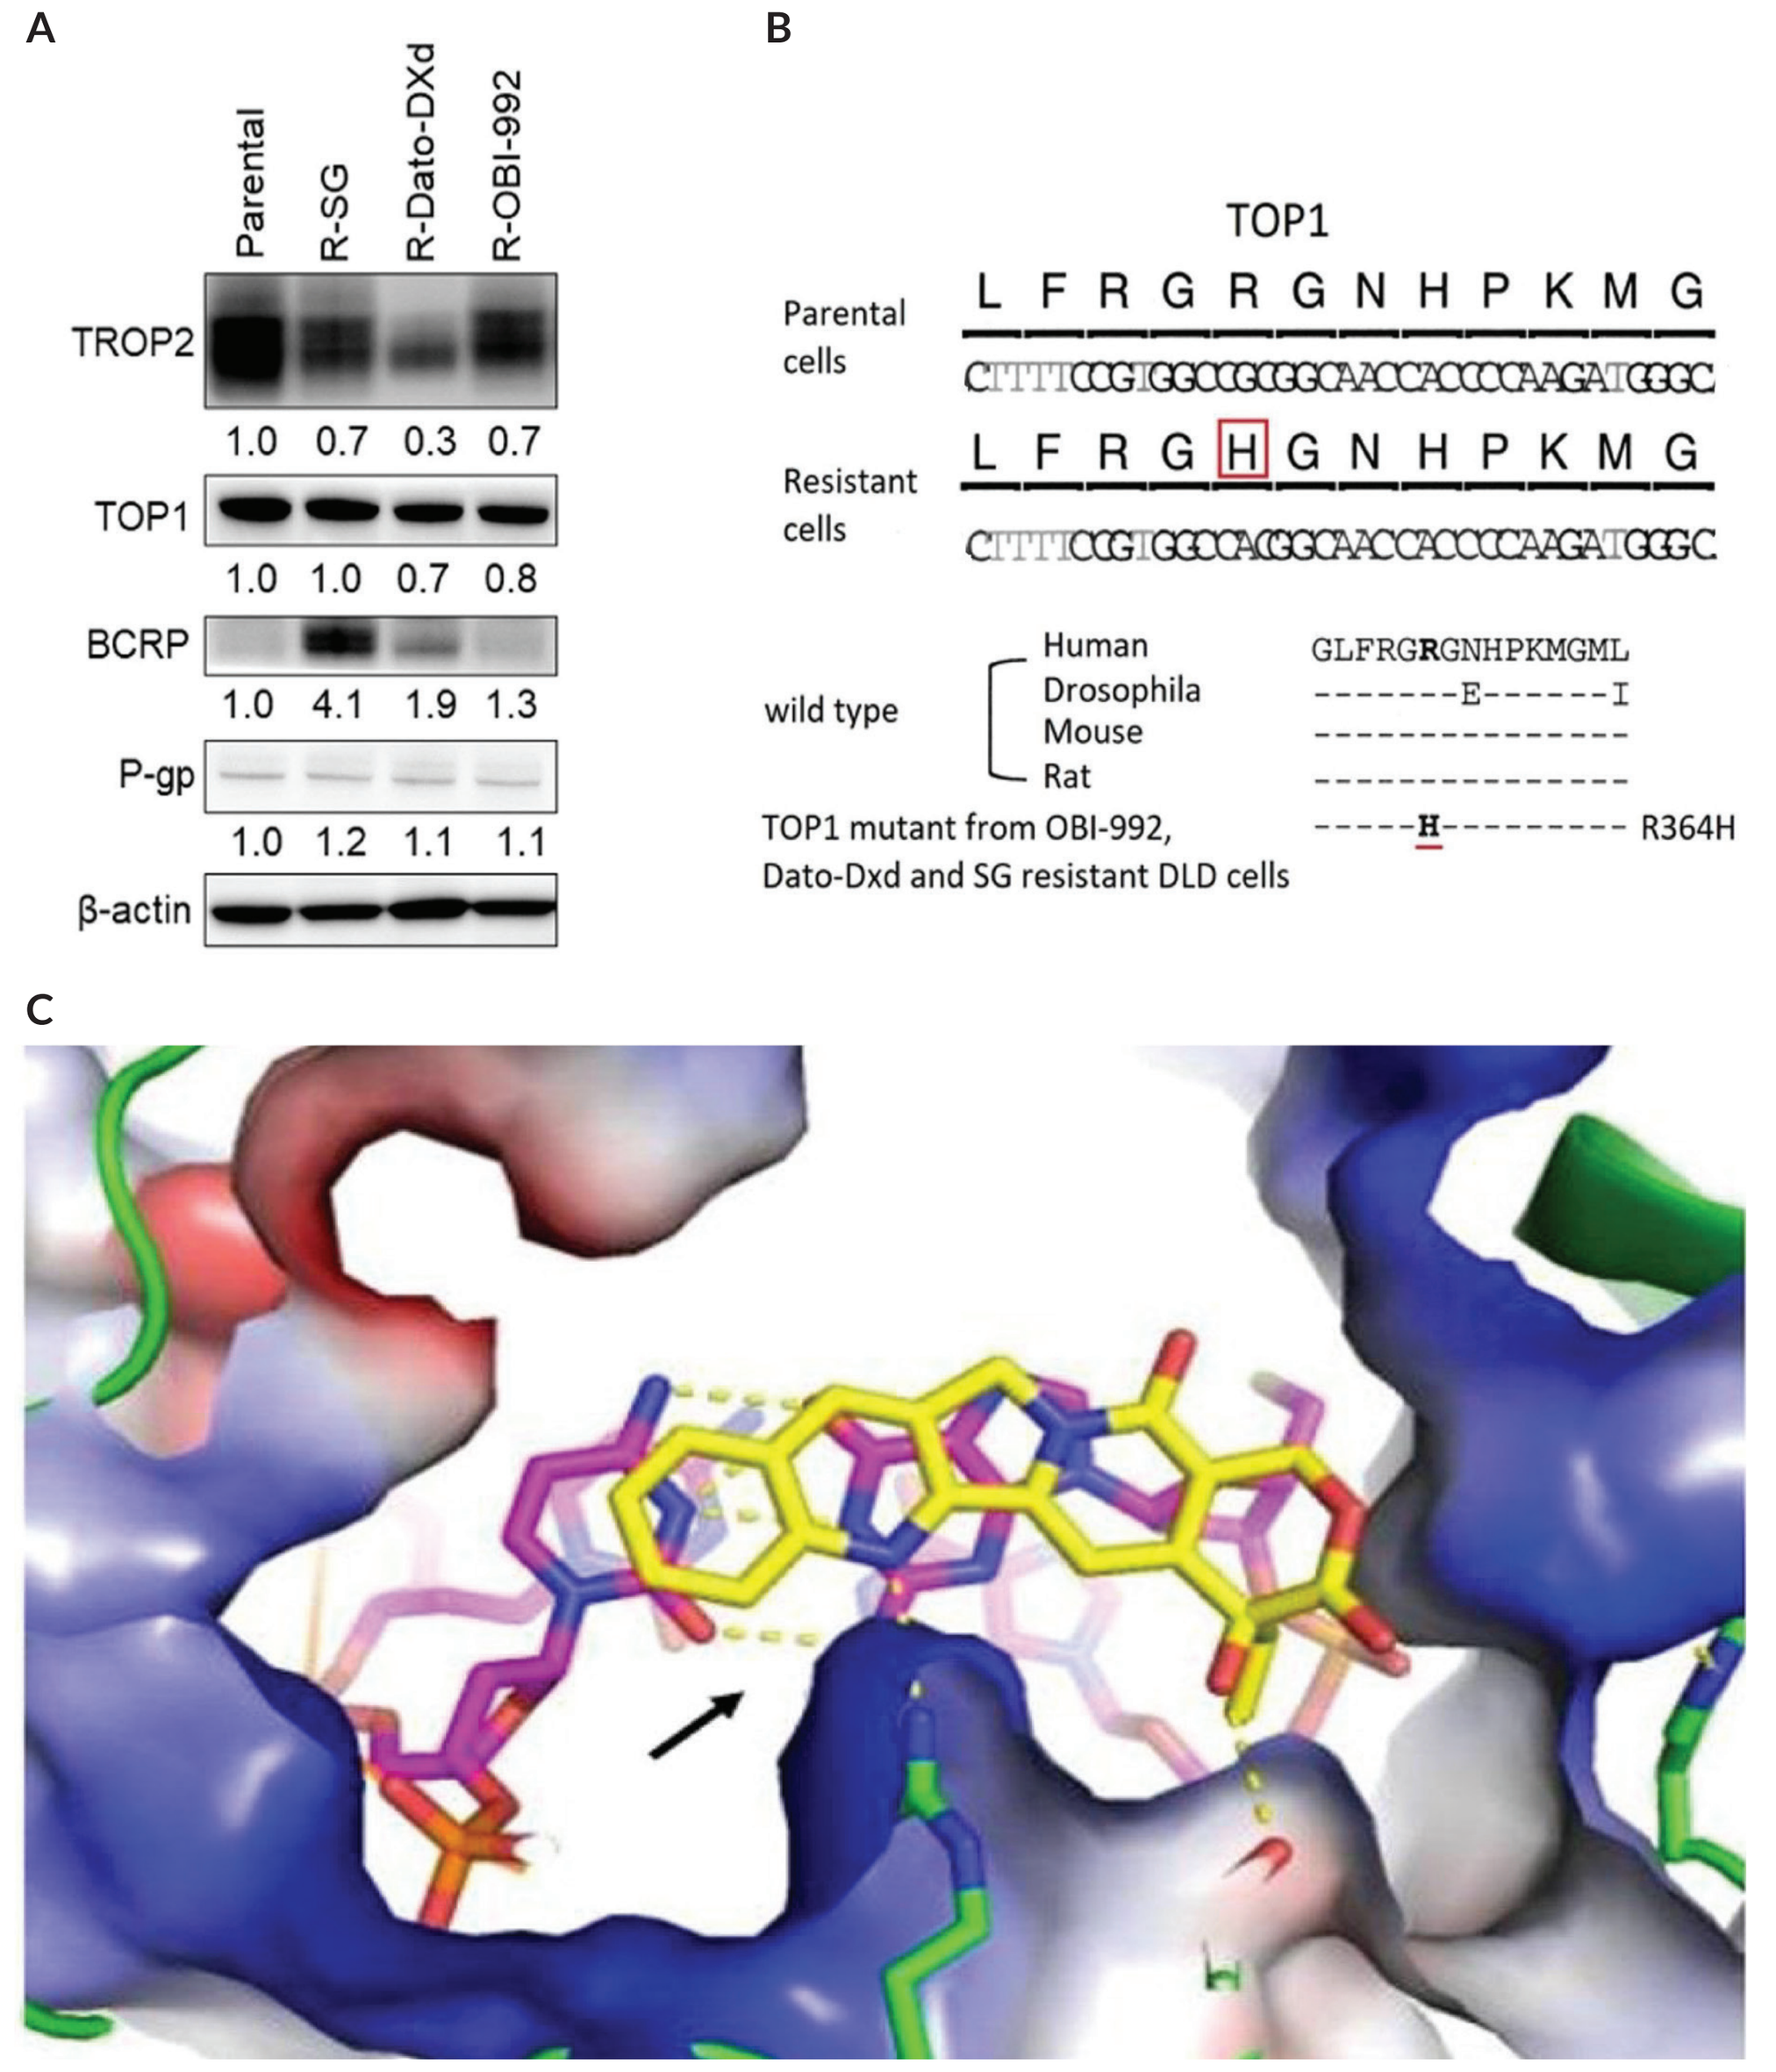
A
Parental R-SG R-Dato-DXd R-OBI-992
TROP2 1.0 0.7 0.3 0.7
TOP1 1.0 1.0 0.7 0.8
BCRP 1.0 4.1 1.9 1.3
P-gp 1.0 1.2 1.1 1.1
β-actin
B
TOP1
Parental cells L F R G R G N H P K M G
Resistant cells L F R G H G N H P K M G
wild type Human Drosophila Mouse Rat
GLFRGRGNHPKMGML
TOP1 mutant from OBI-992, Dato-Dxd and SG resistant DLD cells
R364H
C

(A) Western blot analysis of indicated protein in ADC-resistant DLD1 cells. (B) TOP1 mutation in the ADC-resistant DLD1 cells. (C) Crystal structure of TOP1 inhibitor (TOPi) binding site of TOP1 (PDB=1T8I). TOP1 is shown as ribbons and surfaces. DNA and TOPi are shown in magenta and yellow, respectively. Arrow indicates the R364 site.
ADC, antibody-drug conjugate; BCRP, breast cancer resistance protein; R-SG, sacituzumab govitecan resistant cells; R-Dato-DXd, datopotamab deruxtecan resistant cells; R-OBI-992, OBI-992 resistant cells; TOP1, topoisomerase 1; TOPi, topoisomerase inhibitor; TROP2, trophoblast cell surface antigen 2.

- OBI-992 demonstrated potential synergistic cytotoxicity when combined with PARP inhibitors (**Figure 8**).

Figure 8. Synergistic Effects of OBI-992 in Combination With PARP Inhibitors

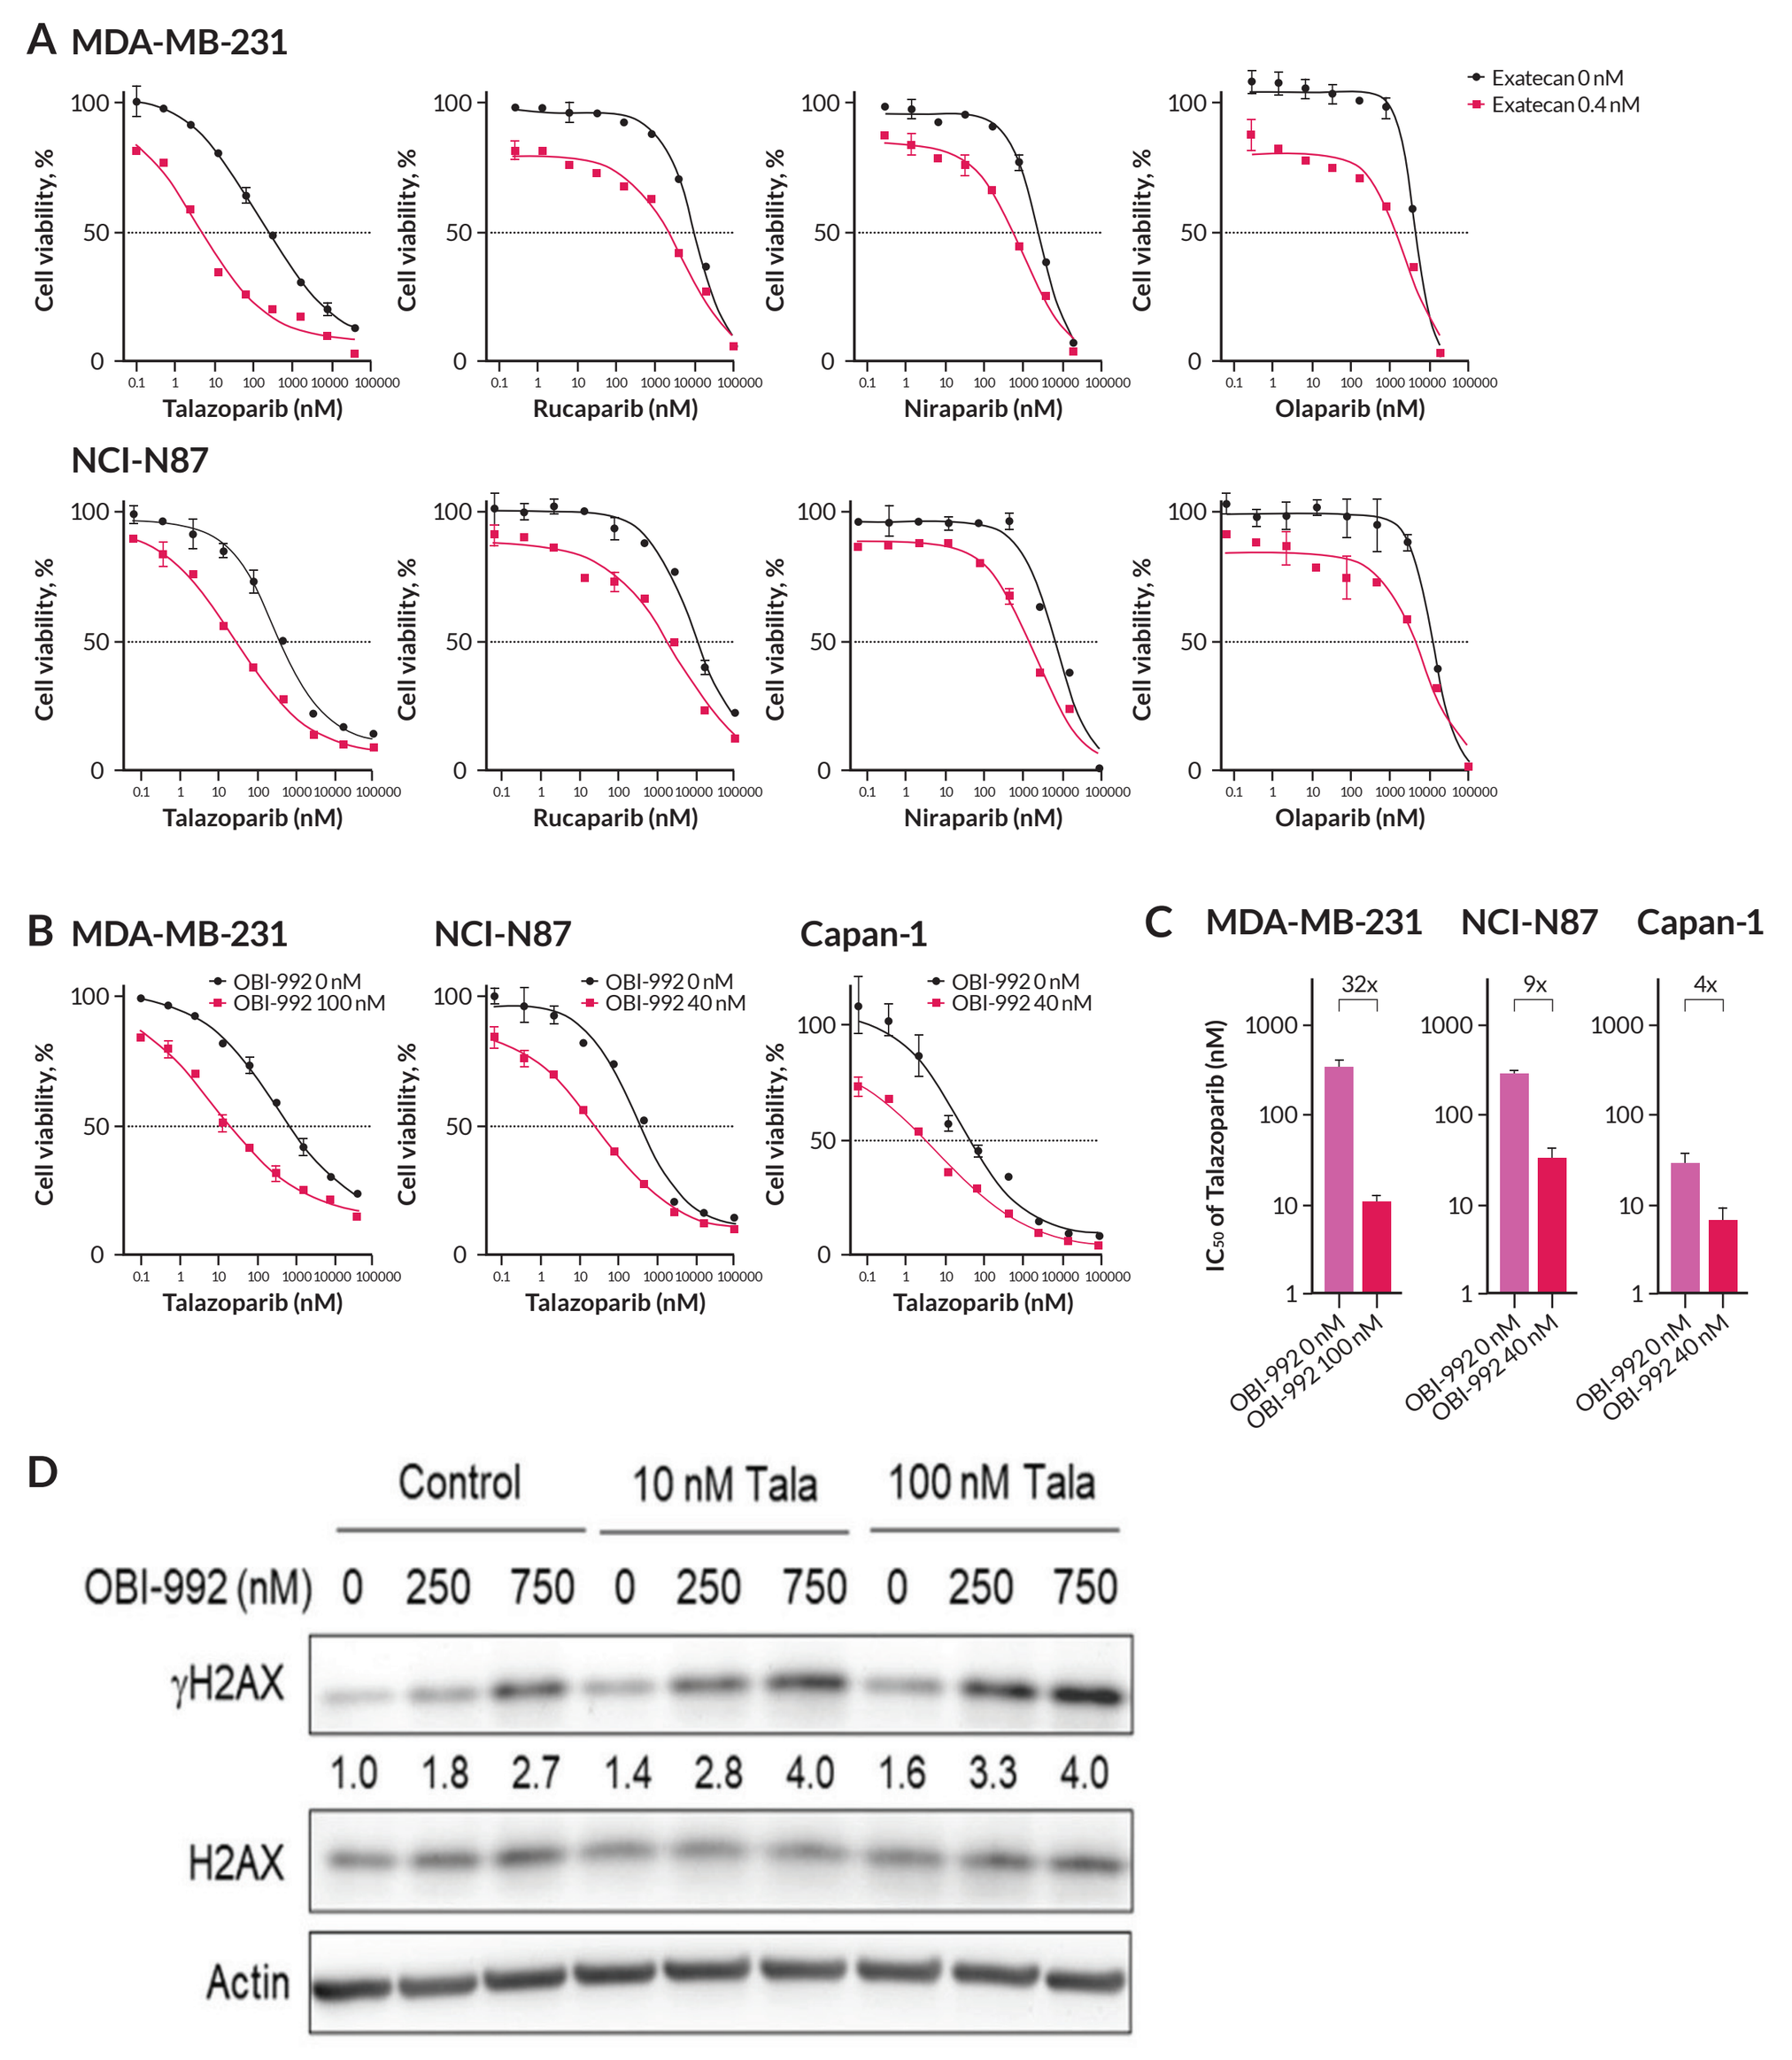
A MDA-MB-231
NCI-N87
Exatecan 0 nM Exatecan 0.4 nM
Talazoparib (nM) Rucaparib (nM) Niraparib (nM) Olaparib (nM)
Cell viability, %
B MDA-MB-231 NCI-N87 Capan-1
OBI-992 0 nM OBI-992 100 nM OBI-992 40 nM
C MDA-MB-231 NCI-N87 Capan-1
32x 9x 4x
IC50 of Talazoparib (nM)
D
Control 10 nM Tala 100 nM Tala
OBI-992 (nM) 0 250 750 0 250 750 0 250 750
γH2AX
1.0 1.8 2.7 1.4 2.8 4.0 1.6 3.3 4.0
H2AX
Actin

(A) Cell viability of MDA-MB-231 and NCI-N87 cells treated with PARP inhibitors and exatecan at exatecan concentrations of 0 or 0.4 nM for 6 days. (B) Cell viability of indicated cells treated with PARP inhibitor and OBI-992 at OBI-992 concentrations of 40 nM or 100 nM for 6 days. Percentages of cell viability were calculated, and results were shown with mean and SD (n=3) The IC_{50} values of each group were calculated by Prism. (C) Fold change of IC_{50}. (D) Western blot analysis of DNA damage marker γH2AX in MDA-MB-231 cells treated with talazoparib and OBI-992 for 24 hours.
IC_{50}, half-maximal inhibitory concentration; PARP, poly (ADP-ribose) polymerases; Tala, talazoparib.

CONCLUSIONS

- OBI-992 is a novel TROP2-targeting ADC with a TOP1 inhibitor exatecan payload.
- High expression of both TROP2 and TOP1 is associated with poor survival in patients with colon cancer.
- R4702 is a novel TROP2 antibody with a unique binding epitope, high binding affinity, and low non-specific binding.
- Exatecan and the exatecan-conjugated ADC OBI-992 showed high cytotoxic activities.
- A slight decrease in TROP2 protein expression and no major change in BCRP expression was observed in OBI-992-resistant cells.
- Combining OBI-992 with PARP inhibitors has the potential for synergistic effects in TROP2-expressing malignancies.

References

1. Khongorzul P, et al. *Mol Cancer Res. 2020;18(1):3-19.*
2. Marei HE, et al. *Cancer Cell Int*. 2022;22(1):255.
3. Ripani E, et al. *Int J Cancer*. 1998;76(5):671-676.
4. Syed YY, et al. *Drugs*. 2020;80(10):1019-1025.
5. Schmitt S, et al. *Mol Cancer Ther*. 2024;23:1-13.
6. Kaplan-Meier Plotter. KMplot.com. Accessed March 21, 2024. https://kmplot.com/analysis/

Disclosures

This study was funded by OBI Pharma, Inc. All authors are employees of OBI Pharma, Inc.

Acknowledgments

The TROP2-targeting antibody was in-licensed from Biosion, Inc. OBI Pharma owns ex-China commercial rights for OBI-992. Graphics support was provided by Prescott Medical Communications Group, a Citrus Health Group, Inc. company (Chicago, IL).

Presented at the American Association for Cancer Research Annual Meeting; April 5-10, 2024; San Diego, CA