AACR 2021
955

Preclinical Characterization of a Novel SSEA-4-Targeting Antibody-Drug Conjugate, OBI-998

I-Ju Chen[1], Chun-Chung Wang[1], Chi-Sheng Shia[1], Chung-Chen Su[1], Chi-Huan Lu[1], Hui-Wen Chang[1], Ping-Tzu Chiu[1], Yueh-Chin Wu[1], Ming-Tain Lai[1], Wei-Chien Tang[1], Hsin-Yi Tung[1], and Ren-Yu Hsu[1]

[1]OBI Pharma, Inc., Taipei, Taiwan

BACKGROUND

Stage-specific embryonic antigen-4 (SSEA-4):

- A Globo-series hexasaccharide
- Overexpressed in multiple cancers, including teratocarcinoma, glioblastoma multiforme, renal cell carcinoma, basaloid lung cancer, epithelial ovarian carcinoma, breast cancer, and oral cancer[1-6]
- A potential cancer stem cell marker[7,8]

Biosynthesis of Globo-series Glycosphingolipids

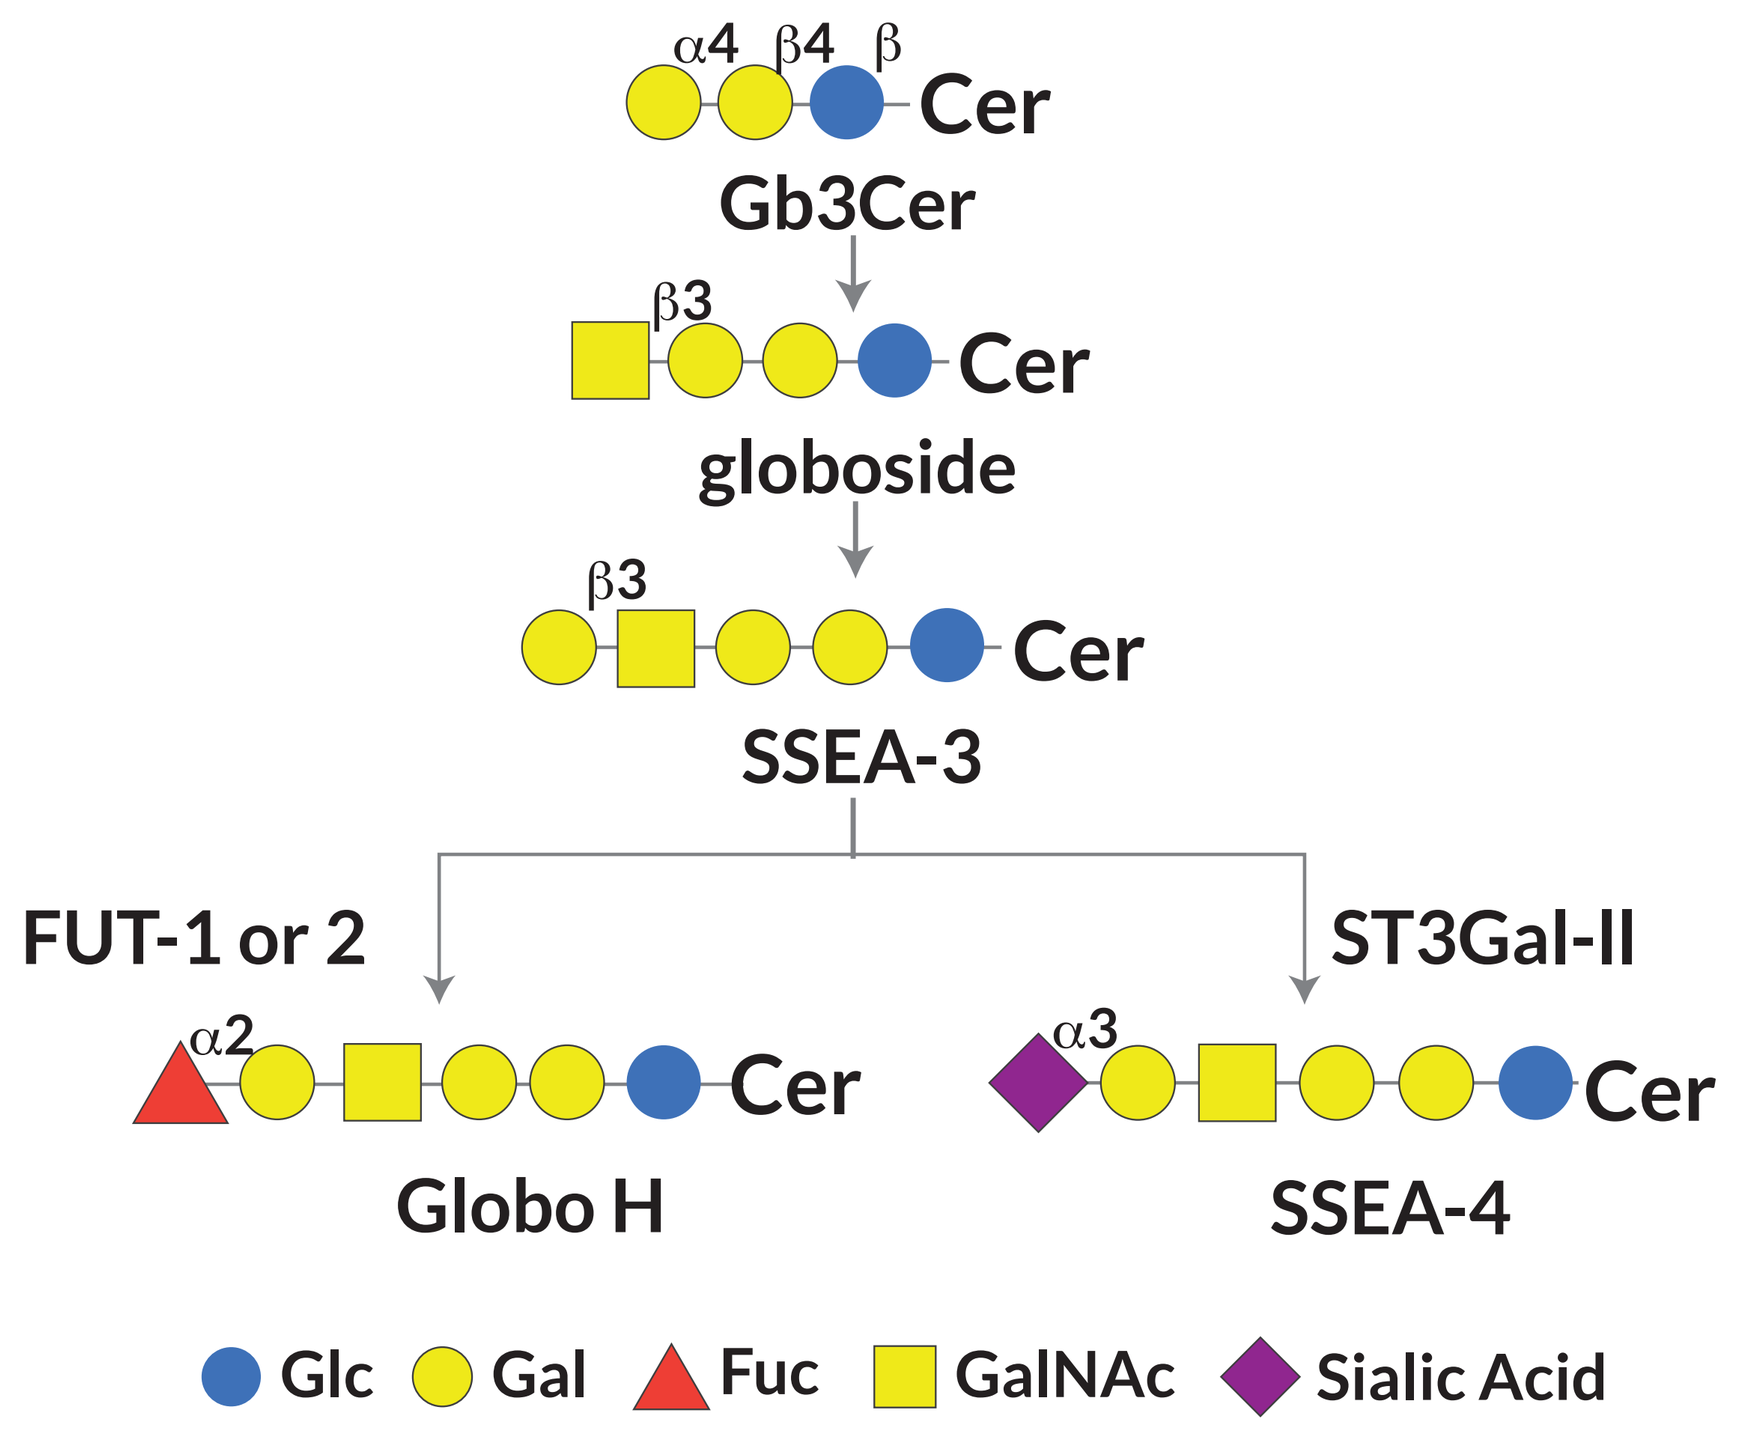
α4 β4 β
Cer
Gb3Cer
β3
Cer
globoside
β3
Cer
SSEA-3
FUT-1 or 2
ST3Gal-II
α2
Cer
Globo H
α3
Cer
SSEA-4
Glc
Gal
Fuc
GalNAc
Sialic Acid

OBI-998 is an antibody drug conjugate (ADC) comprising the humanized anti-SSEA-4 antibody (OBI-898) that is conjugated to the highly potent microtubule-disrupting agent monomethyl auristatin E (MMAE) through maleimide and PEGylated cleavable linkers

OBI-998 Structure

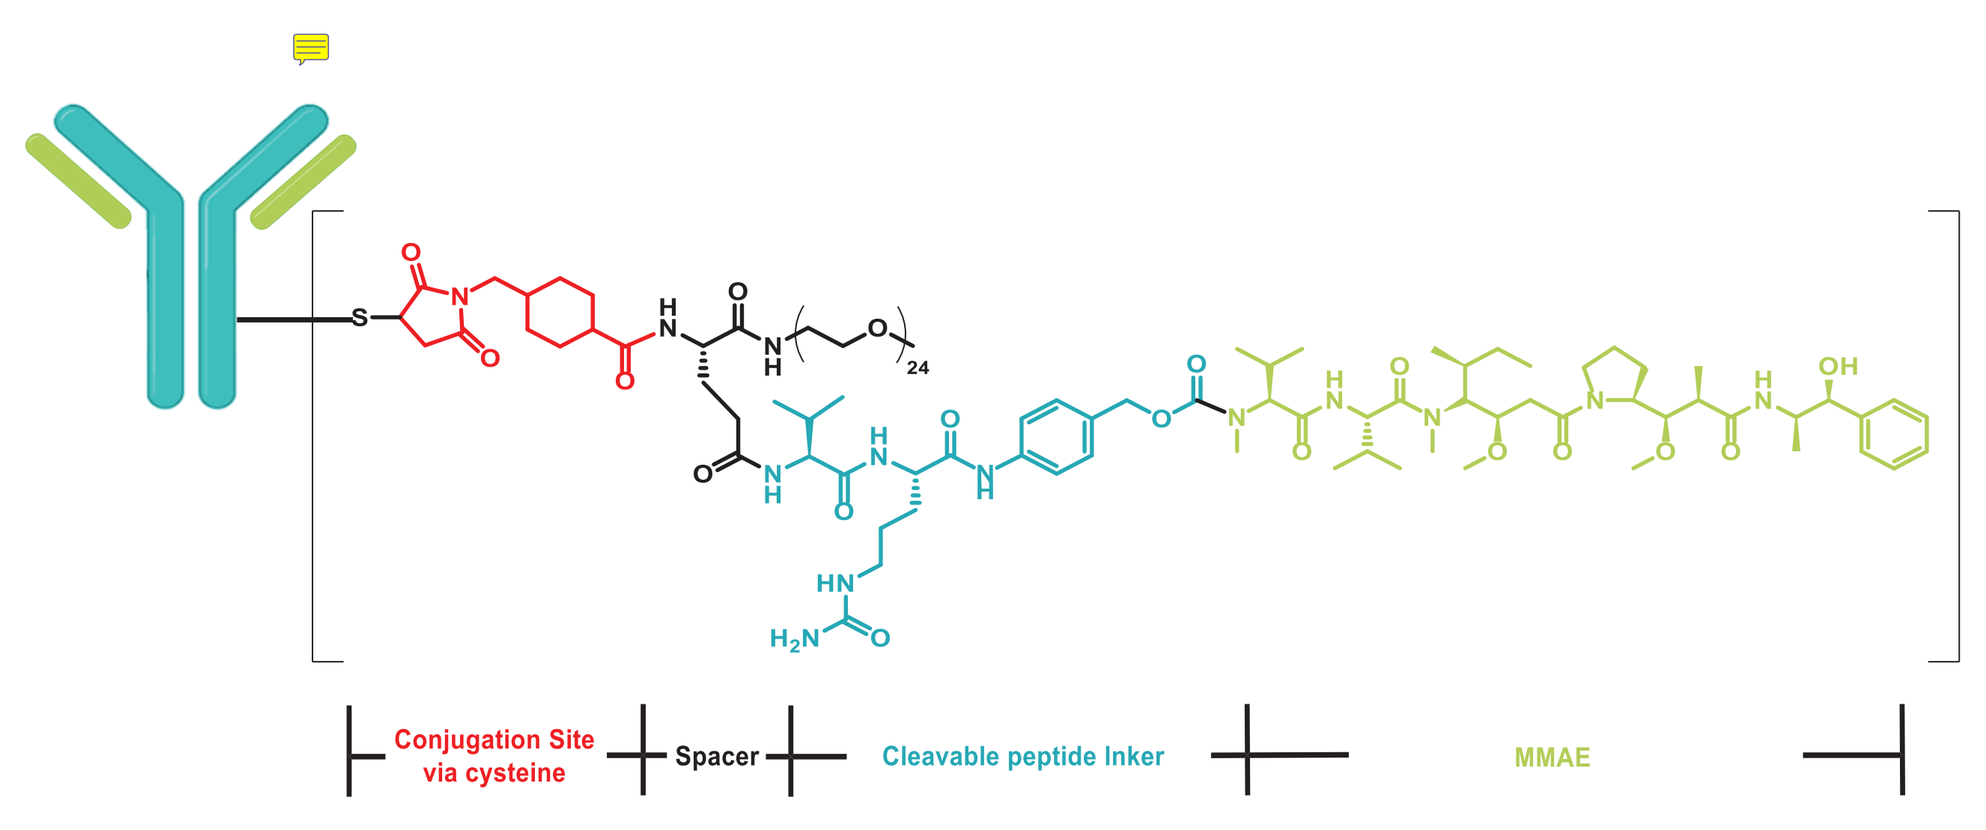
~4
Conjugation Site via cysteine
Spacer
Cleavable peptide linker
MMAE

OBJECTIVE

We investigated a novel antibody-drug conjugate (ADC) targeting SSEA-4 to evaluate its potential as a therapeutic agent for cancer treatment.

RESULTS

Binding specificity

Figure 1. Binding specificity of antibody part of OBI-998.

(A) SSEA-4-specific binding, not Globo H, nor SSEA-3

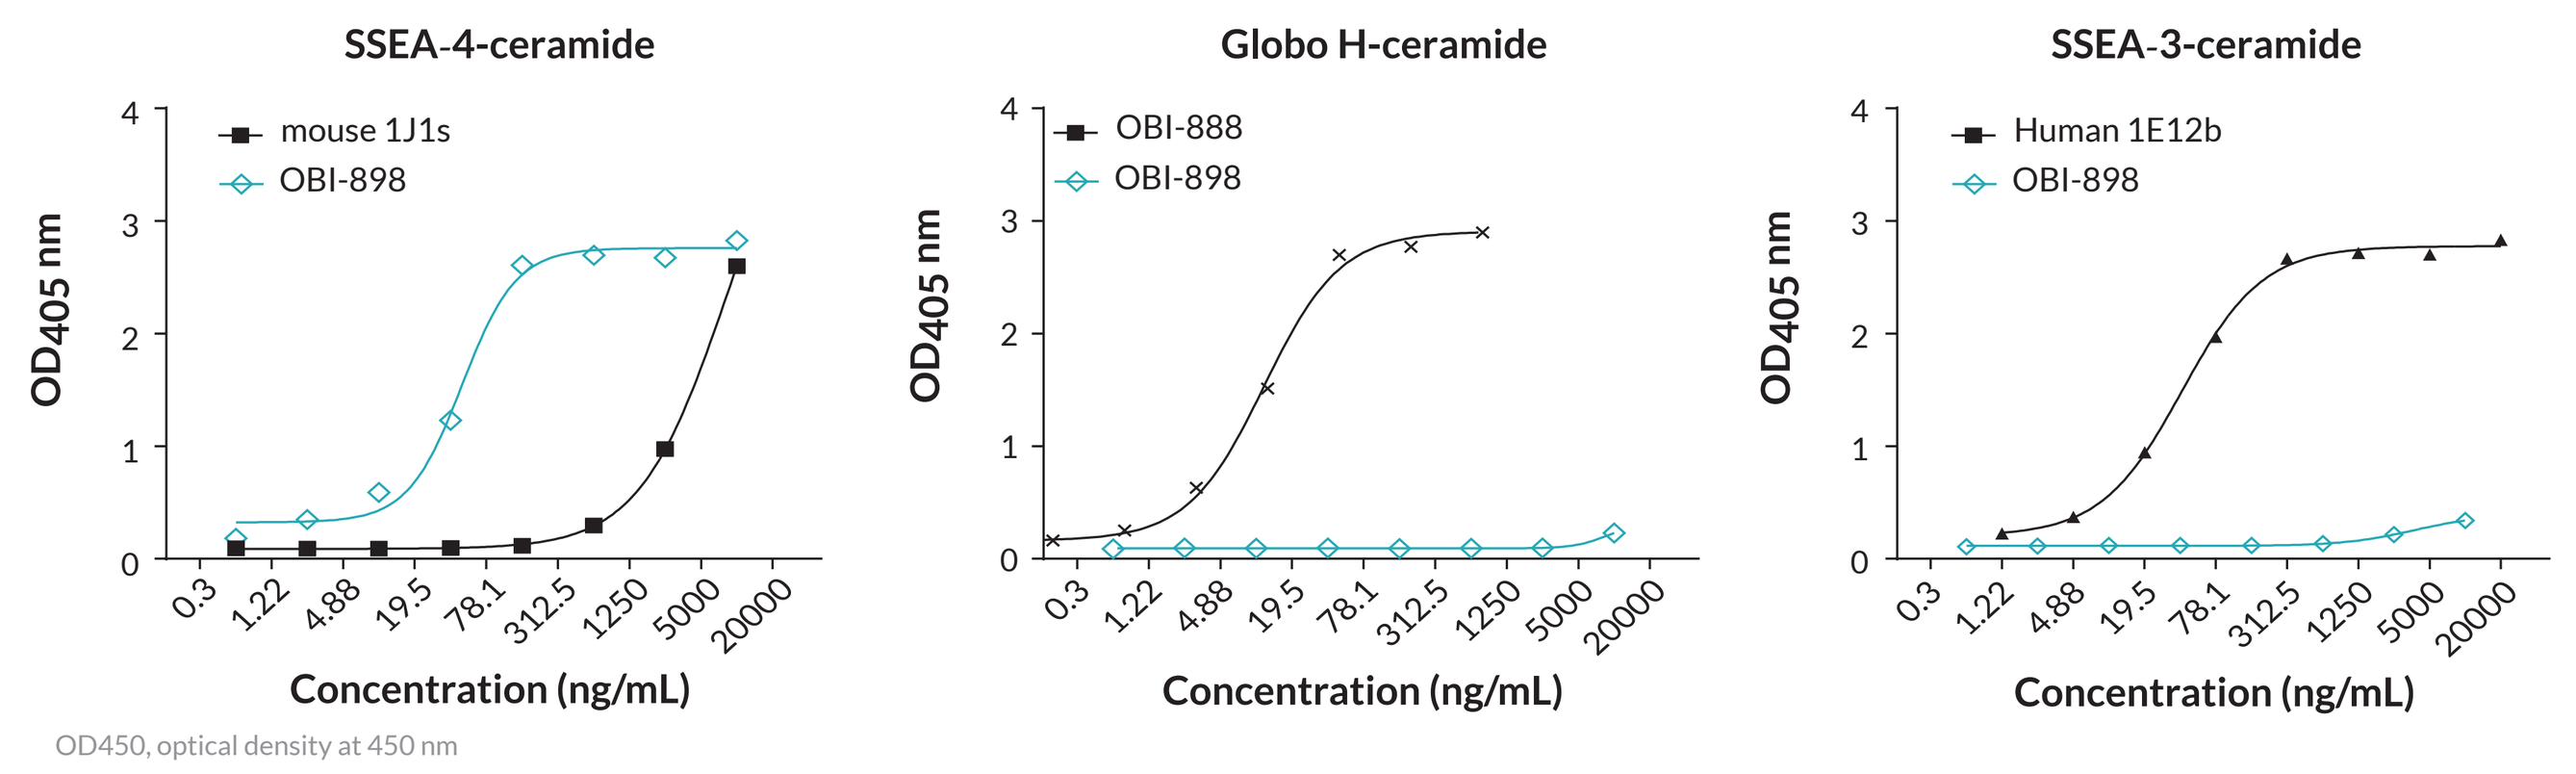
SSEA-4-ceramide
Globo H-ceramide
SSEA-3-ceramide
mouse 1J1s
OBI-898
OBI-888
Human 1E12b
OD405 nm
Concentration (ng/mL)
0.3
1.22
4.88
19.5
78.1
312.5
1250
5000
20000
OD450, optical density at 450 nm

(B) Binding specificity determination by an array of 21 glycans

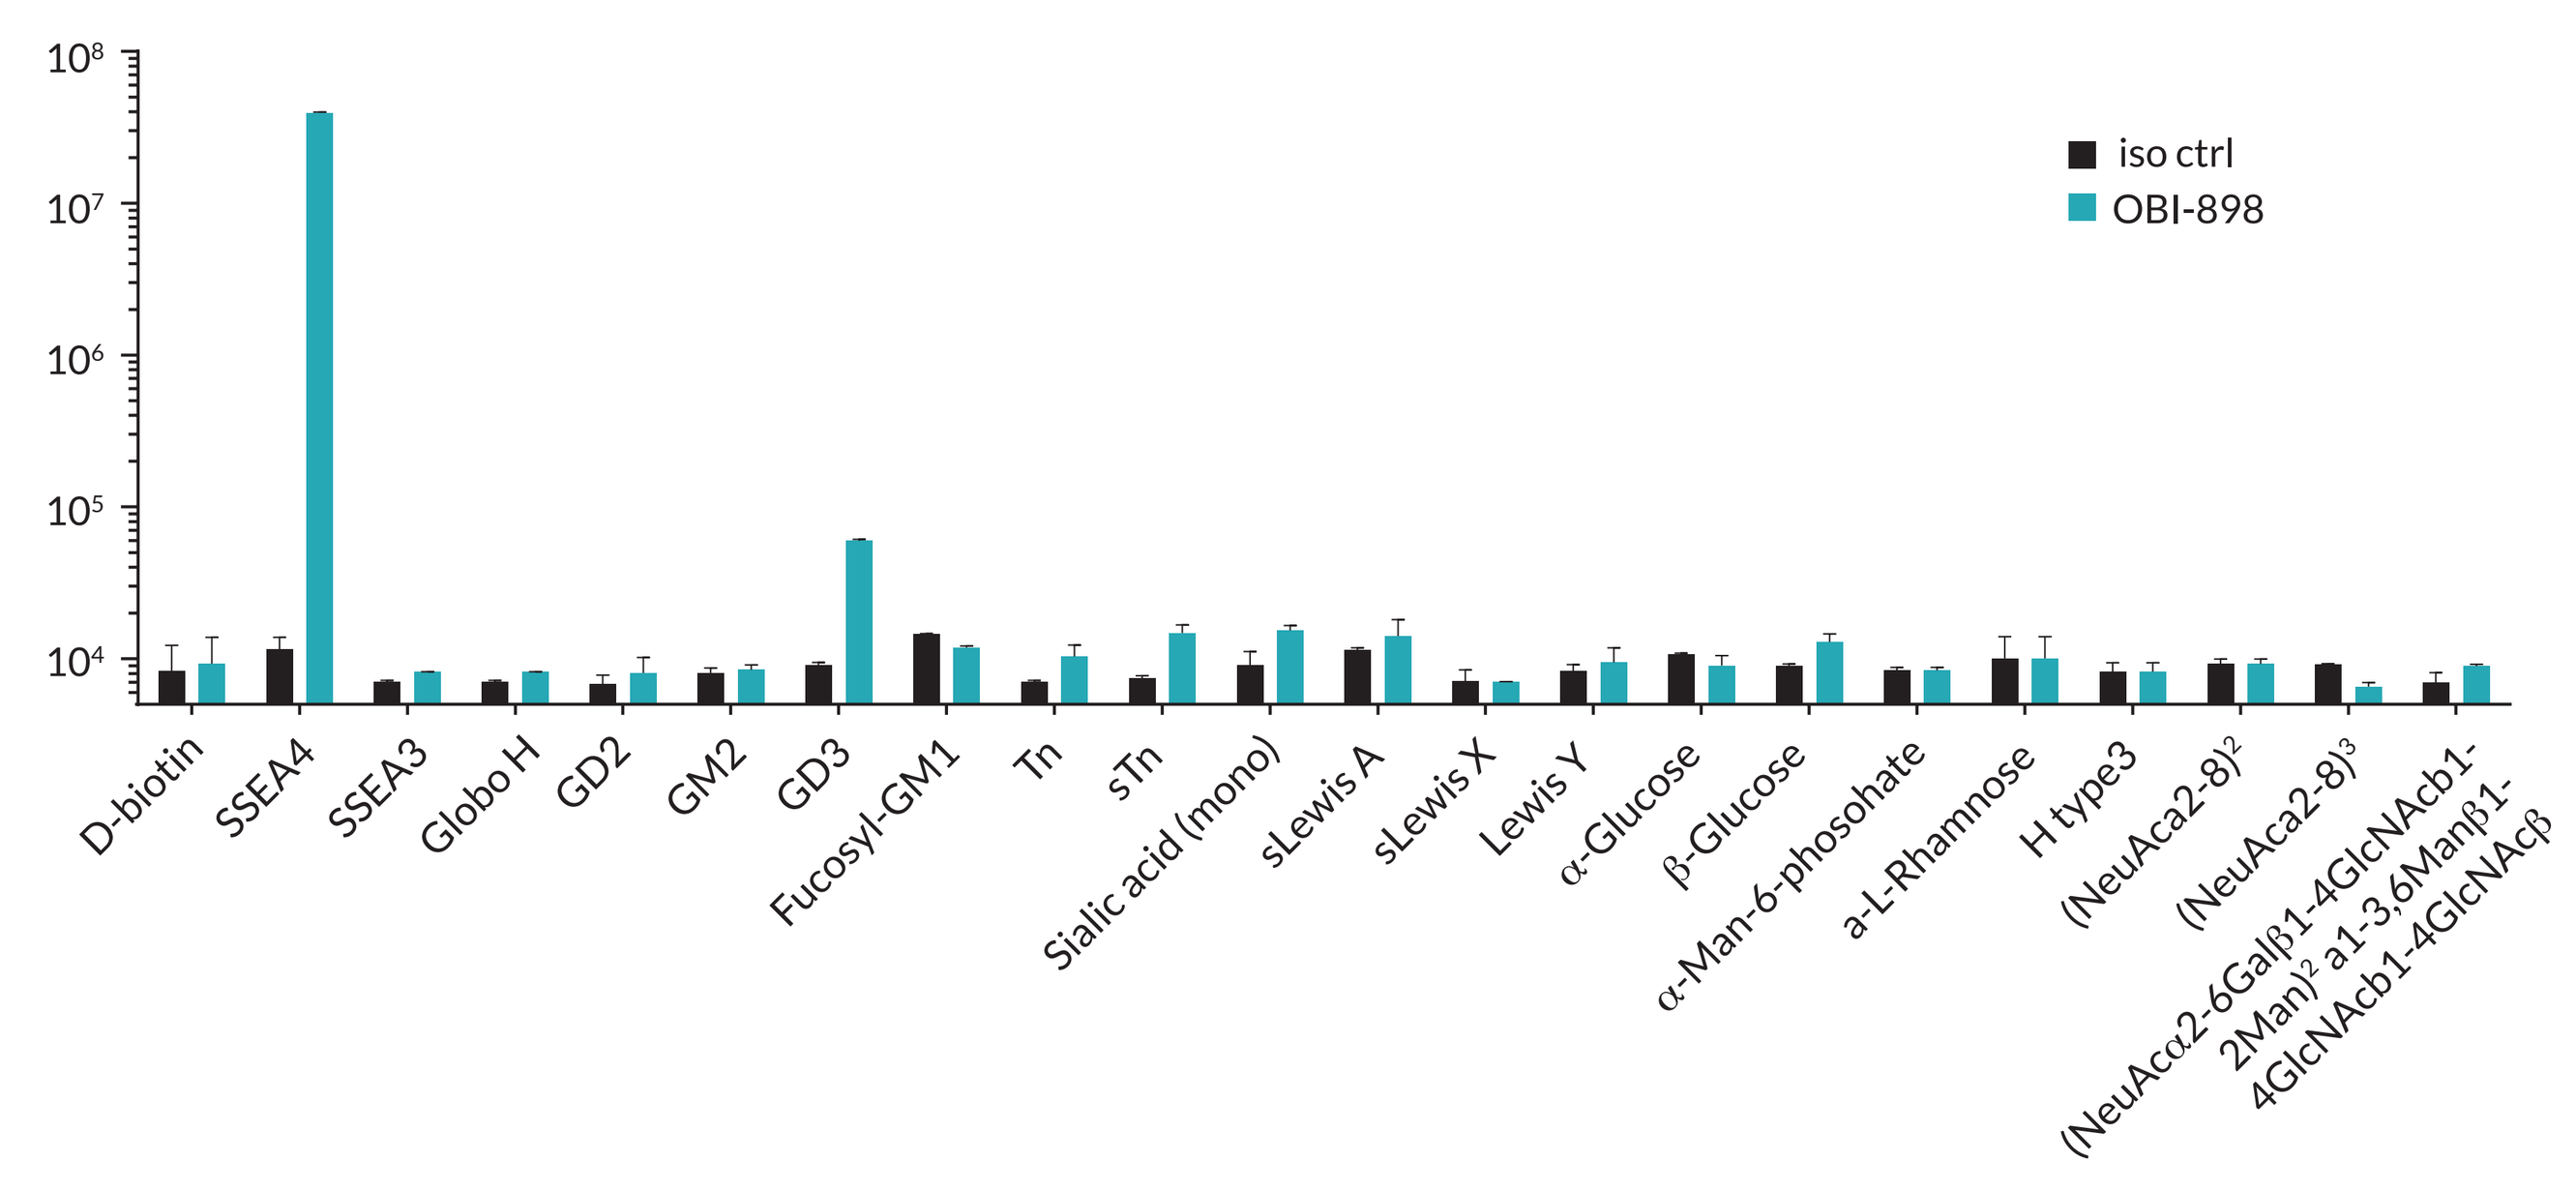
iso ctrl
OBI-898
D-biotin
SSEA4
SSEA3
Globo H
GD2
GM2
GD3
Fucosyl-GM1
Tn
sTn
Sialic acid (mono)
sLewis A
sLewis X
Lewis Y
α-Glucose
β-Glucose
α-Man-6-phosohate
a-L-Rhamnose
H type3
(NeuAcα2-8)²
(NeuAcα2-8)³
(NeuAcα2-6Galβ1-4GlcNAcb1-2Man)² a1-3,6Manβ1-4GlcNAcb1-4GlcNAcβ

(A) In titration enzyme-linked immunosorbent assay (ELISA), serial dilutions of anti-SSEA-4 immunoglobulin G (IgG) (OBI-898, the antibody part of OBI-998 ADC) were incubated with 20 μg SSEA-4-ceramide, Globo H-ceramide, or SSEA-3-ceramide. Mouse anti-SSEA-4 IgG (1J1s), human anti-Globo H antibody (OBI-888), and human anti-SSEA-3-IgG antibody (1E12b) were used as a positive control, respectively. Dose-response binding curves were shown. **(B)** Biotinylated sugars at 50 ng were coated onto the plate and incubated with 2.5 μg/mL of OBI-898 or isotype control (iso ctrl). The amount of OBI-898 bound was examined by chemiluminescent sandwich ELISA analysis.

Cytotoxicity

Figure 2. Cytotoxicity of OBI-998.

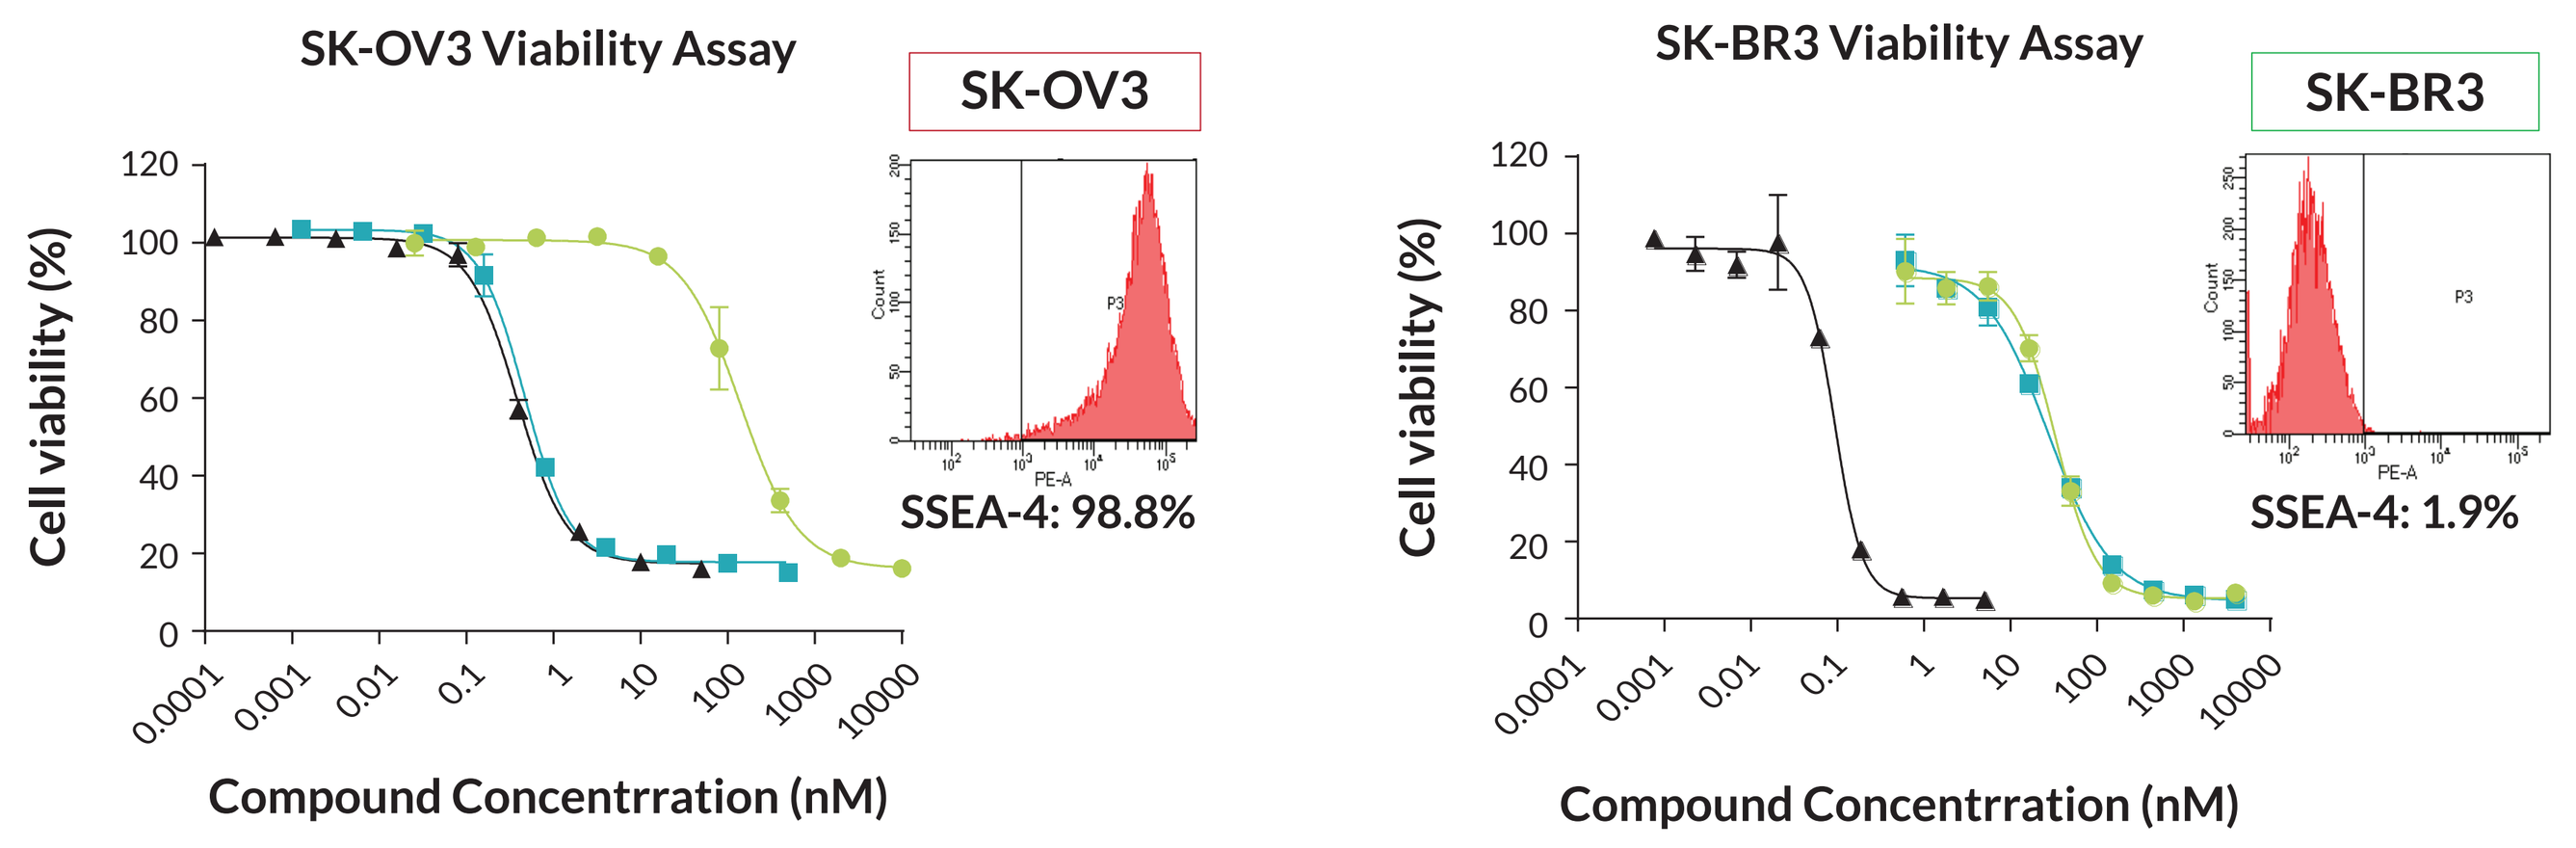
SK-OV3 Viability Assay
SK-OV3
SSEA-4: 98.8%
SK-BR3 Viability Assay
SK-BR3
SSEA-4: 1.9%
Cell viability (%)
Compound Concentrration (nM)

SK-OV3:

		IC_{50} (nM)
	Ctrl ADC	140.3
	OBI-998	0.4684
	MMAE	0.3871

SK-BR3:

		IC_{50} (nM)
	Ctrl ADC	33.4
	OBI-998	27.29
	MMAE	0.09322

OBI-998 showed potent cytotoxic activity on SSEA-4 high expression cancer cell line, SK-OV3, but not on low expression cell, SK-BR3.

SSEA-4 expression level of SK-OV3 and SK-BR3 cell lines are determined using flow cytometry (FACS) surface staining. The SK-OV3 and SK-BR3 cells were treated with OBI-998, anti-CD30 ADC (Ctrl ADC), or MMAE at respective concentrations for 4 days. The cell viability was determined by CellTiter-Glo® viability assay kit.

In vitro bystander effect

Figure 3. *In vitro* bystander effect of OBI-998 in conditioned medium transfer (A) and co-culture system (B).

(A) Conditioned medium transfer

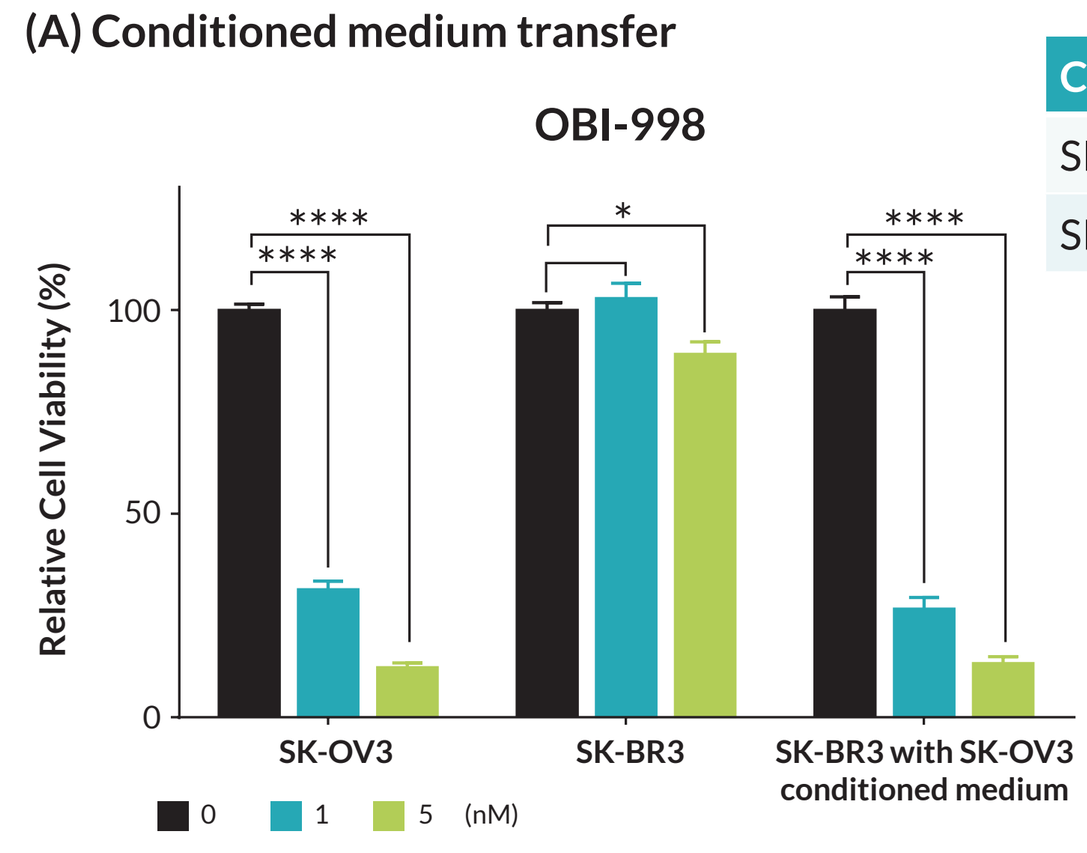
OBI-998
Relative Cell Viability (%)
SK-OV3
SK-BR3
SK-BR3 with SK-OV3 conditioned medium
0
1
5
(nM)

Cell line	SSEA-4 (%)
SK-BR3	98
SK-OV3	2

Cell seeding number: 1 x 10^3
Data represented mean ± SEM
ns > 0.05
*P ≤ 0.05
**P ≤ 0.01
***P ≤ 0.001
****P ≤ 0.0001

(B) Co-culture system

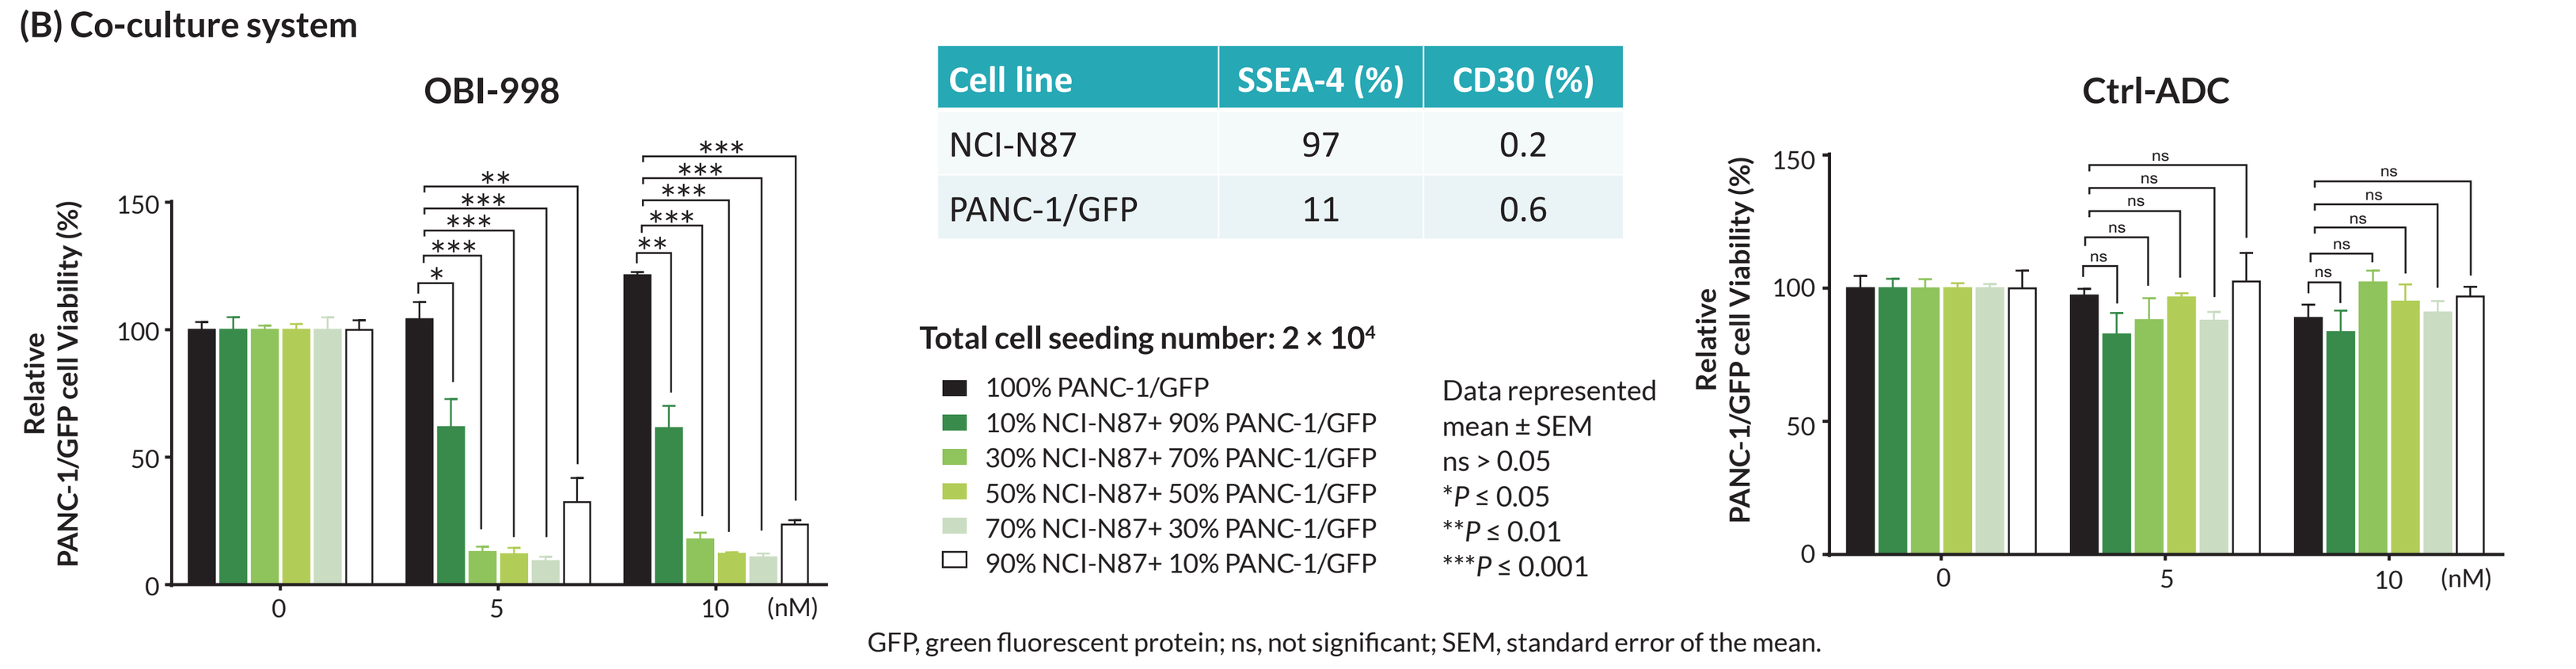
OBI-998
Ctrl-ADC
Relative PANC-1/GFP cell Viability (%)
(nM)

Cell line	SSEA-4 (%)	CD30 (%)
NCI-N87	97	0.2
PANC-1/GFP	11	0.6

Total cell seeding number: 2 × 10^4

100% PANC-1/GFP
10% NCI-N87+ 90% PANC-1/GFP
30% NCI-N87+ 70% PANC-1/GFP
50% NCI-N87+ 50% PANC-1/GFP
70% NCI-N87+ 30% PANC-1/GFP
90% NCI-N87+ 10% PANC-1/GFP

Data represented mean ± SEM
ns > 0.05
**P* ≤ 0.05
***P* ≤ 0.01
****P* ≤ 0.001

GFP, green fluorescent protein; ns, not significant; SEM, standard error of the mean.

(A) SSEA-4 expression levels of SK-OV3 and SK-BR3 cell lines are determined using FACS surface staining. SK-OV3 cells were treated with 0, 1, and 5 nM of OBI-998 for 5 days; then the supernatant was transferred to treat SK-BR3 cells for another 5 days (**SK-BR3 with SK-OV3 conditioned medium**). The SK-OV3 and SK-BR3 cells were treated with OBI-998 at respective concentrations for 5 days as positive (**SK-OV3**) and negative (**SK-BR3**) controls, respectively. The cell viability of treated cells relative to corresponding untreated cells (0 nM) was determined by CellTiter-Glo® viability assay kit. Statistically significant differences were indicated (P < 0.05).

(B) SSEA-4 and CD30 expression levels in NCI-N87 and PANC-1 transfected with green fluorescent protein (GFP) (PANC-1/GFP) were determined using FACS surface staining. PANC-1/GFP cells alone or co-cultured with NCI-N87 cells in different ratios were treated with either OBI-998 or Ctrl-ADC (anti-CD30-MMAE) at various concentrations (0, 5, 10 nM) for 8 days. The cell viability of PANC-1/GFP cells relative to corresponding untreated cells (0 nM) was calculated by GFP fluorescence intensity. Statistically significant differences were indicated (P < 0.05).

Internalization

Figure 4. Internalization kinetics of OBI-998 in HCC1428 cells.

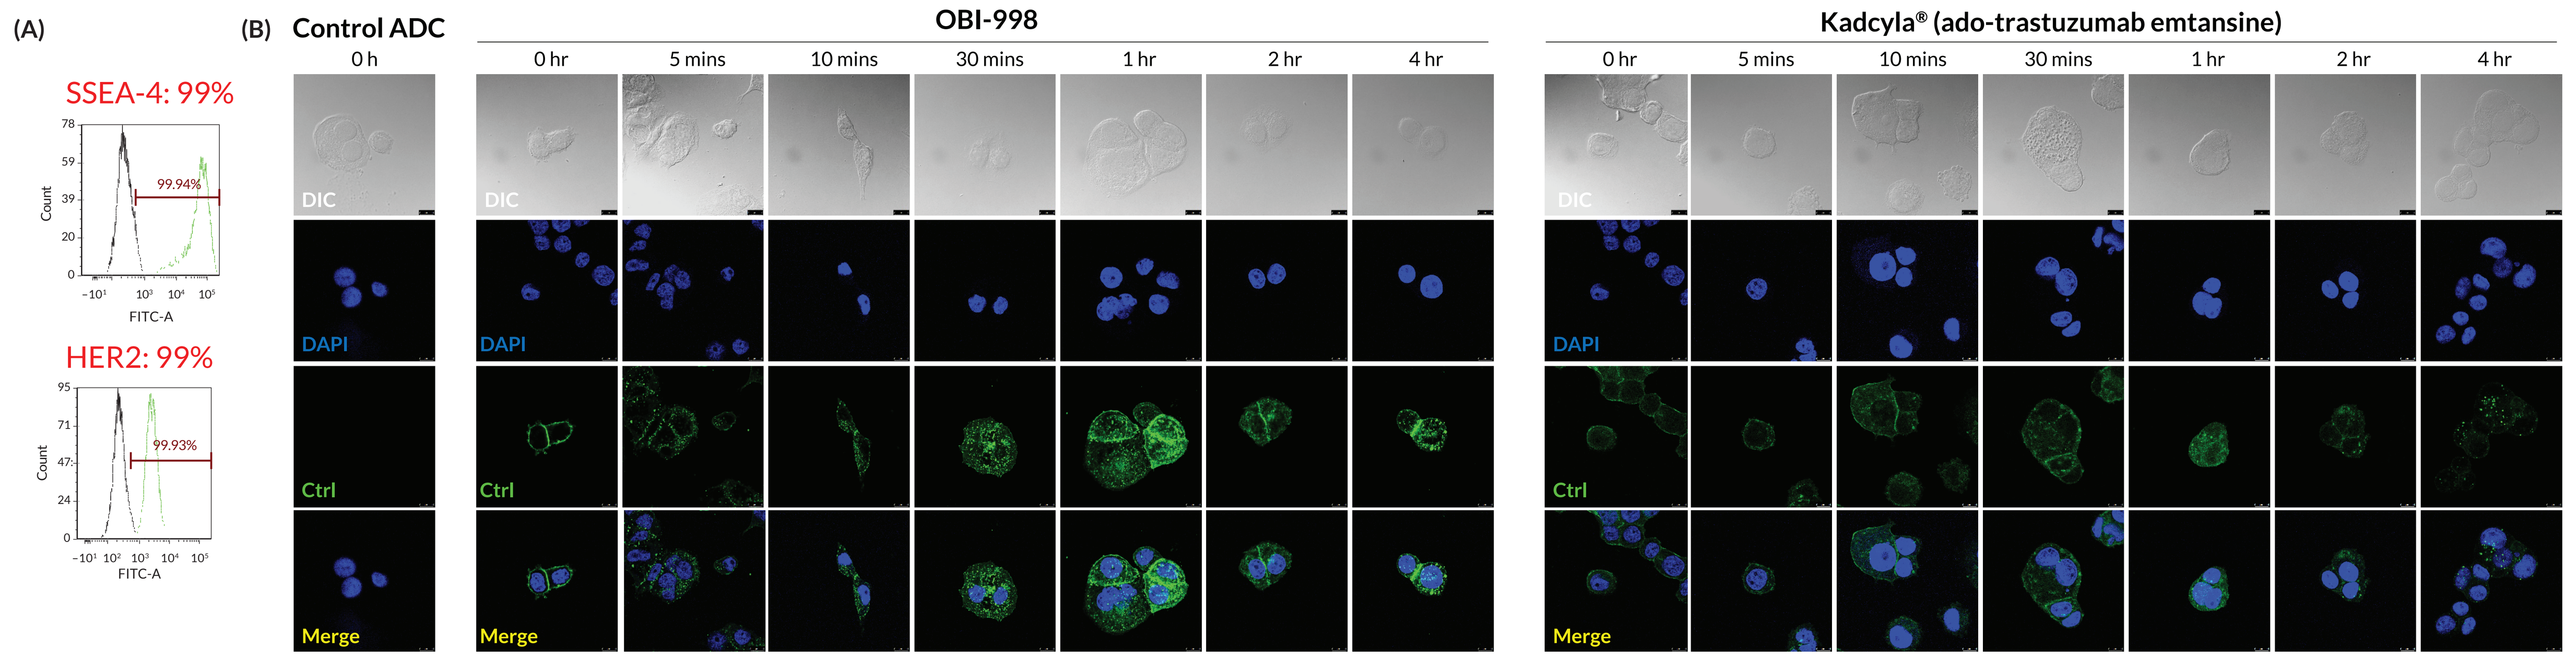
(A)
SSEA-4: 99%
99.94%
HER2: 99%
99.93%
FITC-A
Count
(B)
Control ADC
0 h
OBI-998
0 hr
5 mins
10 mins
30 mins
1 hr
2 hr
4 hr
Kadcyla® (ado-trastuzumab emtansine)
DIC
DAPI
Ctrl
Merge

(A) SSEA-4 and human epidermal growth factor receptor-2 (HER2) expression levels of HCC1428 cell line are determined using FACS surface staining. **(B)** HCC1428 cells were incubated with OBI-998 or Kadcyla® (ado-trastuzumab emtansine; 1 μg/mL) for 1 hr at 4°C and then fixed (0 hr) or transferred to prewarmed media and maintained at 37°C for 5 mins, 10 mins, 30 mins, 1 hr, 2 hr, and 4 hr before fixation. They were then fixed and incubated with Alexa Fluor® 488-conjugated anti-human immunoglobulin G (IgG) for detection of SSEA-4 or HER2. Cells were incubated with Control ADC, anti-CD30-MMAE (1 μg/mL) as a negative target-binding control (ctrl) and only observed at 0 hr. Cell nuclei were stained by DAPI.

Anti-tumor efficacy

Figure 5. Anti-tumor efficacy of OBI-998 in multiple cancer types.

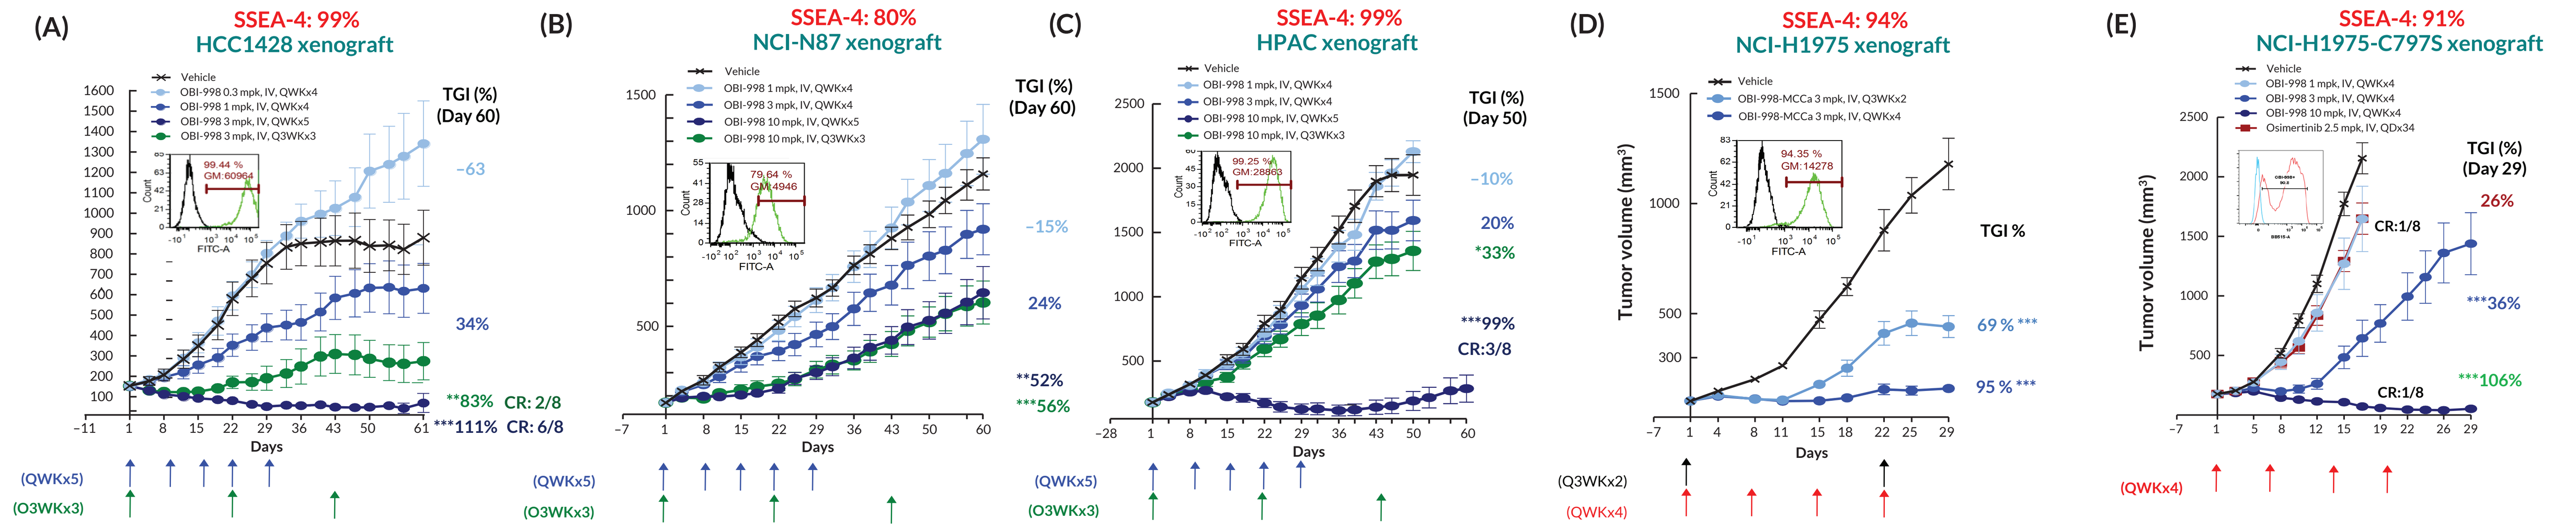
(A)
SSEA-4: 99%
HCC1428 xenograft
TGI (%) (Day 60)
−63
34%
**83% CR: 2/8
***111% CR: 6/8
(QWKx5)
(O3WKx3)
(B)
SSEA-4: 80%
NCI-N87 xenograft
TGI (%) (Day 60)
−15%
24%
**52%
***56%
(C)
SSEA-4: 99%
HPAC xenograft
TGI (%) (Day 50)
−10%
20%
*33%
***99%
CR:3/8
(D)
SSEA-4: 94%
NCI-H1975 xenograft
TGI %
69 % ***
95 % ***
(Q3WKx2)
(QWKx4)
(E)
SSEA-4: 91%
NCI-H1975-C797S xenograft
TGI (%) (Day 29)
26%
***36%
***106%
CR:1/8
Tumor volume (mm³)
Days

SSEA-4 expression levels in HCC1428 breast cancer **(A)**, NCI-N87 gastric cancer **(B)**, HPAC pancreatic cancer **(C)**, NCI-H1975 lung cancer cell line (*EGFR* L858R/T790M mutation and is sensitive to osimertinib) **(D)**, and NCI-H1975-C797S lung cancer (*EGFR* exon 19 deletion/T790M/C797S mutation and is resistant to osimertinib) **(E)** cell lines are determined by FACS surface staining. 6- to 8-week-old BALB/c nude female mice were subcutaneously implanted with viable respective cancer cells. Tumor implanted mice were divided into treatment groups, each group containing 8 animals. Dose administrations were initiated at 11 days **(A)** or 7 days **(B-D)** post tumor cell implantation (denoted as day 1). Vehicle and OBI-998 at indicated doses were administered intravenously once weekly or once every 3 weeks. Tumor size was monitored and recorded twice weekly for 61 days **(A)**, 60 days **(B,C)**, or 29 days **(D,E)**. Tumor growth inhibition (TGI) rate was calculated at the end of study. Data are represented as mean ± SEM. CR: complete regression.

Pharmacokinetics and tissue distribution

Figure 6. Pharmacokinetic profile of OBI-998.

(A) Pharmacokinetic profiles in serum

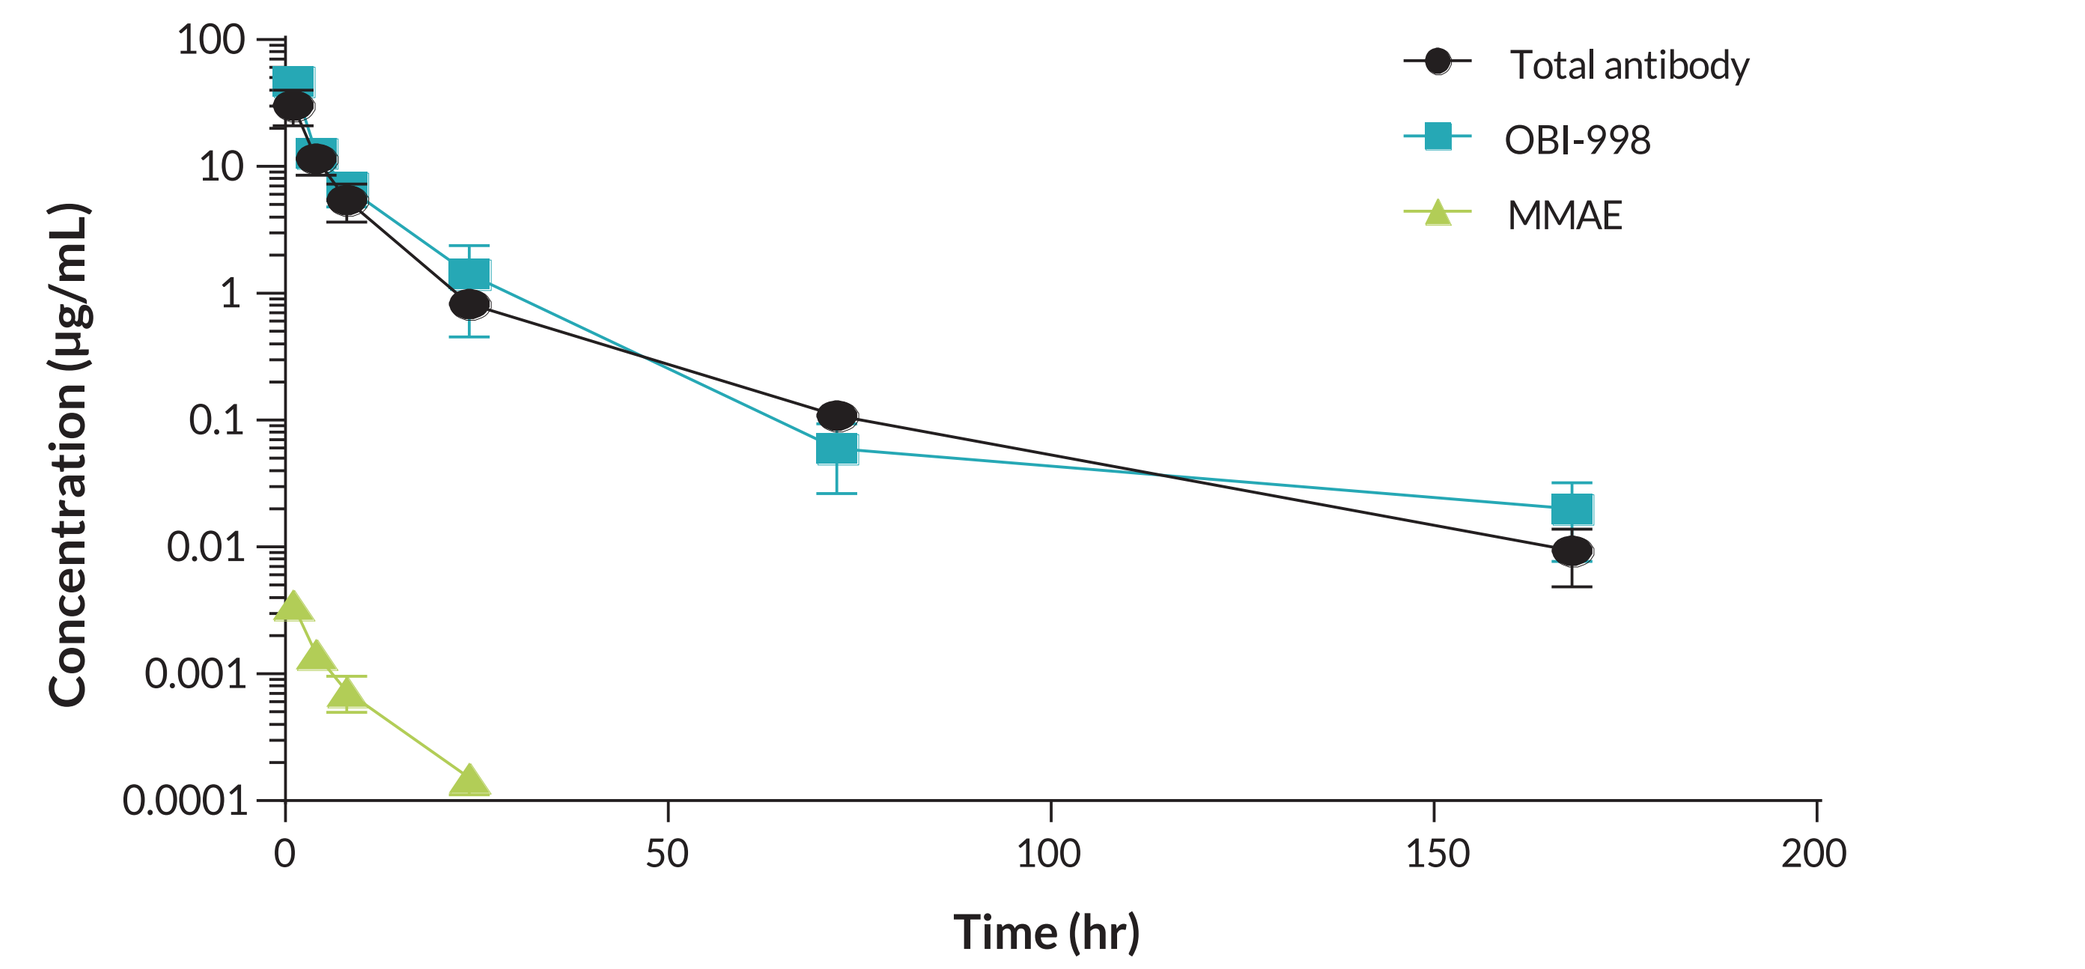
Total antibody
OBI-998
MMAE
Concentration (μg/mL)
Time (hr)

(B) Tissue distribution

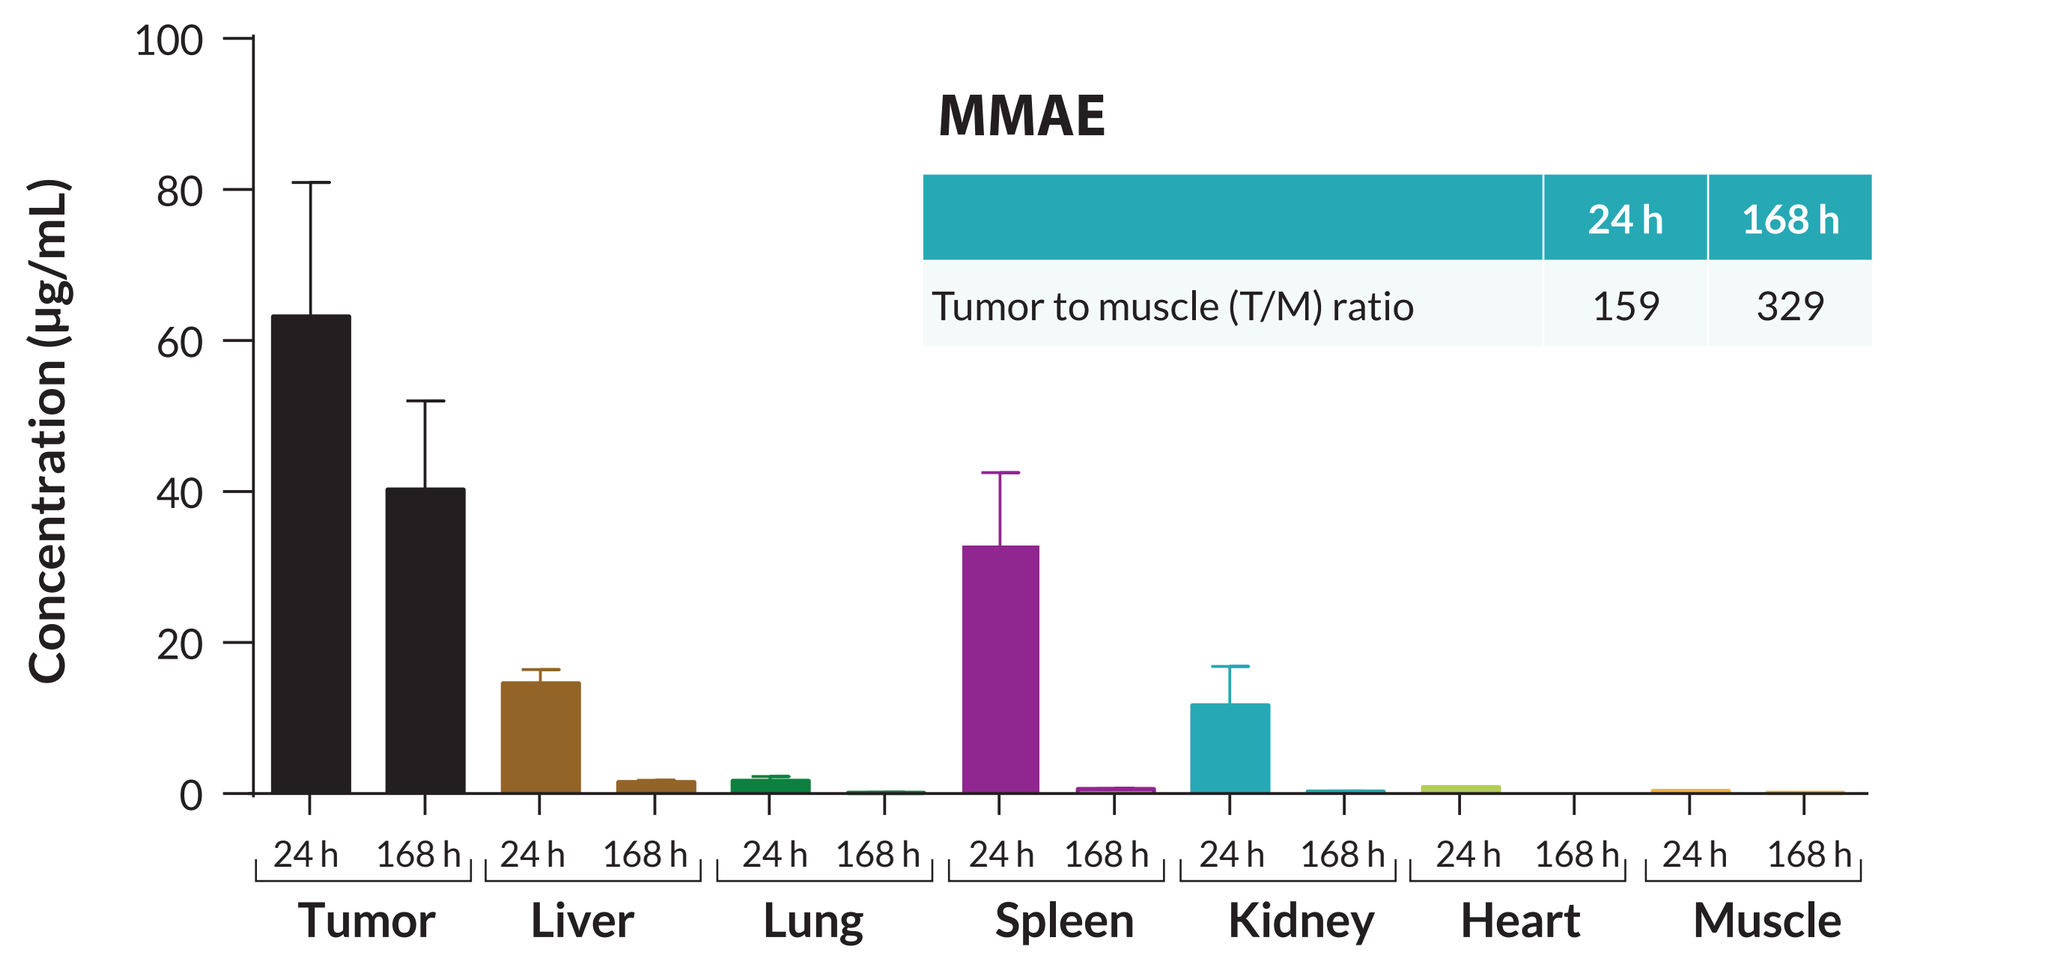
MMAE
Concentration (μg/mL)
24 h
168 h
Tumor
Liver
Lung
Spleen
Kidney
Heart
Muscle

	24 h	168 h
Tumor to muscle (T/M) ratio	159	329

(A) *In vivo* pharmacokinetic profiles from 5 mg/kg OBI-998-injected HCC1428 tumor-bearing mice (n=5). **(B)** Organs and tumor tissues from HCC1428 tumor-bearing mice were harvested and homogenized to evaluate free MMAE concentration at 24 to 168 hr post injection. Total antibody and OBI-998 concentrations were determined by ELISA coated with SSEA-4-lipid and captured by anti-human antibody and anti-MMAE antibody, respectively. The concentration of MMAE was evaluated by liquid chromatography tandem mass spectrometry (LC/MS/MS).

CONCLUSIONS

- OBI-998 is a novel ADC targeting SSEA-4 that possesses desired properties such as high target specificity, rapid internalization, potent cytotoxicity, and significant bystander effects
- OBI-998 showed a high level of deposition and a persistent presence of MMAE in tumors and significant anti-tumor efficacy in a variety of animal models
- The results support the further development of OBI-998 as a therapeutic agent for SSEA-4-targeting cancer therapy

REFERENCES

1. Kannagi R, et al. *EMBO J.* 1983;2:2355–2361.
2. Lou YW, et al. *Proc Natl Acad Sci U S A.* 2014;111:2482–2487.
3. Saito S, et al. *Jpn J Cancer Res.* 1997;88:652–659.
4. Gottschling S, et al. *Eur Respir J.* 2013;41:656–663.
5. Huang YL, et al. *Proc Natl Acad Sci U S A.* 2013;110:2517–2522.
6. Ye F, et al. Int *J Gynecol. Cancer.* 2010;20:958–964.
7. Noto Z, et al. *Oral Oncol.* 2013;49:787–795.
8. Aloia A, et al. *Breast Cancer Res.* 2015;17:146.

Prepared for American Association for Cancer Research (AACR) 2021 Virtual Meeting, April 10-15 and May 17-21, 2021.