Inhibitory Activity of Globo H and SSEA-4 on Activated T-cells

Tzer-Min Kuo[1], Chin-Chan Lee[1], Jiann-Shiun Lai[1], Chung-Chen Su[1], and Ming-Tain Lai[1]

[1]OBI Pharma, Inc., Taipei, Taiwan

AACR 2021
3176

BACKGROUND

- Globo H (GH) and stage-specific embryonic antigen-4 (SSEA-4) are Globo-series glycosphingolipid (GSL) antigens that are highly expressed on a variety of epithelial cancers
- GH and SSEA-4 are reported to play an important role in cancer cell development and survival and are therefore promising targets as immunotherapeutic agents for cancer treatment
- Shedding of GSLs by tumor cells and uptake by neighboring cells may generate a hospitable environment for tumor progression and survival within the tumor microenvironment
- GH shed from tumor cells was shown to promote angiogenesis and suppress immune responses in vitro. In addition, exogenous Globo H ceramide (GHCer) inhibits proliferation of human peripheral blood mononuclear cells (PBMCs) and murine CD4+ T-cells
- Given the important role of sialic acid in the regulation of the immune system, SSEA-4, a polysaccharide with sialic acid at the terminal end, may play an important role in the regulation of T-cell responses

OBJECTIVE

- This study aims to investigate the GHCer and SSEA-4 ceramide (SSEA-4Cer) effects on T-cell activation

RESULTS

Globo H (GH) ceramide (GHCer) inhibited tumor-infiltrating lymphocytes (TILs) in mice

Figure 1. GHCer-enhanced syngeneic B16F10 melanoma tumor growth and decreased tumor-infiltrating lymphocytes in mice.

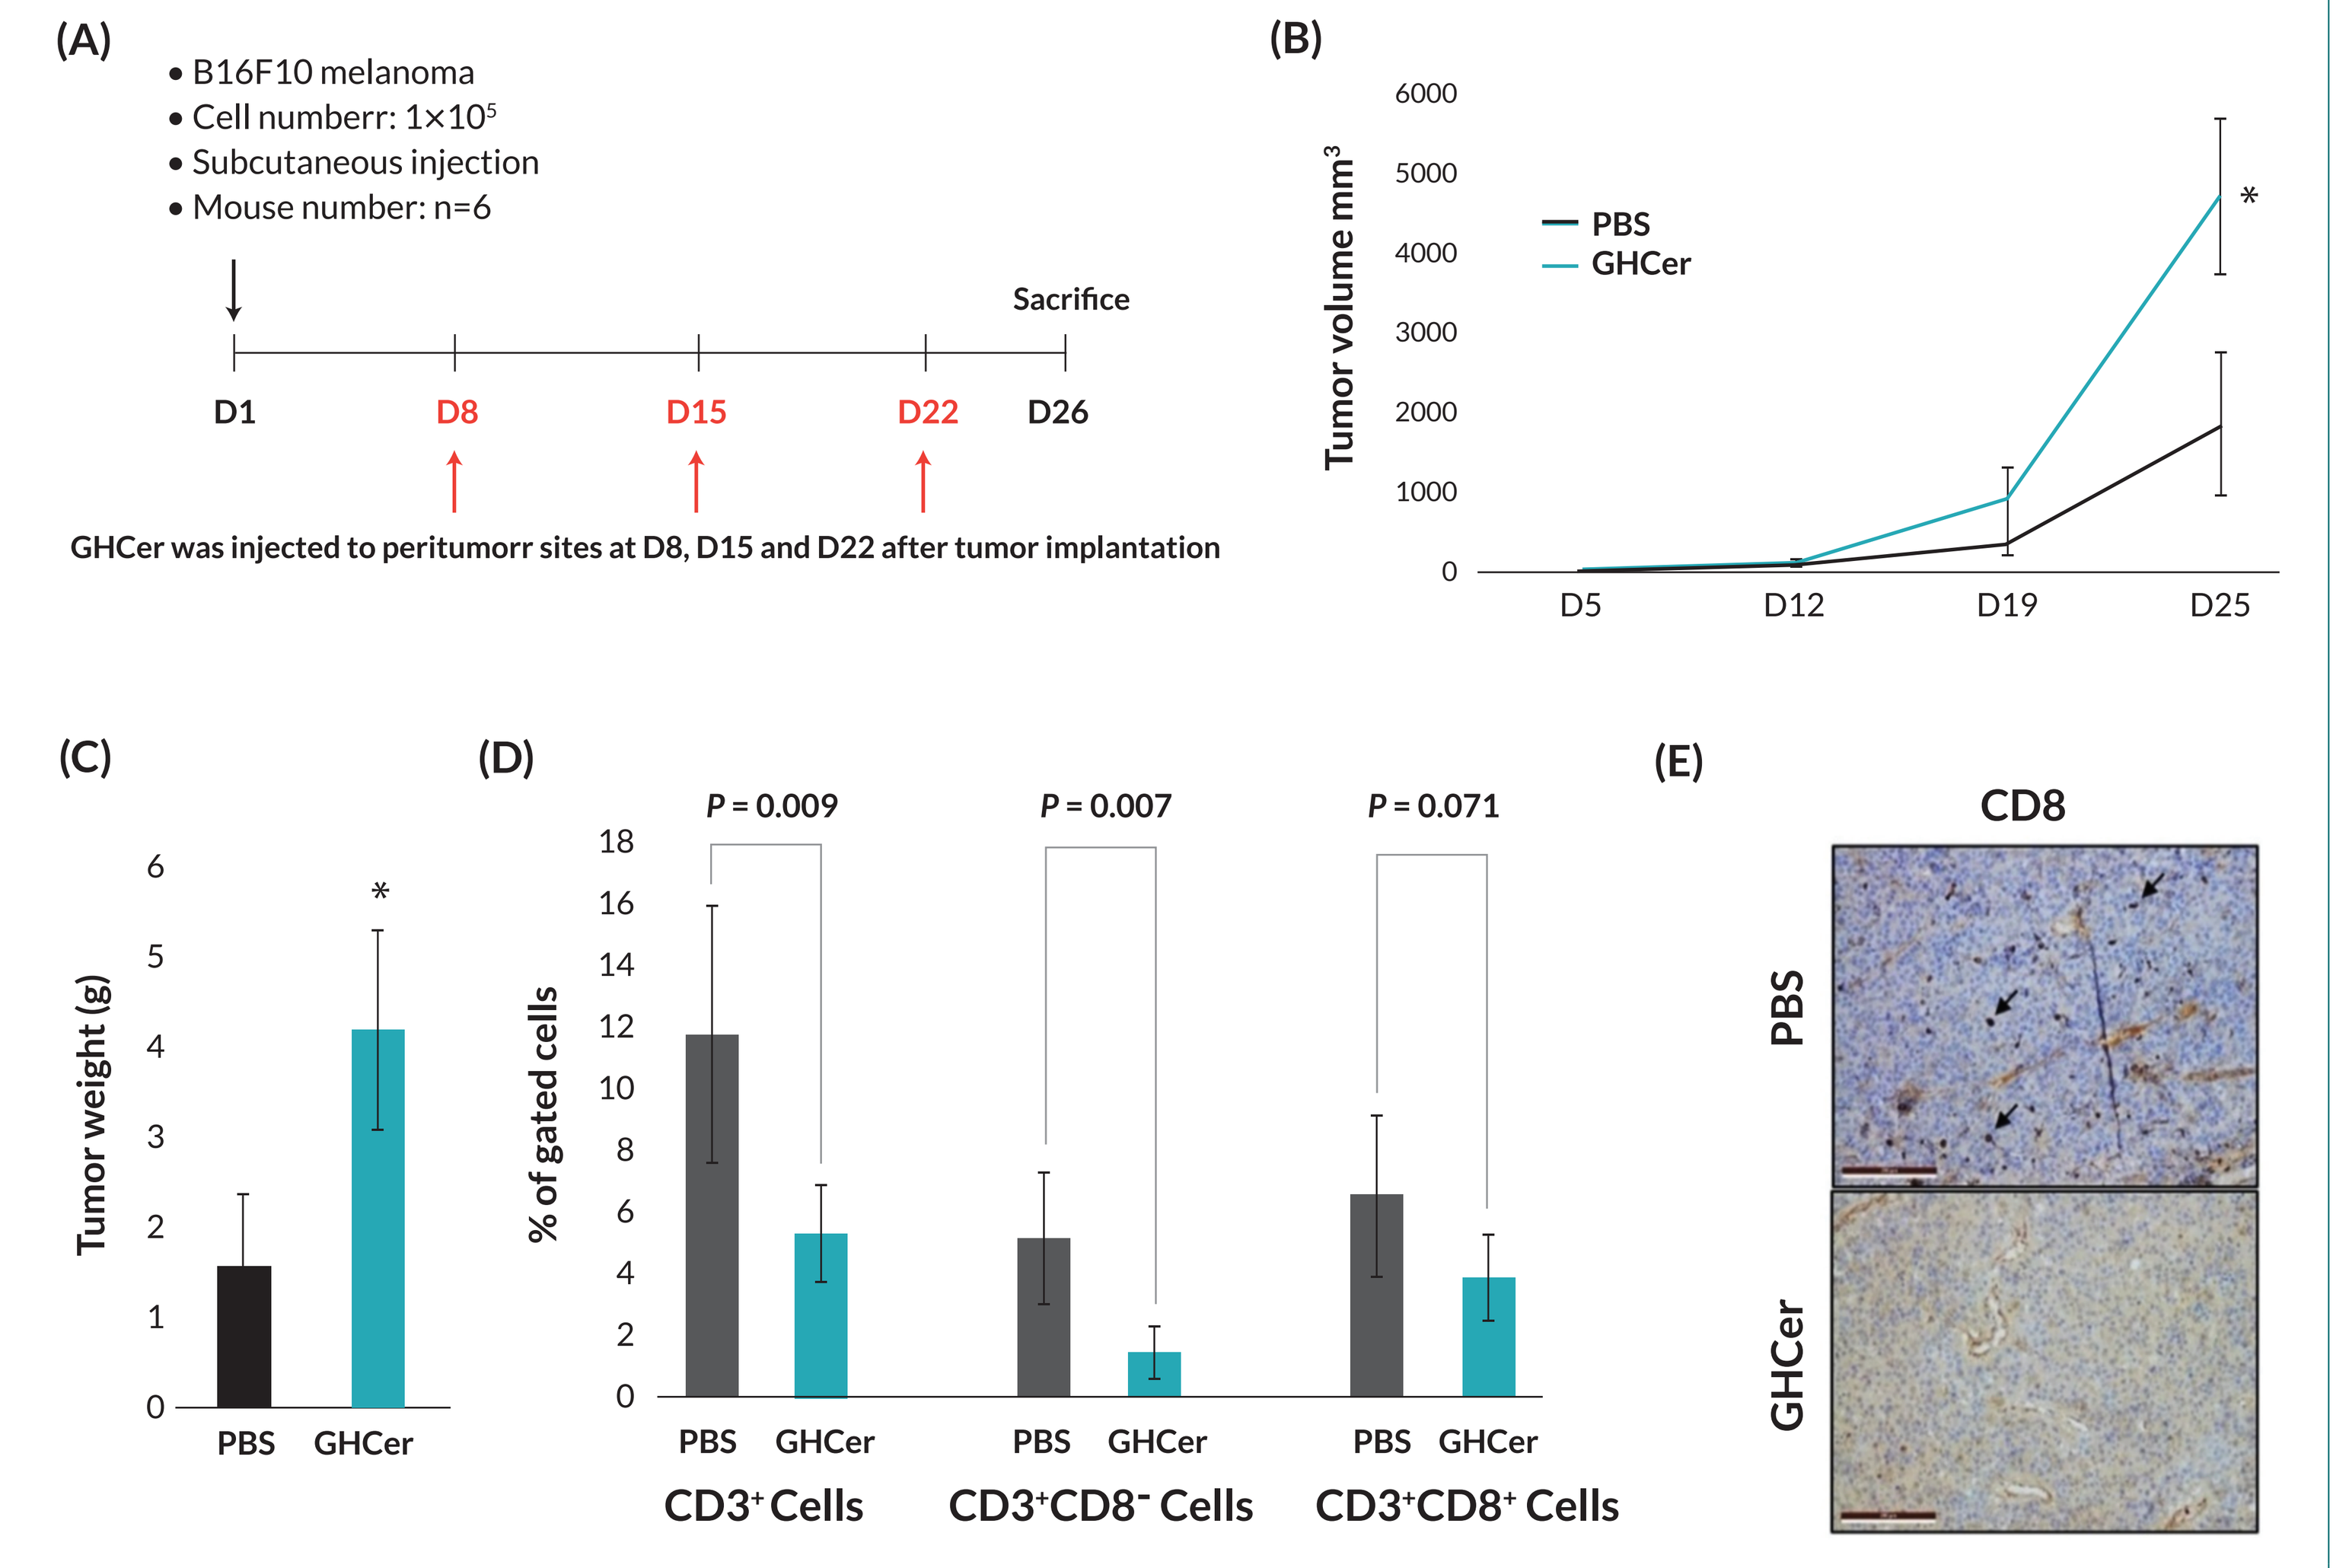
(A)
• B16F10 melanoma
• Cell numberr: 1×10⁵
• Subcutaneous injection
• Mouse number: n=6
Sacrifice
D1
D8
D15
D22
D26
GHCer was injected to peritumorr sites at D8, D15 and D22 after tumor implantation
(B)
Tumor volume mm³
PBS
GHCer
D5
D12
D19
D25
(C)
Tumor weight (g)
PBS
GHCer
(D)
% of gated cells
P = 0.009
P = 0.007
P = 0.071
CD3⁺ Cells
CD3⁺CD8⁻ Cells
CD3⁺CD8⁺ Cells
(E)
CD8
PBS
GHCer

(A) Schematic view of experimental design and time line. C57BL/6 mice (n=6) aged 5 weeks were allografted with B16F10 cells subcutaneously 7 days prior to treatment with 5 μM GHCer or phosphate-buffered saline (PBS) buffer control 3 times by peritumoral injection. The tumor size was monitored by a caliper. All mice were killed on day 26; tumors were collected for further analysis. GHCer increased on B16F10 melanoma tumor growth in **(B)** size (2.54-fold increase of tumor volume; *$P < 0.05$) and **(C)** weight in mice. **(D)** Flow cytometry (FACS) quantification of the percentage of CD3+, CD3+CD8−, and CD3+CD8+ cells in tumor-infiltrating lymphocytes (TILs) of B16F10 melanoma tumor in mice. GHCer significantly decreased intratumoral total CD3+ and CD3+CD8− cells. **(E)** Lower numbers of CD8+ cells were observed by immunohistochemistry staining in the internal tumor tissues of the GHCer group as compared with PBS control. Scale bar, 200 μm.

Uptake of GHCer and stage specific embryonic antigen-4 (SSEA-4) ceramide (SSEA-4Cer) by T-cells

Figure 2. Uptake of GHCer and SSEA-4Cer by T-cells.

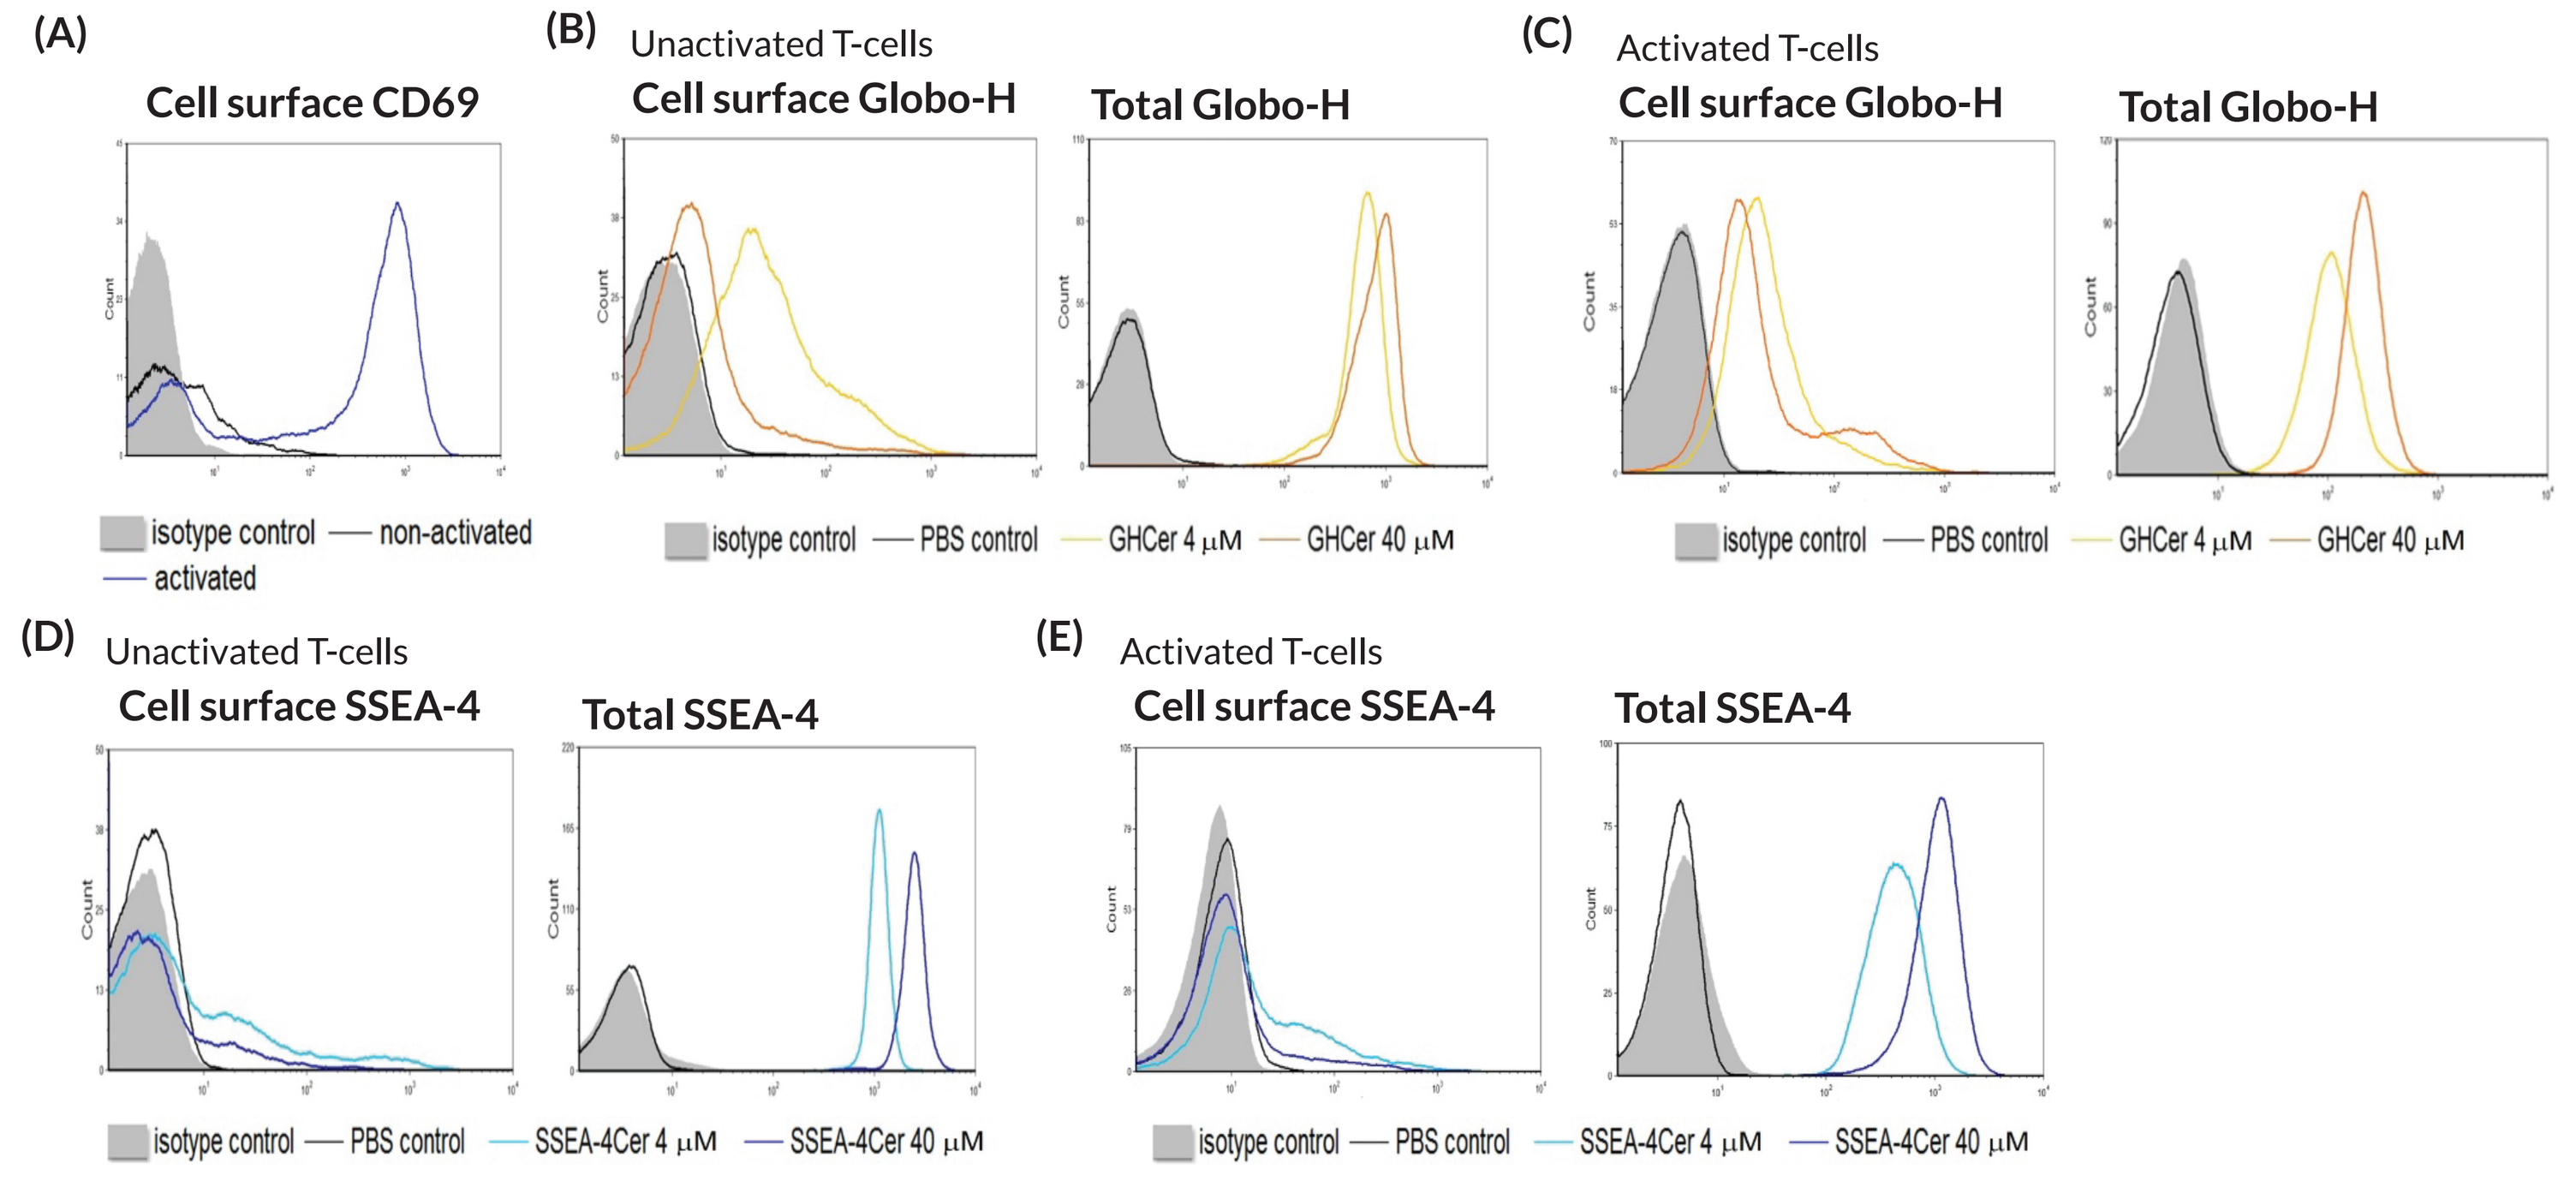
(A)
Cell surface CD69
isotype control
non-activated
activated
(B) Unactivated T-cells
Cell surface Globo-H
Total Globo-H
isotype control
PBS control
GHCer 4 μM
GHCer 40 μM
(C) Activated T-cells
Cell surface Globo-H
Total Globo-H
(D) Unactivated T-cells
Cell surface SSEA-4
Total SSEA-4
SSEA-4Cer 4 μM
SSEA-4Cer 40 μM
(E) Activated T-cells
Cell surface SSEA-4
Total SSEA-4

T-cells isolated from human PBMCs were stimulated using Dynabeads® coated with anti CD3/CD28 monoclonal antibodies (mAbs) for 48 hr. **(A)** Activation was determined by monitoring the upregulation of CD69 on live T-cells by flow cytometry. Unactivated and activated T-cells were treated with or without 4 and 40 μM GHCer or SSEA-4Cer for 48 hr. Cell surface and total (combined cell surface and intracellular staining) GHs in **(B)** unactivated and **(C)** activated T-cells were detected, as well as SSEA-4 in **(D)** unactivated and **(E)** activated T-cells.

GHCer and SSEA-4Cer reduced cell proliferation of activated T-cells

Figure 3. GHCer and SSEA-4Cer reduced cell proliferation of activated but not unactivated T-cells.

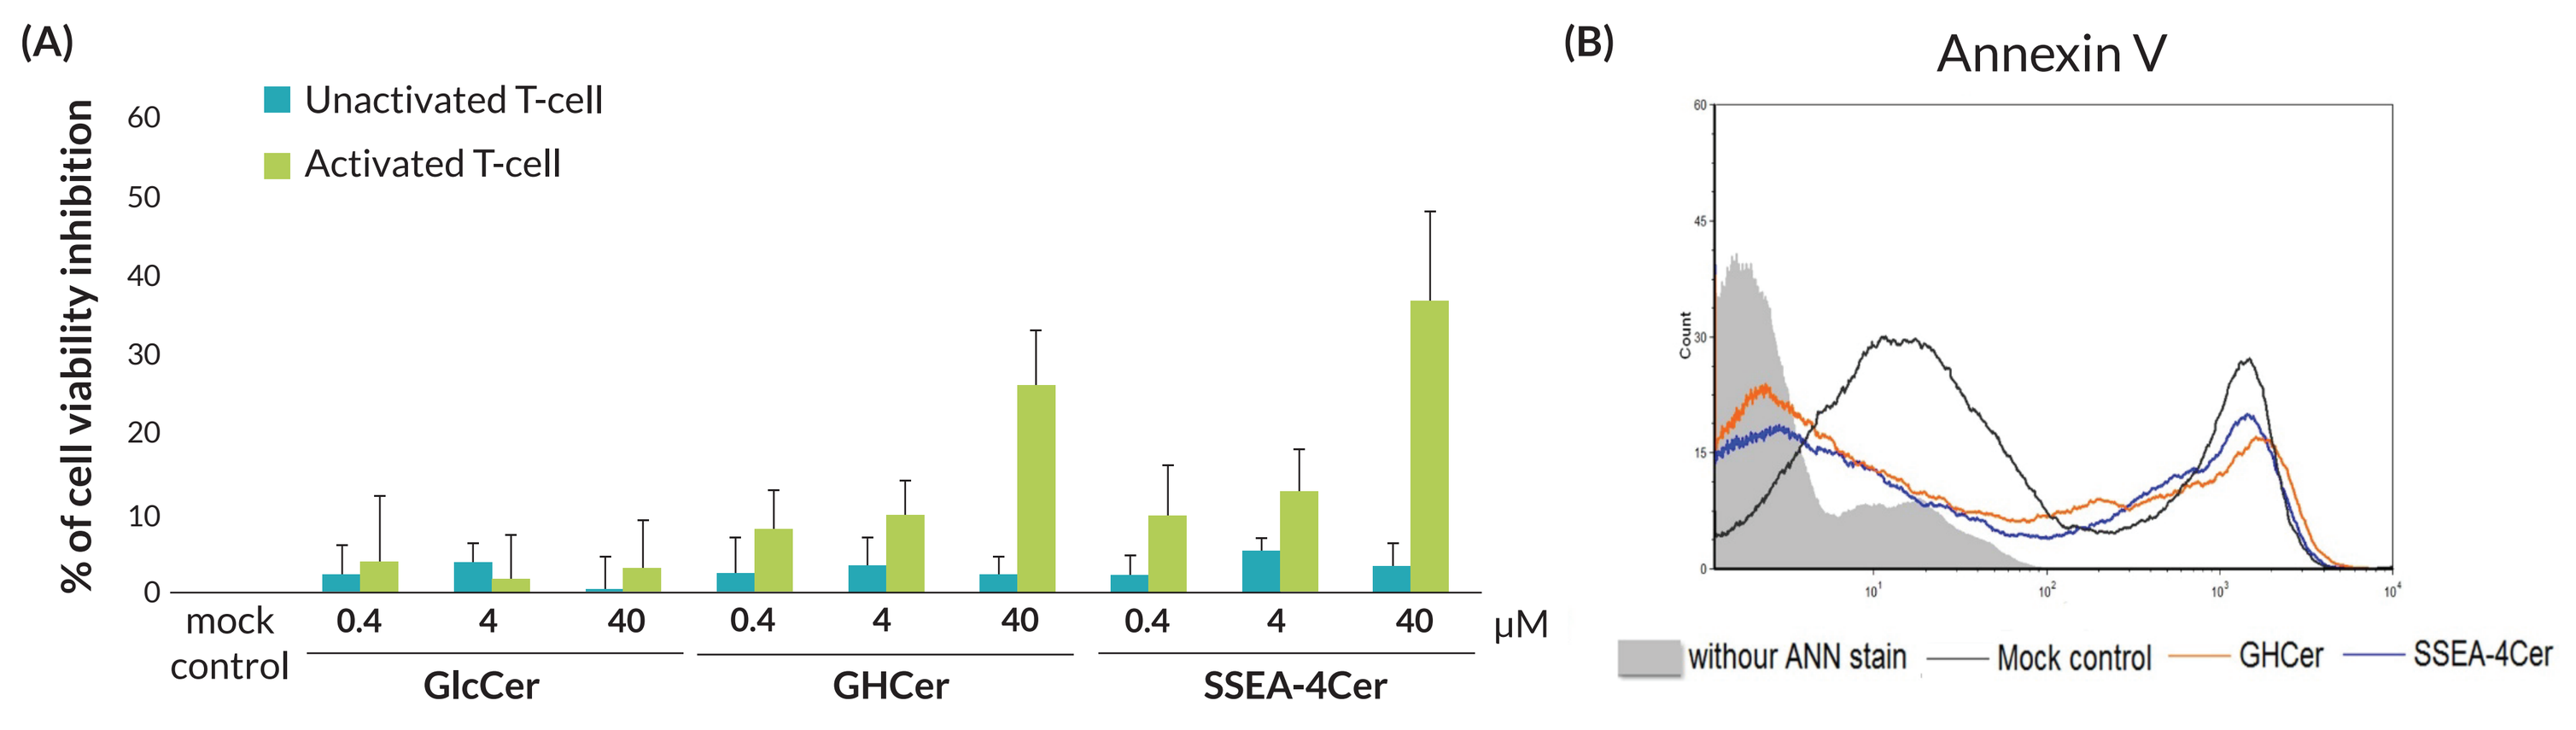
(A)
% of cell viability inhibition
Unactivated T-cell
Activated T-cell
mock control
0.4
4
40
μM
GlcCer
GHCer
SSEA-4Cer
(B)
Annexin V
withour ANN stain
Mock control
GHCer
SSEA-4Cer

T-cells isolated from human PBMC were activated with anti-CD3/CD28 mAbs for 48 hr. **(A)** Reduction of cell viability in activated T-cells by GHCer and SSEA-4Cer. MTS assay was performed in unactivated and activated T-cells at 48 hr glucosylceramide (GlcCer), GHCer, and SSEA-4Cer treatment (0, 0.4, 4, or 40 μM). Results depict mean ± standard deviation (SD) of >5 independent experiments using T-cells from 5 individuals (*$P < 0.05$ vs mock controls). **(B)** Detection of GHCer and SSEA-4Cer-induced apoptosis in activated T-cells by Annexin V staining. After treatment for 48 hr, 40 μM GHCer and SSEA-4Cer have no impact on apoptosis induction in activated T-cells.

GHCer and SSEA-4Cer induced T-cell arrest

Figure 4. Inhibition of T-cell activation by GHCer and SSEA-4Cer.

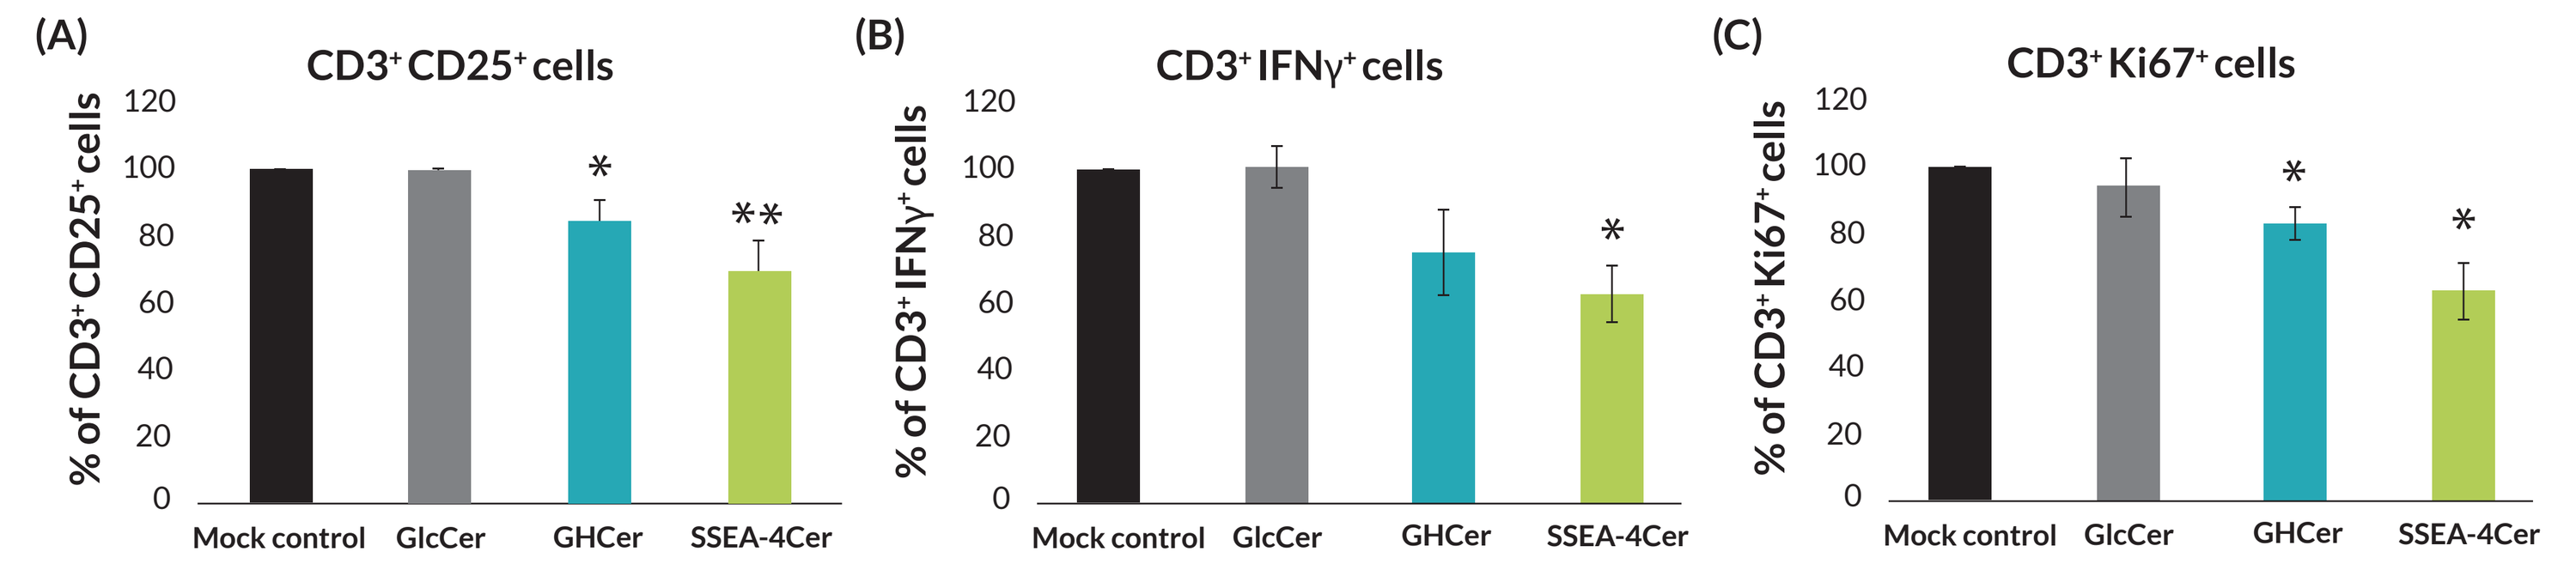
(A)
CD3⁺CD25⁺ cells
% of CD3⁺CD25⁺ cells
(B)
CD3⁺IFNγ⁺ cells
% of CD3⁺IFNγ⁺ cells
(C)
CD3⁺Ki67⁺ cells
% of CD3⁺Ki67⁺ cells
Mock control
GlcCer
GHCer
SSEA-4Cer

Human PBMCs were activated *in vitro* with anti-CD3/CD28 for 48h and then were treated with 40 μM GlcCer, GHCer and SSEA-4Cer for 48 h. Cells were further evaluated for the percentage of CD3+ cells expressing T-cell activation marker CD25, cytokine IFNγ and cell proliferation Ki67 by a flow cytometry. Value in FACS plots indicates the percentage of total cells within the quadrant shown. GHCer and SSEA-4Cer reduced **(A)** CD3+CD25+, **(B)** CD3+IFNγ+ and **(C)** CD3+Ki67+ cells in PBMCs. Compiled data from more than three experiments are shown and are expressed as mean ± SD. *$P < 0.05$ versus mock controls . **$P < 0.01$ versus mock controls.

GHCer and SSEA-4Cer induced T-cell suppression is primarily through reduction of CD8+ T-cell proliferation

Figure 5. GHCer and SSEA-4Cer inhibit proliferation of CD8+ T-cells.

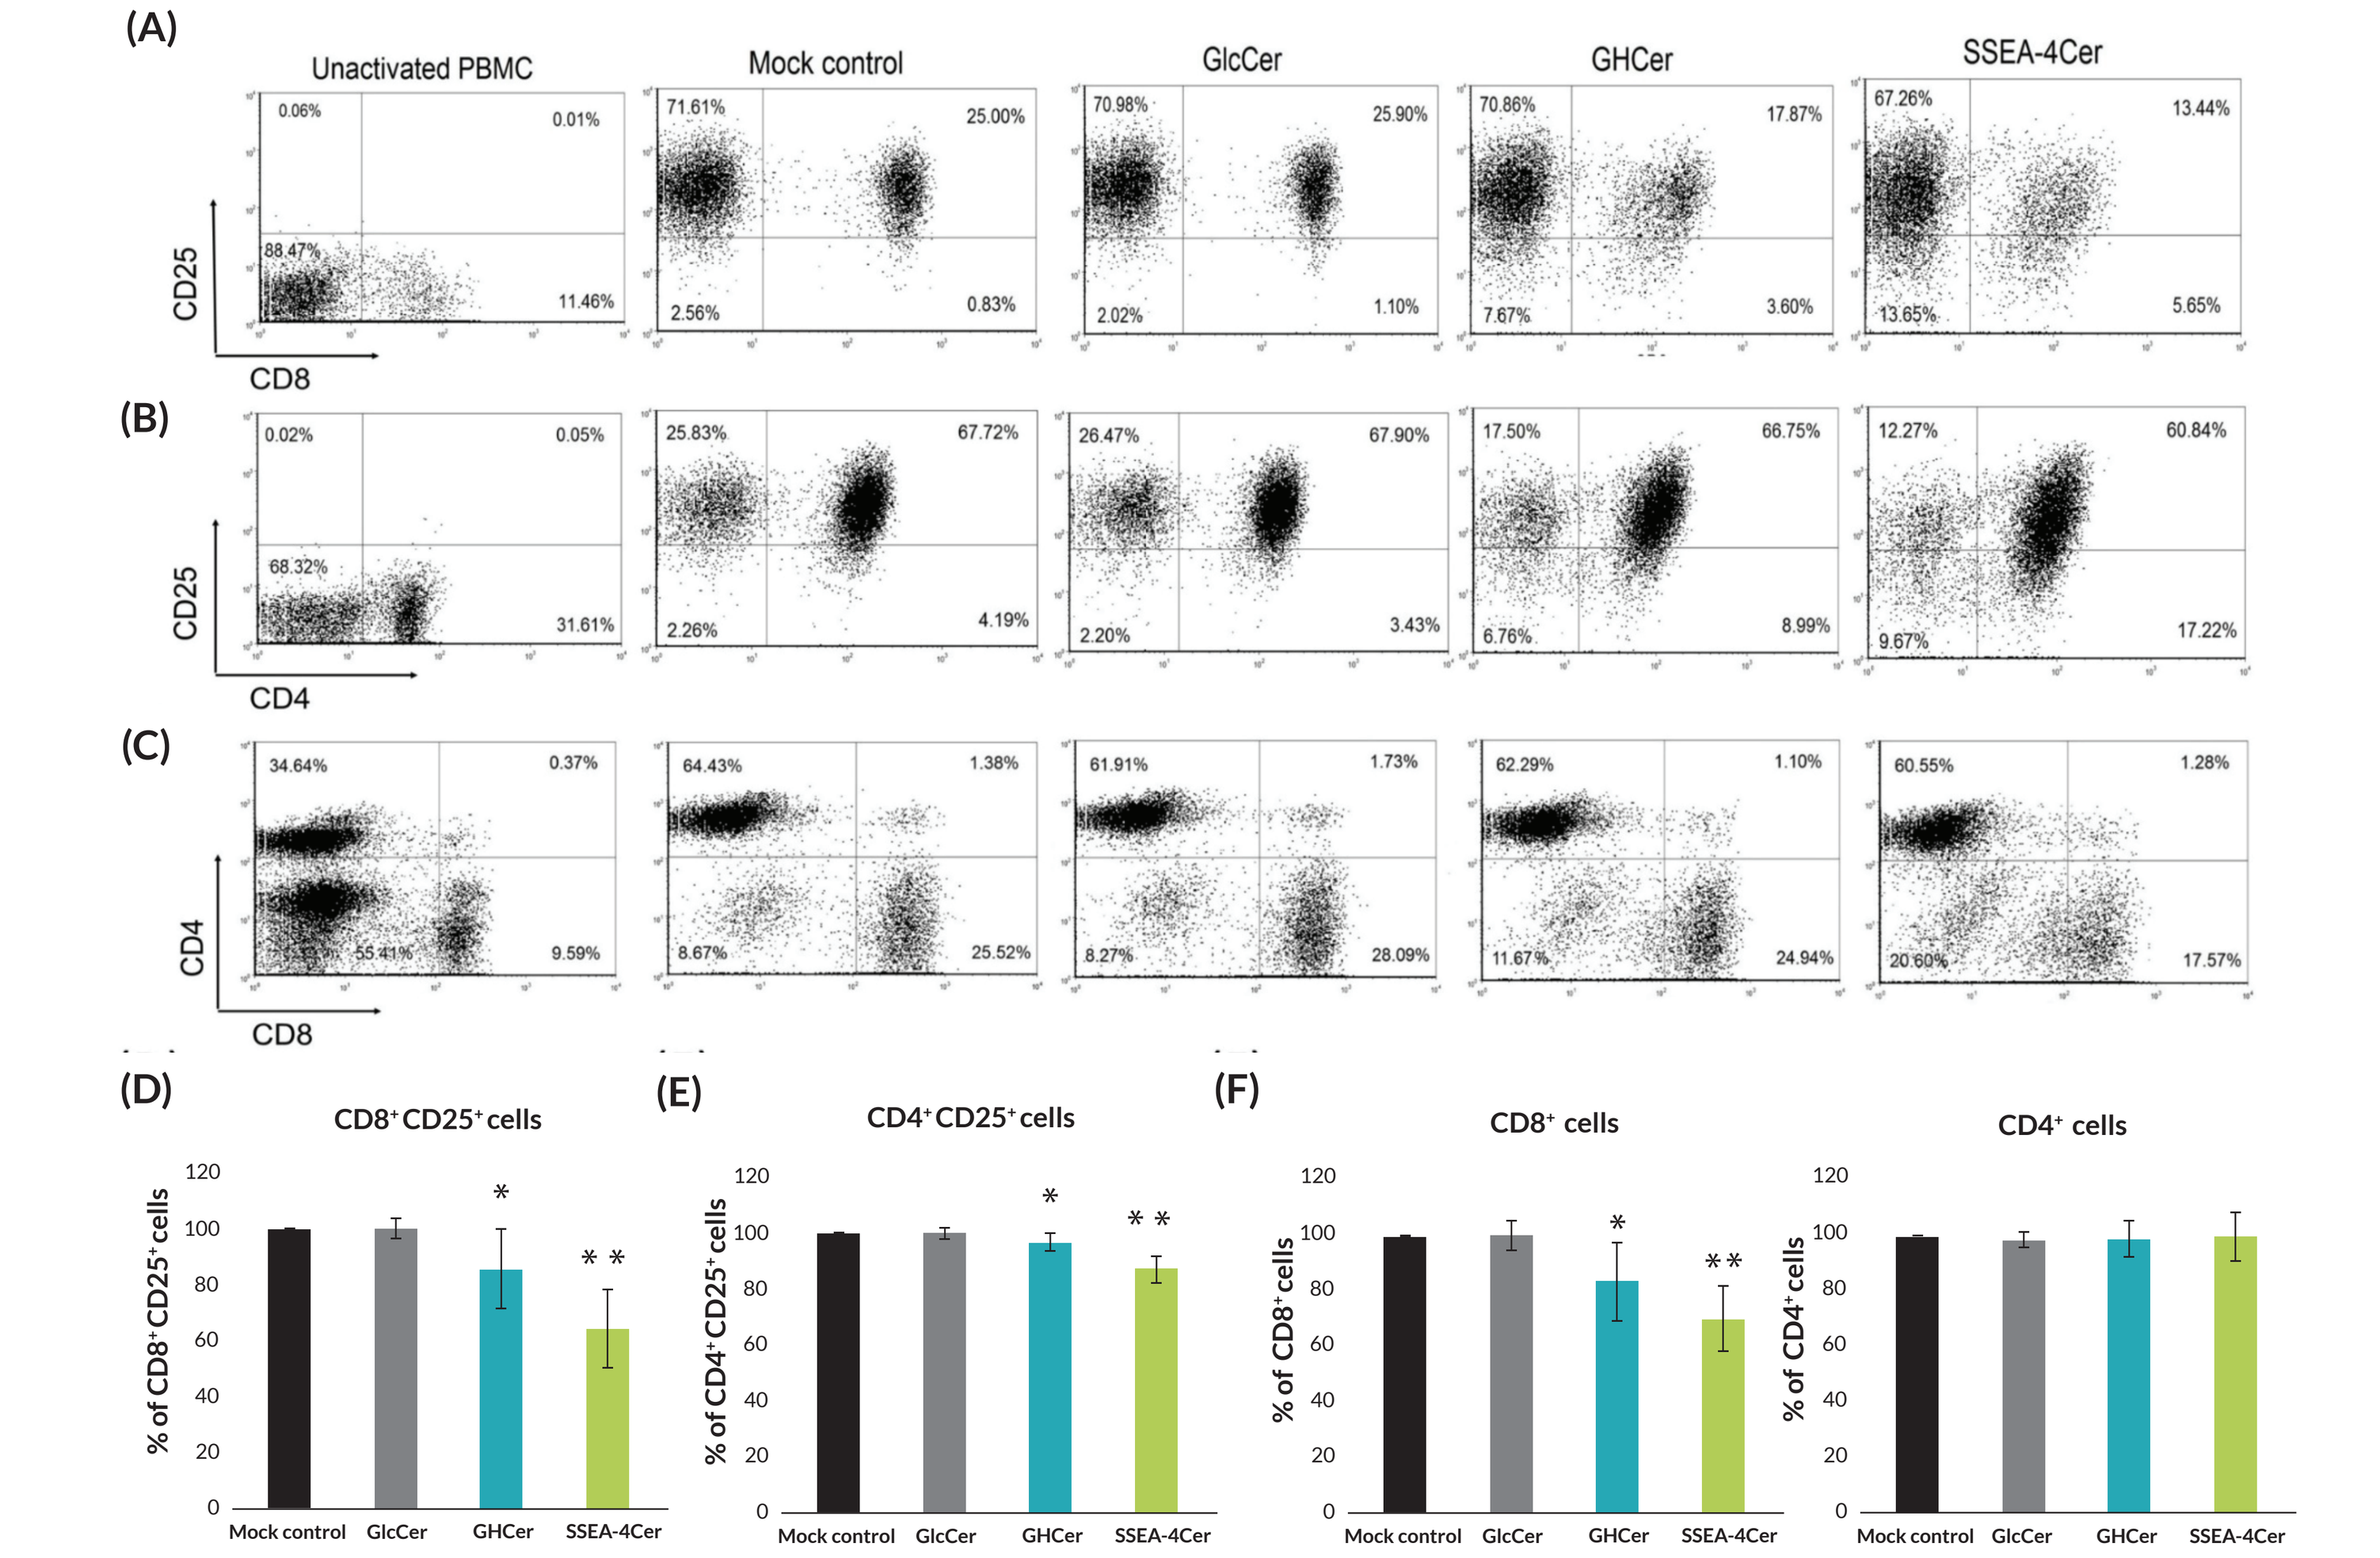
(A)
Unactivated PBMC
Mock control
GlcCer
GHCer
SSEA-4Cer
CD25
CD8
(B)
CD25
CD4
(C)
CD4
CD8
(D)
CD8⁺CD25⁺ cells
(E)
CD4⁺CD25⁺ cells
(F)
CD8⁺ cells
CD4⁺ cells
Mock control
GlcCer
GHCer
SSEA-4Cer

Human PBMCs were stimulated with anti-CD3/CD28 for 48 hr and then were treated with 40 μM GlcCer, GHCer, and SSEA-4Cer for 48 hr. Percentages of activated CD8 and CD4 T-cells and CD25 surface expression were measured by a flow cytometer. Representative experiment shown for **(A)** CD25/CD8, **(B)** CD25/CD4, and **(C)** CD8/CD4. Mean ± SD from >5 independent experiments shown for **(D)** CD25/CD8, **(E)** CD25/CD4, and **(F)** CD8 and CD4. *$P < 0.05$ vs mock controls. **$P < 0.01$ vs mock controls.

Analysis of GHCer- and SSEA-4Cer-signaling in activated T-cells

Figure 6. Analysis of protein expression in GHCer- and SSEA-4Cer-treated T-cells by micro-western array (MWA) assay.

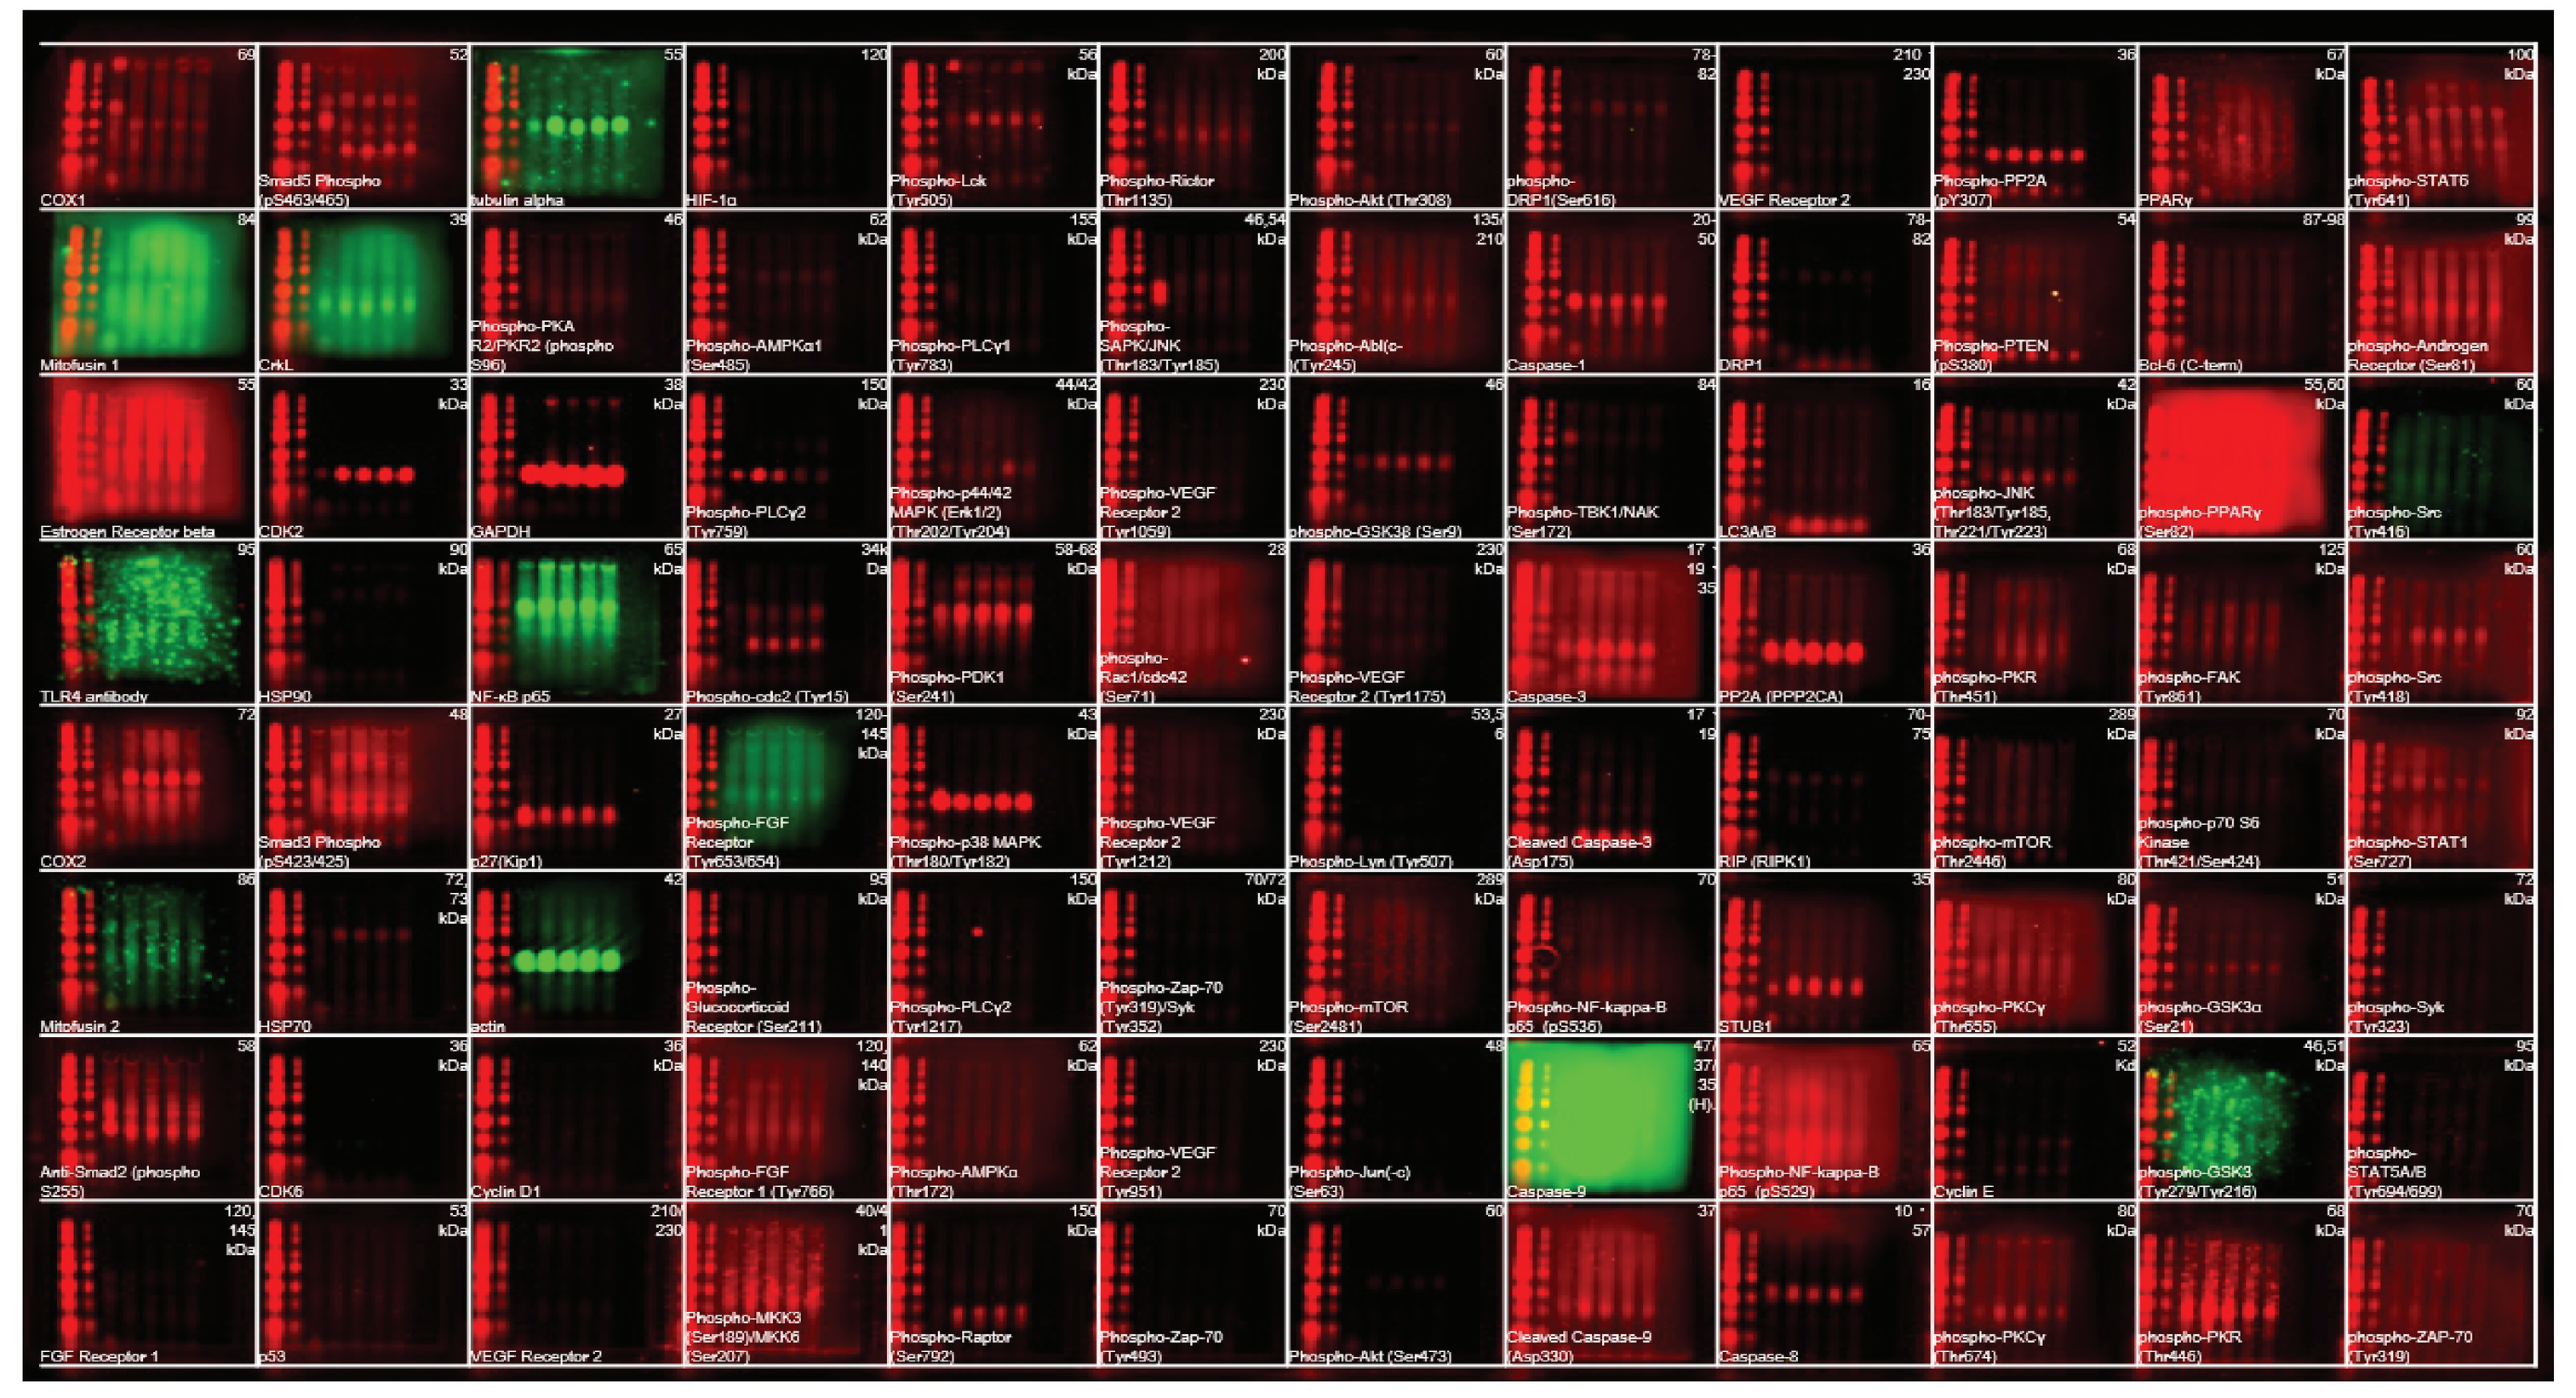

Analysis of protein expression in GHCer- and SSEA-4Cer-treated T-cells by micro-western array (MWA) assay Activated T-cells were treated with or without 40 μM GlcCer, GHCer, or SSEA-4Cer for 48 hr. Changes in abundance of indicated proteins or their phosphorylated forms were determined by MWA. Five samples printed in each well (from left to right) were condition control (1), mock control cells (2), and cells treated with 40 μM GlcCer, GHCer, or SSEA-4Cer (3-5), respectively. Artificial coloring differentiates the used secondary antibodies in species (red and green for anti-rabbit and anti-mouse, respectively).

GHCer and SSEA-4Cer inhibited T-cell activation-related signaling

Figure 7. GHCer and SSEA-4Cer interfered with T-cell receptor-mediated signaling.

Antibodies		Relative protein abundance		
		GlcCer	GHCer	SSEA-4Cer
Decreased	Phospho-Lck (Tyr505)	1.05	1.096	0.601
	RIPK1	0.893	0.873	0.759
	Phospho-PKR (Thr446)	0.79	0.547	0.452
	Phospho-STAT1 (Ser423/Ser425)	0.781	0.669	0.601
	Phospho-Smad3 (Ser423/Ser425)	0.838	0.662	0.653
	Cleaved Caspase-3 (Asp175)	1.042	0.741	1.021
	Cleaved Caspase-9 (Asp330)	0.629	0.626	0.605
Increased	P27	1.016	1.126	1.541
	Phospho-Akt (Thr308)	0.869	1.043	1.237
	Phospho-Akt (Thr473)	0.948	1.174	1.463
	Phospho-GSK3β (Ser9)	0.937	1.062	1.332
	Phospho-p38 MAPK (Thr180/Tyr182)	1.069	1.329	1.199
	Phospho-p44/42 MAPK (Erk1/2)	1.247	1.241	1.648
	(Thr202/Tyr204)	0.927	2.832	1.291

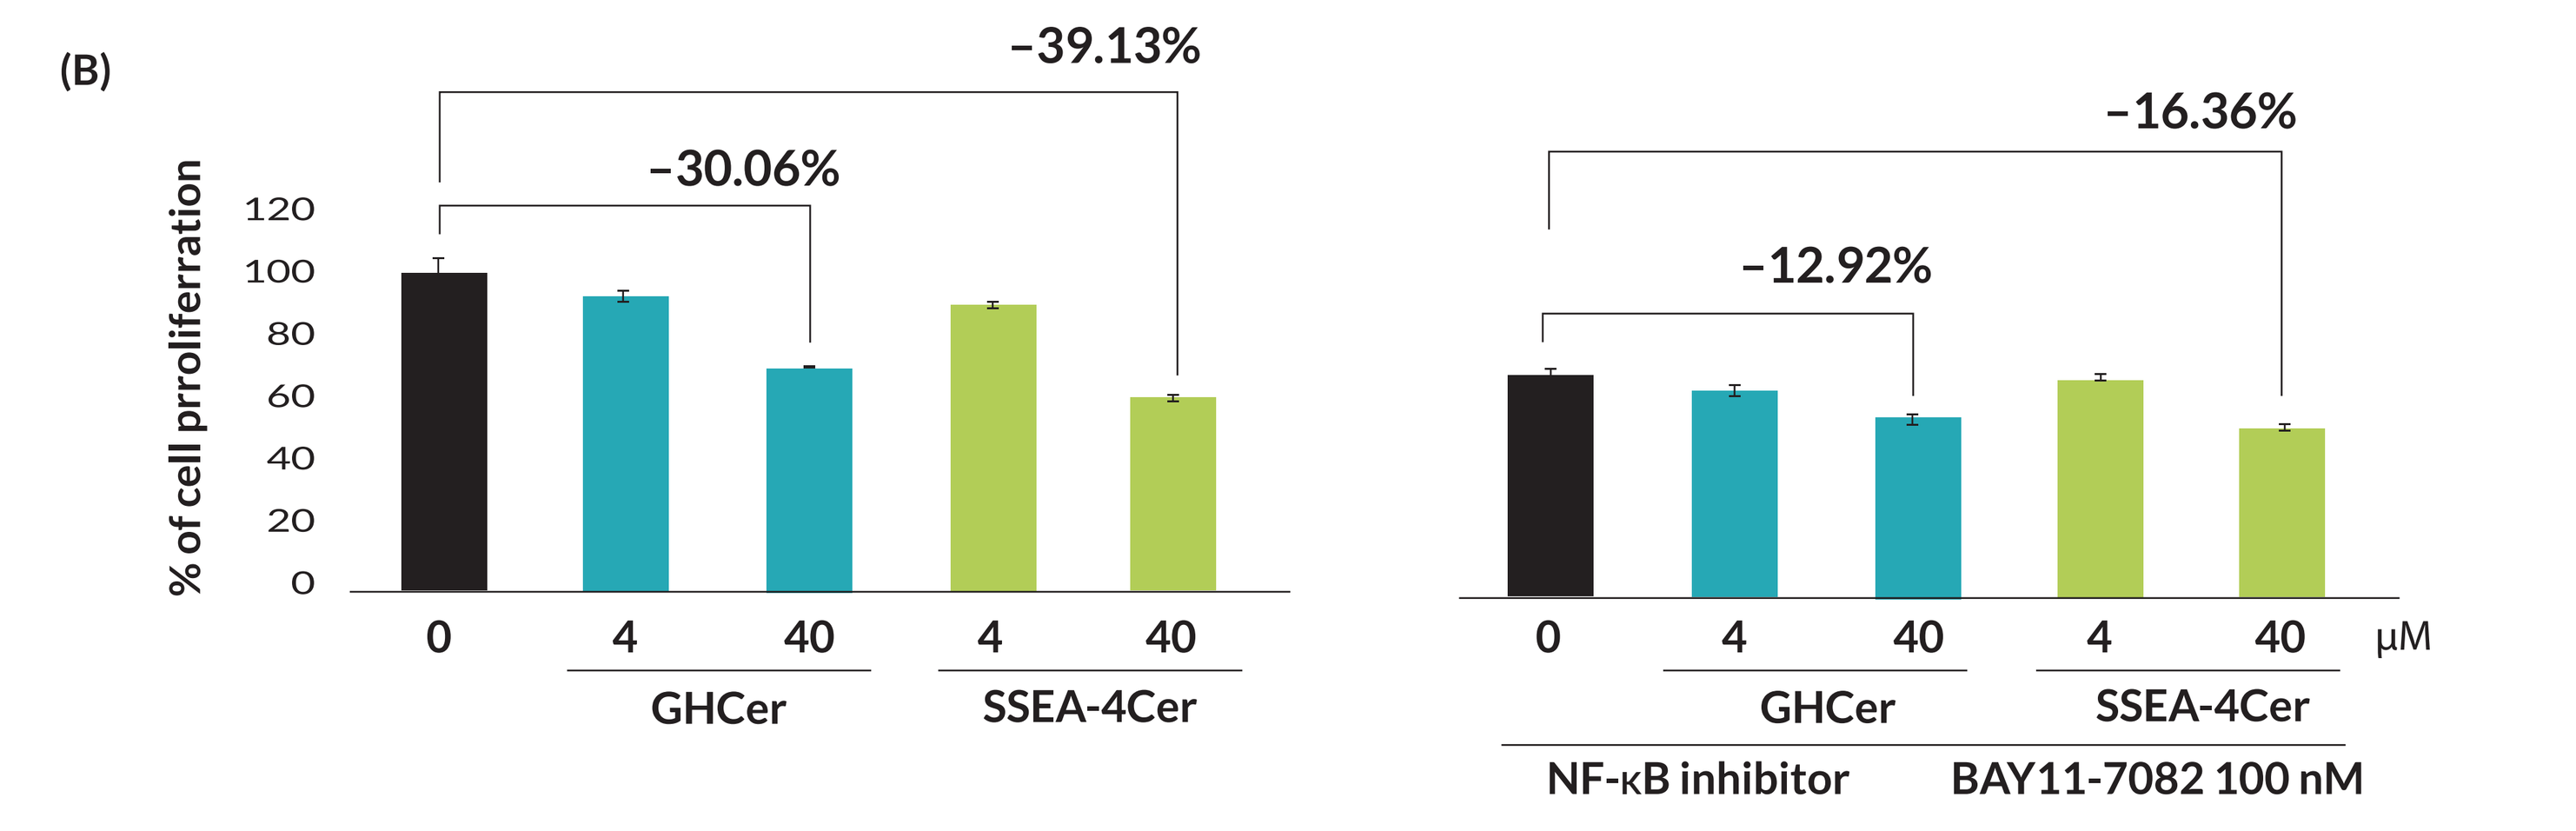
(B)
% of cell proliferration
−39.13%
−30.06%
−16.36%
−12.92%
0
4
40
μM
GHCer
SSEA-4Cer
NF-κB inhibitor
BAY11-7082 100 nM

(A) Selected data of relative protein abundance are listed in the table. Relative protein abundance was normalized to the average of glyceraldehyde 3-phosphate dehydrogenase (GAPDH) and actin. A fold change of 1.2 or 0.8 in expression was used as a cutoff for upregulation or downregulation, respectively. **(B)** Pretreatment with nuclear factor kappa light chain enhancer of activated B cells (NF-κB) inhibitor BAY11-7082 inhibited GHCer and SSEA-4Cer-induced cell viability enhancement. Activated T-cells were stimulated with or without 4 or 40 μM of GHCer and SSEA-4Cer for 48 hr after pretreatment with 0.1 μM BAY11-7082 for 30 mins. After removing GHCer and SSEA-4Cer, cell viability was determined by MTS assay. Each experiment was repeated 3 times, and similar results were obtained.

SUMMARY

- Globo H ceramide (GHCer) enhanced B16F10 melanoma tumor growth with decreased tumor-infiltrating lymphocytes (TILs) in mice
- Significant uptake of GHCer and SSEA-4Cer by T-cells was demonstrated via exogenous addition of the Globo ceramides
- GHCer- and SSEA-4Cer-treated activated T-cells showed a downregulated proliferative activity
- GHCer and SSEA-4Cer inhibited CD3+ T-cell activation of human PBMCs
- GHCer and SSEA-4Cer induced T-cell arrest primarily through reduction of CD8+ T-cell proliferation
- T-cell activation–related signaling is downregulated in GHCer and SSEA-4Cer-treated activated T-cells
- NF-κB inhibitor reduced the effects of GHCer and SSEA-4Cer on inhibition of T-cell proliferation

CONCLUSIONS

- GHCer inhibits T-cell infiltration into the tumor in vivo. GH and SSEA-4 exhibit immunosuppressive activity on activated T-cells, which suggests that both GHCer and SSEA-4Cer may play a role in regulating immune responses in the tumor microenvironment.

Prepared for American Association for Cancer Research (AACR) 2021 Virtual Meeting, April 10-15 and May 17-21, 2021.